ATOR-1017, a tumor directed Fcγ-receptor cross-linking dependent 4-1BB agonistic antibody

Karin Enell Smith*, Anna Rosén, Anna Dahlman, Karin Barchan, Ida Åberg, Doreen Werchau, Mia Thagesson, Niina Veitonmäki, Christina Furebring, Peter Ellmark
Alligator Bioscience AB, Lund, Sweden *Presenting author

Background and Mode of Action

ATOR-1017 format

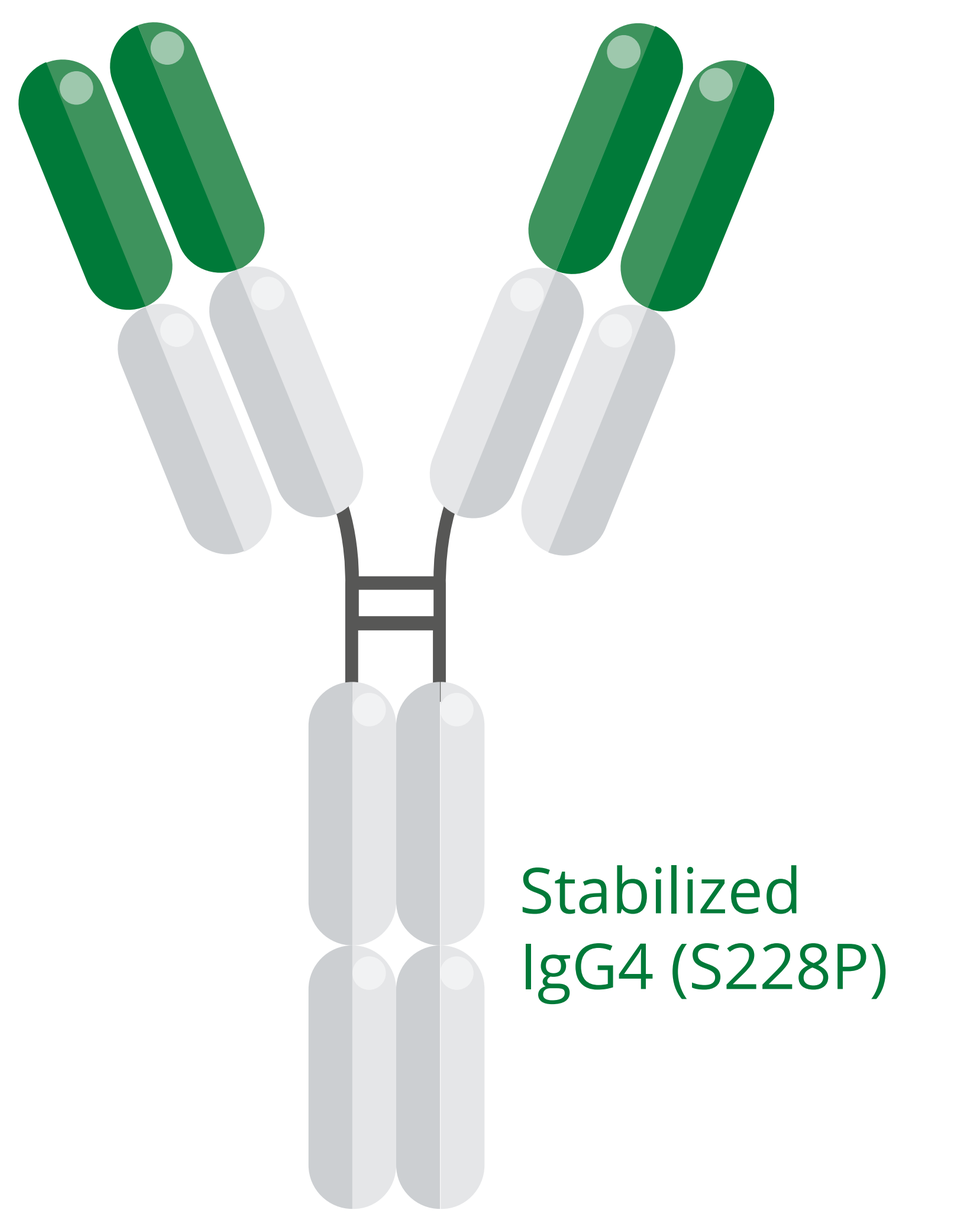
Stabilized
IgG4 (S228P)

Agonistic effect that depends on cross-linking with specific FcγRs

Binding to the 4-1BB receptor

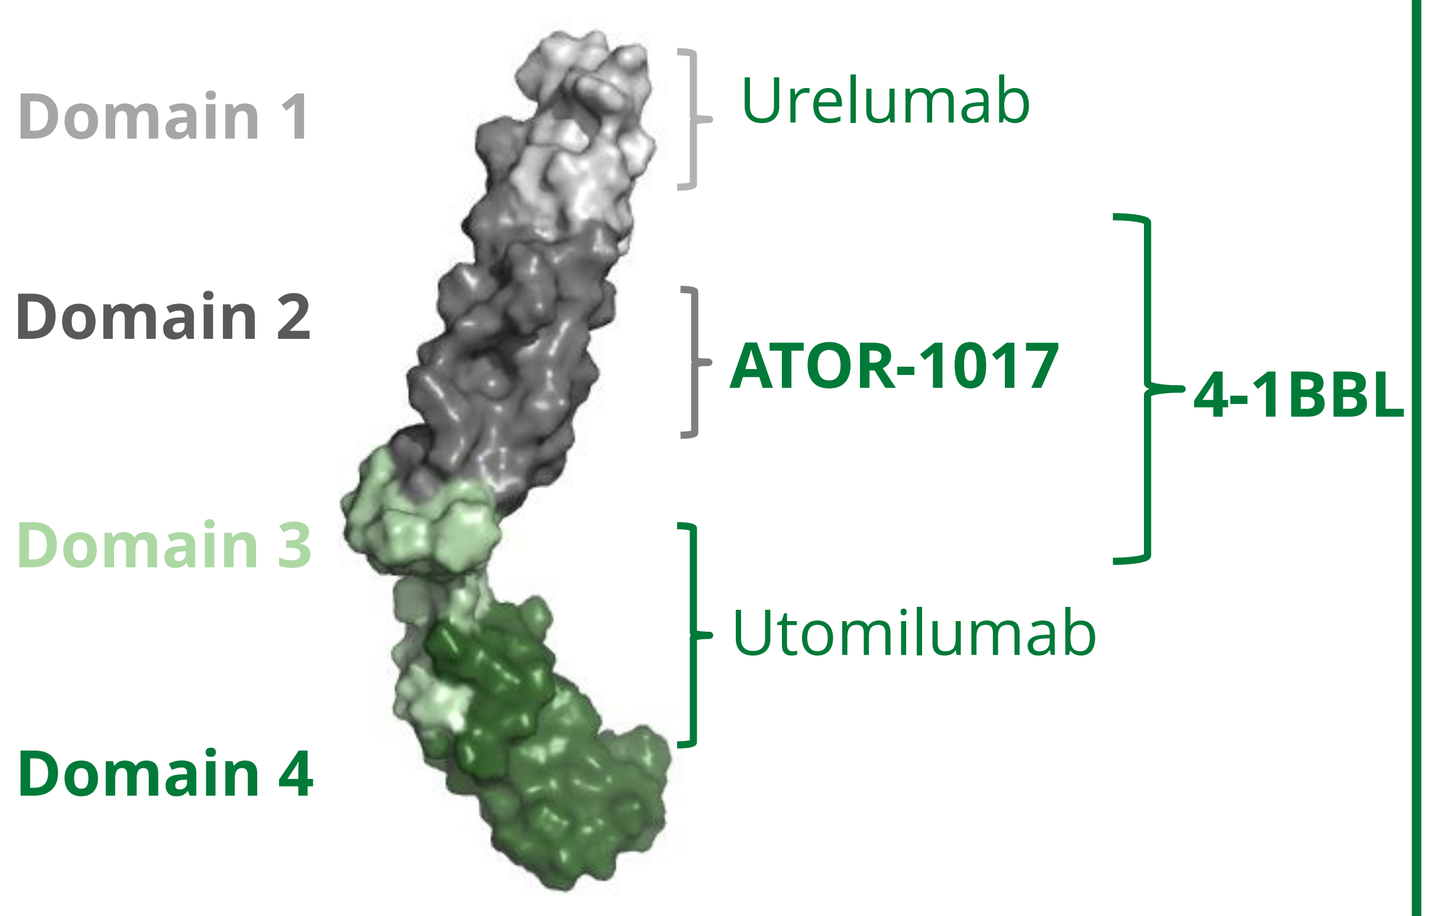
Domain 1
Domain 2
Domain 3
Domain 4
Urelumab
ATOR-1017
4-1BBL
Utomilumab

Unique binding domain of the 4-1BB receptor

Mode of action

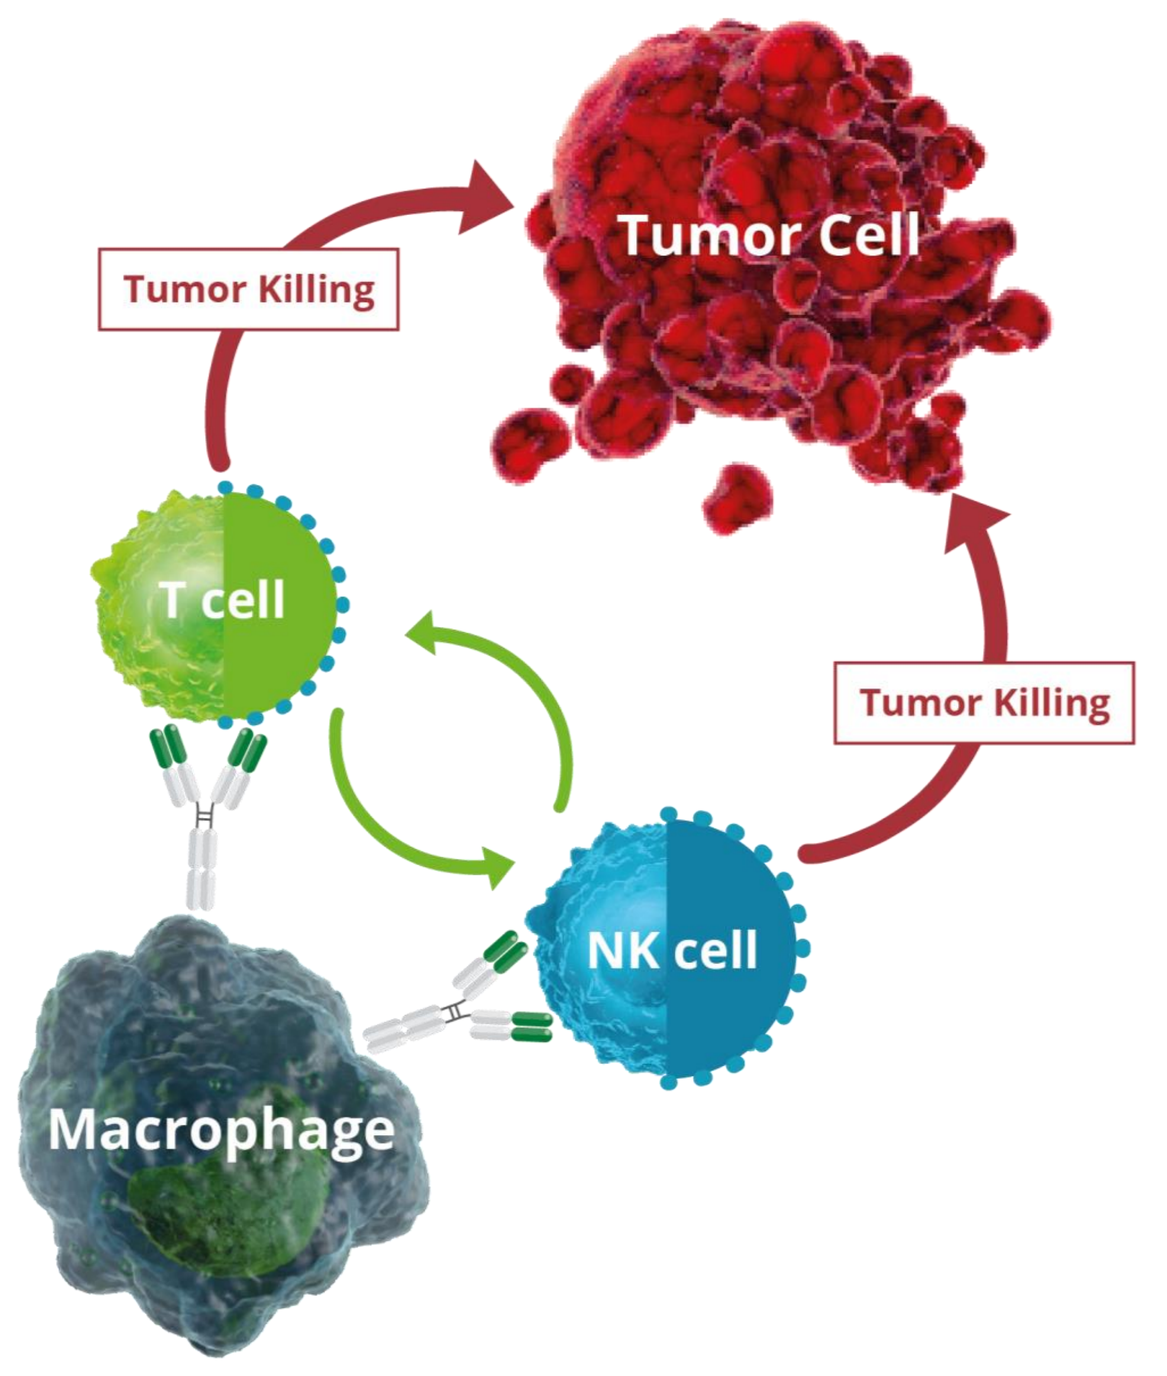
Tumor Cell
Tumor Killing
T cell
Tumor Killing
NK cell
Macrophage

Activates tumor infiltrating effector T cells and NK cells expressing 4-1BB

- ATOR-1017 is designed for an optimal efficacy and improved safety, by combining the IgG4-format that mediates a potent FcγR cross-linking with a unique binding epitope on 4-1BB
- ATOR-1017 generates a strong tumor directed immune response in patients with tumors infiltrated with 4-1BB expressing T cells and NK cells and cells expressing specific FcγRs

4-1BB and specific FcγRs are co-expressed and co-localized in tumors

4-1BB and FcγR mRNA are co-expressed in solid and hematological tumors

No 4-1BB and FcγR mRNA co-expression in normal tissue

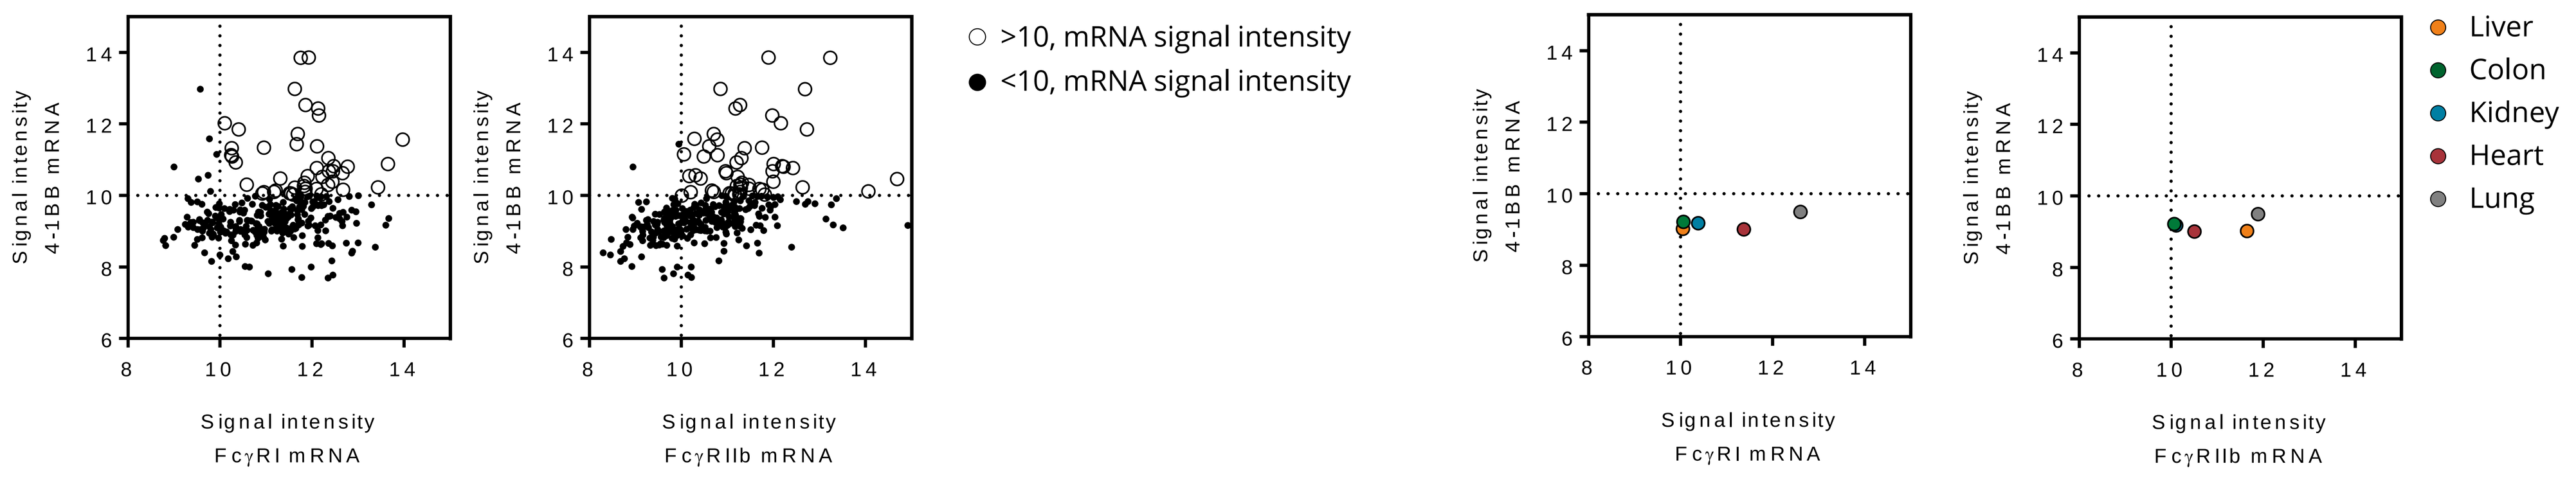
Signal intensity 4-1BB mRNA
Signal intensity FcγRI mRNA
Signal intensity FcγRIIb mRNA
>10, mRNA signal intensity
<10, mRNA signal intensity
Liver
Colon
Kidney
Heart
Lung

4-1BB and FcγRI or FcγRIIb mRNA are co-expressed in in tumor tissue. mRNA expression values were obtained from the public Affymetrix microarray platform. mRNA expression signal intensity range is between 5-19 on a log2 scale. Tumors with high level of co-expression (>10) include renal, breast, ovarian tumor and lung cancer as well as T and B cell lymphomas. No 4-1BB and FcγRI or FcγRIIb mRNA co-expression was observed in normal tissues such as liver, colon, kidney, heart and lung.

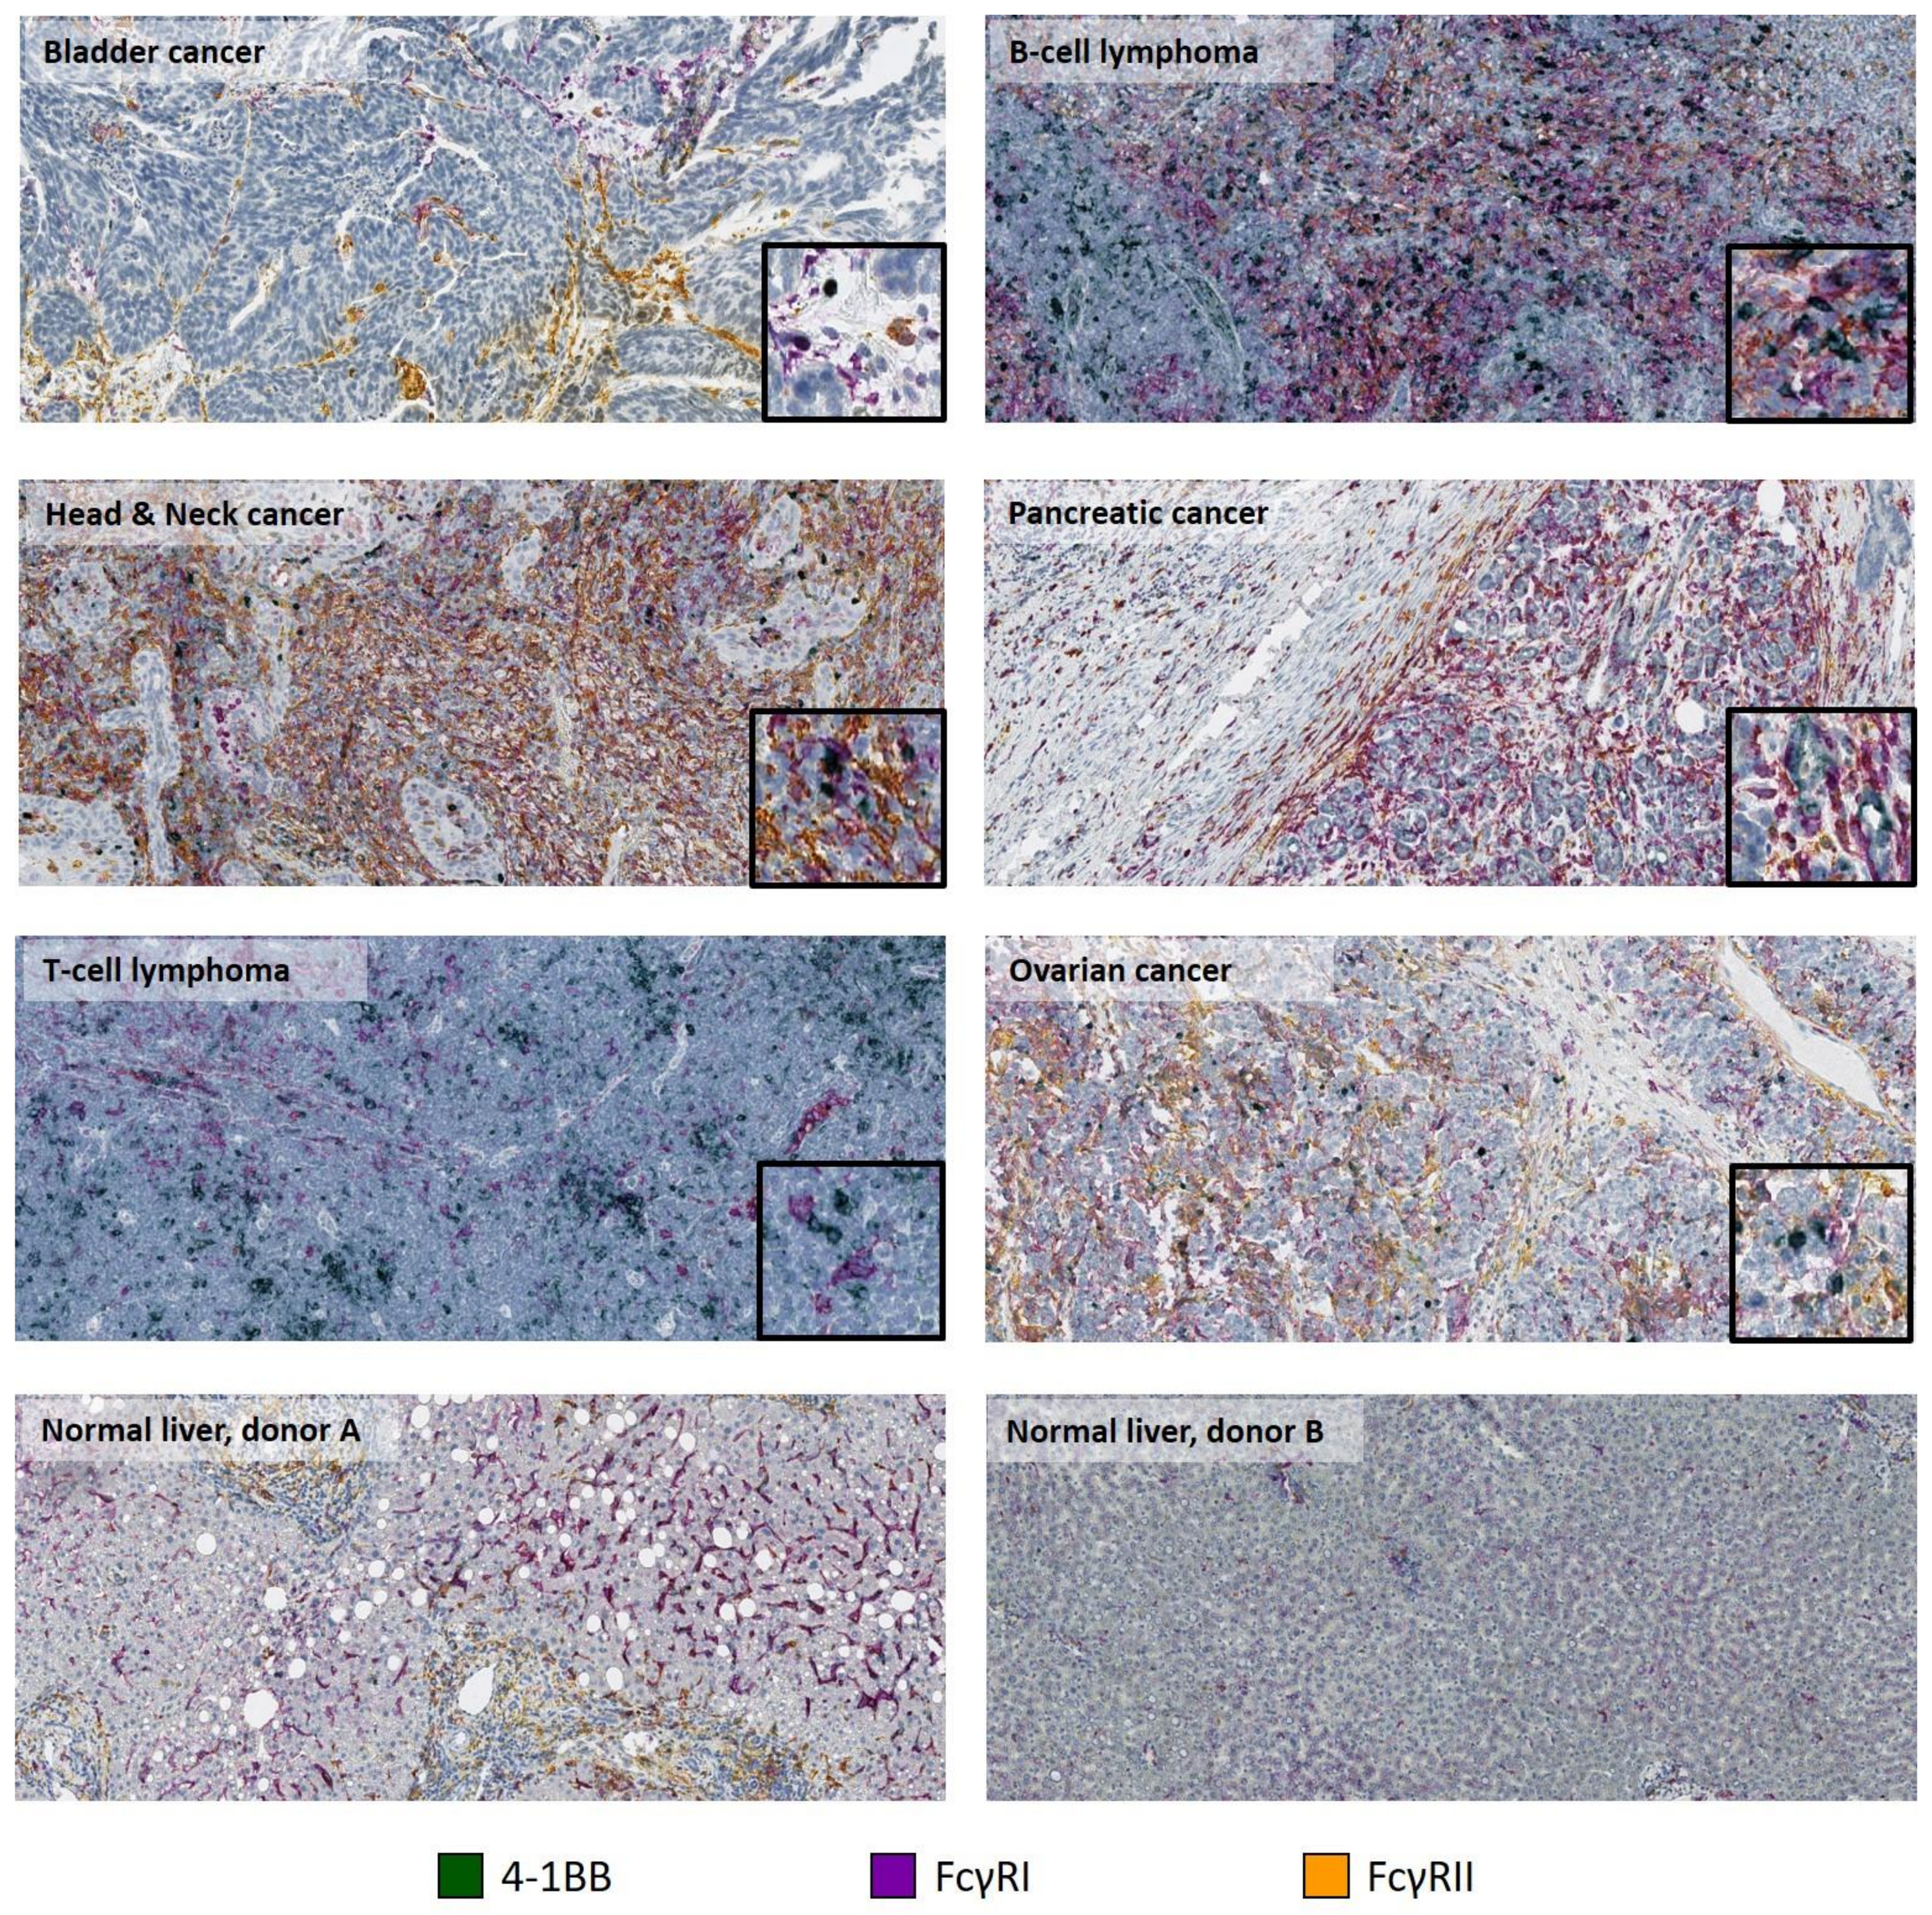
Bladder cancer
B-cell lymphoma
Head & Neck cancer
Pancreatic cancer
T-cell lymphoma
Ovarian cancer
Normal liver, donor A
Normal liver, donor B
4-1BB
FcγRI
FcγRII

Indication	4-1BB co-localized w. FcγRs on immune cells	Highest freq. on immune cells	4-1BB co-localized w. FcγRs in intra-tumoral stroma	Highest freq. in intra-tumoral stroma
Bladder cancer	3/3	FcgRI or FcgRII	3/3	FcgRI or FcgRII
Lung cancer	1/2	FcgRII	2/2	FcgRII or 4-1BB
Head and neck cancer	2/2	FcgRI	2/2	FcgRI
Ovarian cancer	2/3	FcgRI or FcgRII	2/3	FcgRI or FcgRII
Pancreatic cancer	3/3	FcgRI	3/3	FcgRI
B cell lymphoma	3/3	4-1BB (2/3)	2/3	FcgRI or FcgRII
T cell lymphoma	1/3	4-1BB (2/3)	1/3	4-1BB (2/3)

4-1BB and FcγRI or FcγRII protein are co-expressed in tumor tissue but not in normal human liver. A multiplex immunohistochemical (IHC) staining method was developed and employed to assess the expression of FcγRI, FcγRII and 4-1BB in whole sections of formalin-fixed paraffin embedded (FFPE) tumors from different indications (n=2-3 donors per indication). 4-1BB (clone BBK-2, ThermoFisher) is visualized in green; FcγRI (clone OTI3D3, Abcam) in purple; and FcγRII (clone EPR6658, Abcam) in orange.

The agonistic effect of ATOR-1017 depends on cross-linking with FcγRs

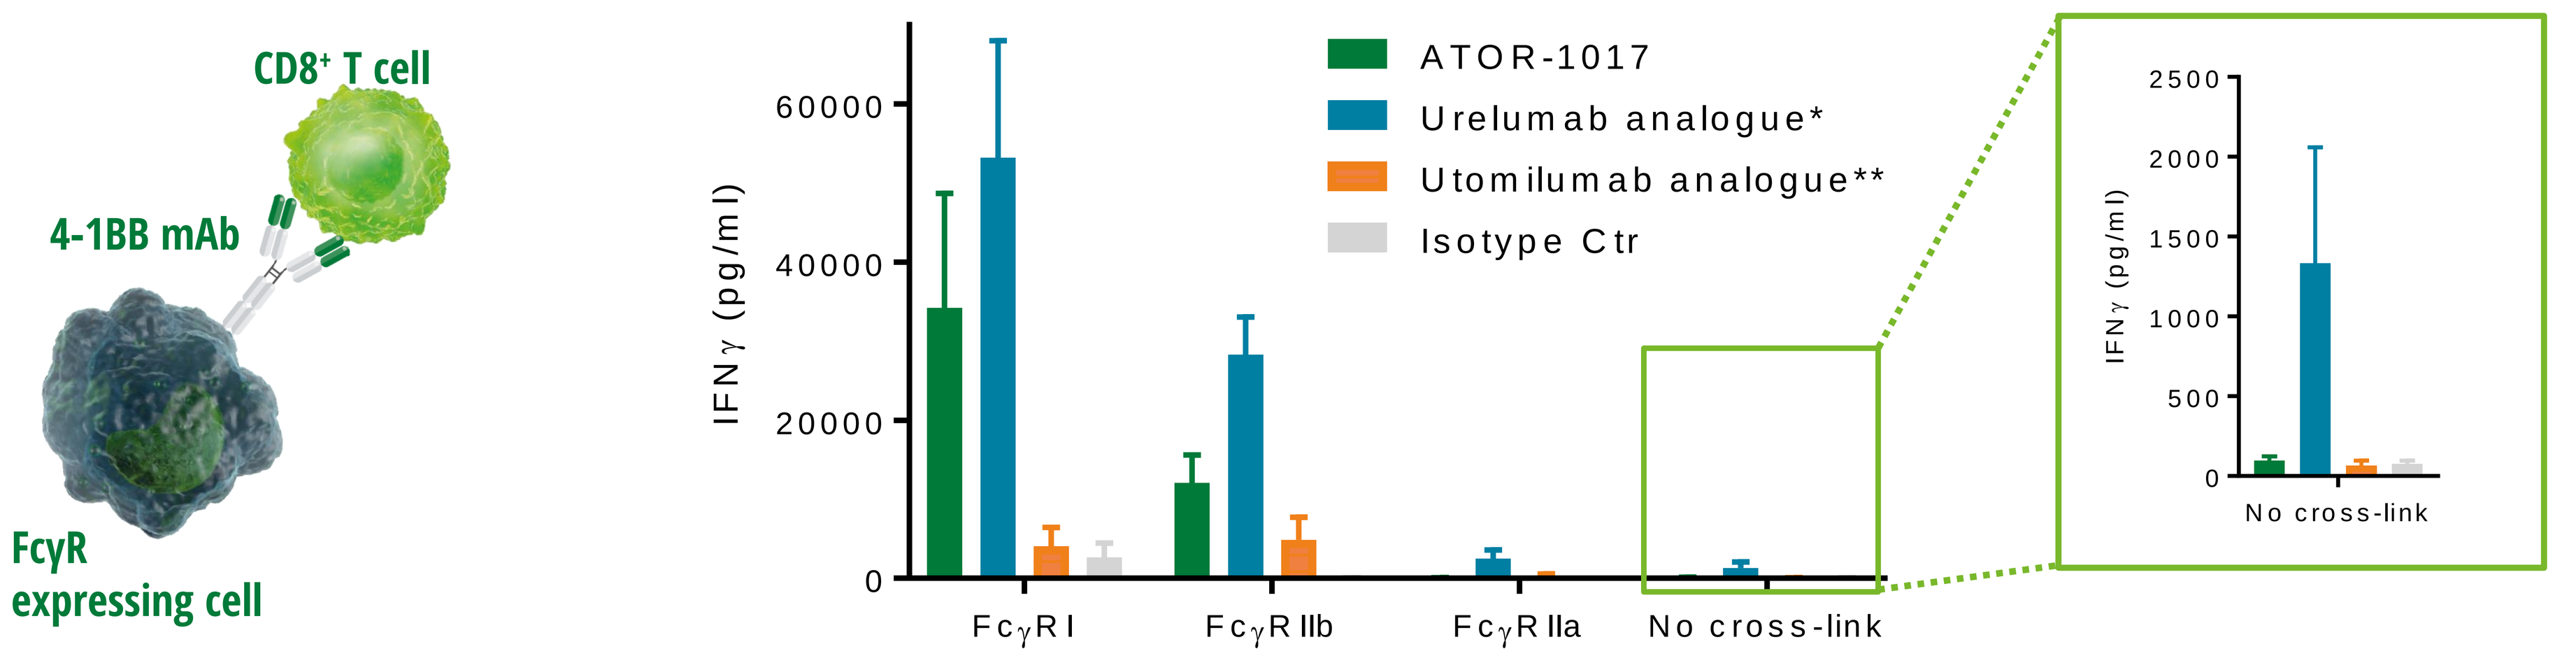
CD8+ T cell
4-1BB mAb
FcγR
expressing cell
ATOR-1017
Urelumab analogue*
Utomilumab analogue**
Isotype Ctr
IFNγ (pg/ml)
FcγRI
FcγRIIb
FcγRIIa
No cross-link

ATOR-1017 is dependent on cross-linking with specific FcγRs. FcγR cross-linking dependency of ATOR-1017 was demonstrated using primary human CD8+ T cells and FcγR-transfected CHO cells. CD8+ T cells were stimulated with a suboptimal concentration of anti-CD3 mAb, and co-stimulated with ATOR-1017, or analogues of the 4-1BB antibodies urelumab and utomilumab in the presence or absence of FcγRI, FcγRIIa or FcγRIIb-transfected cells. IFN-γ concentrations were determined in supernatants by ELISA and shown as mean ± SEM (n≥5) .
*US8137667 seq 1 and 4, **US8337850 seq 43 and 45

ATOR-1017 activates cytotoxic CD8+ T cells

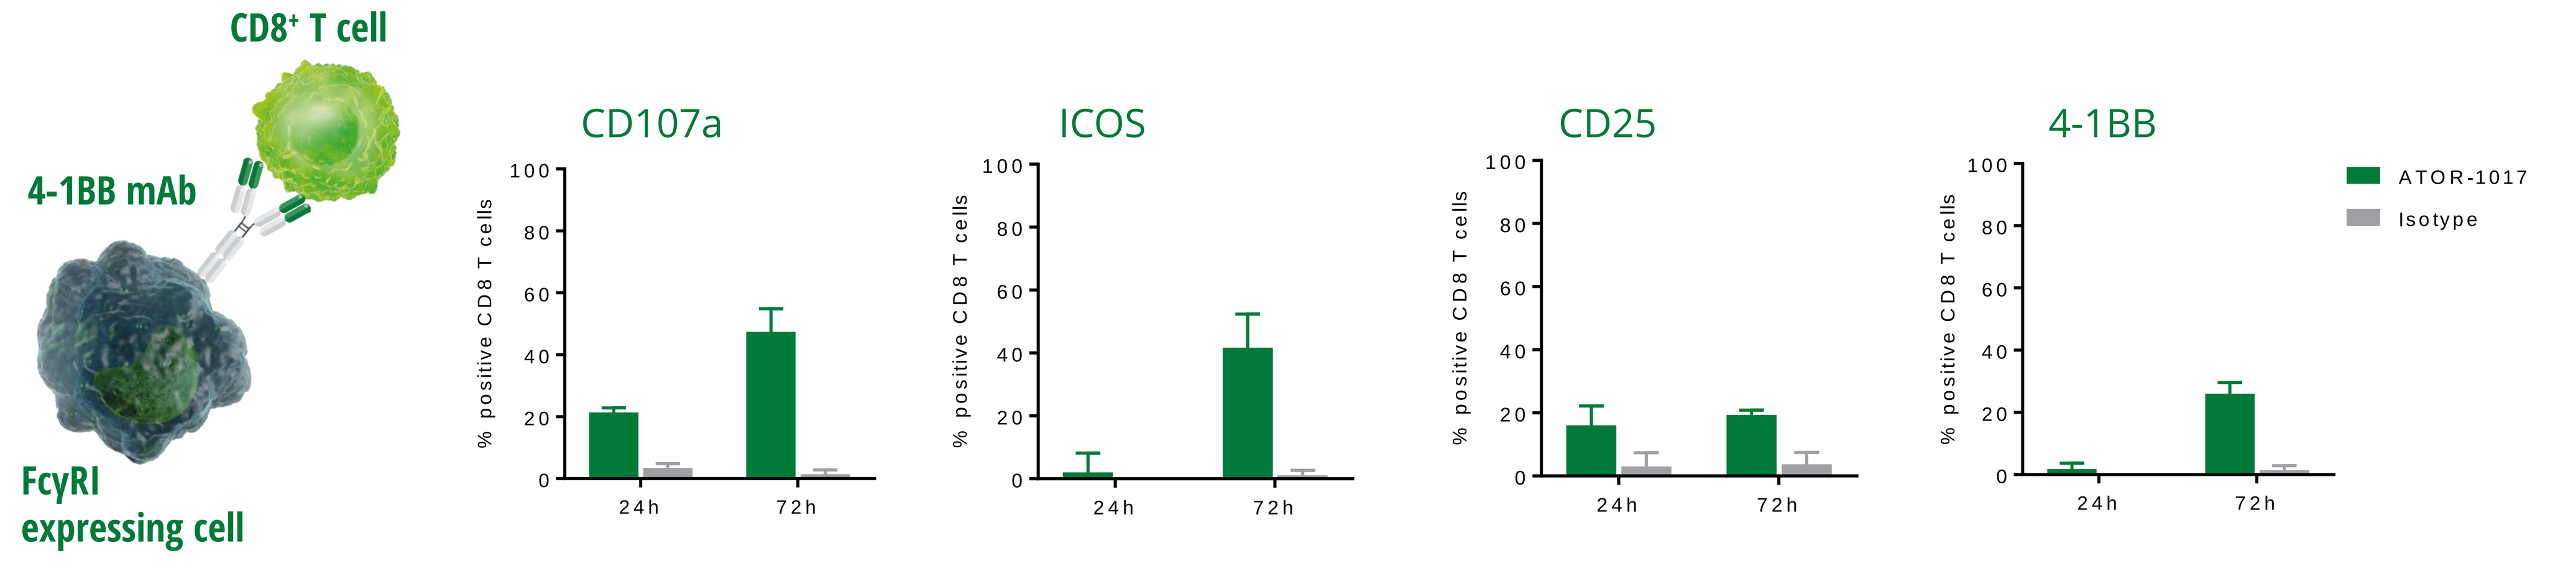
CD8+ T cell
4-1BB mAb
FcγRI
expressing cell
CD107a
ICOS
CD25
4-1BB
% positive CD8 T cells
24h
72h
ATOR-1017
Isotype

ATOR-1017 activates cytotoxic CD8+ T cells. CD8+ T cell activation of ATOR-1017 was demonstrated using primary human CD8+ T cells stimulated with a suboptimal concentration of anti-CD3 mAb, and co-stimulated with ATOR-1017 in the presence of FcγRI-transfected cells. Following incubation, CD8+ T cells were analyzed with flow cytometry for expression of CD107a, ICOS, CD25 and 4-1BB and shown as mean ± SD (n=3).

ATOR-1017 activates cytotoxic NK cells

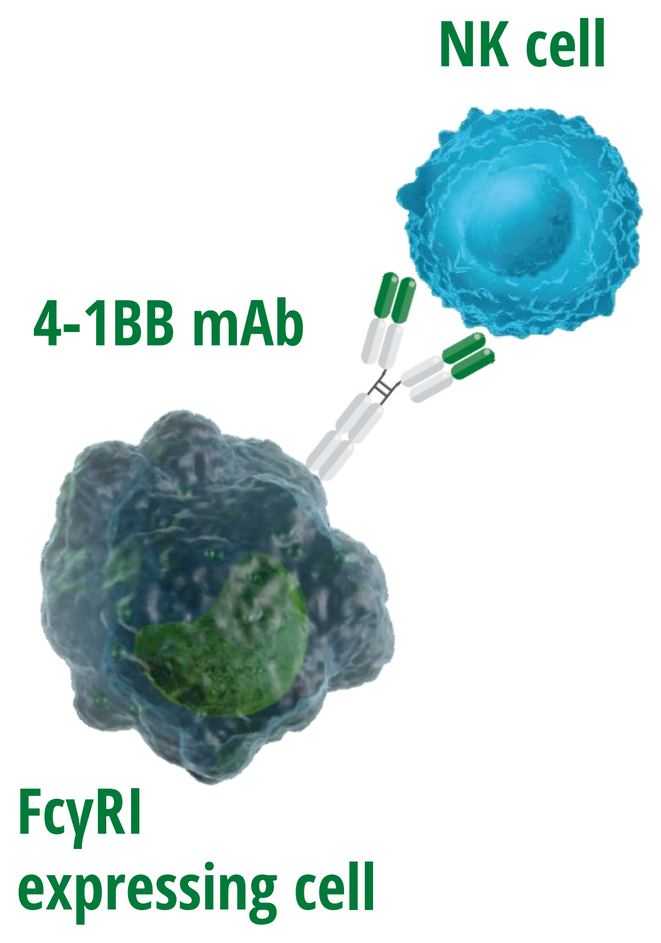
NK cell
4-1BB mAb
FcγRI
expressing cell

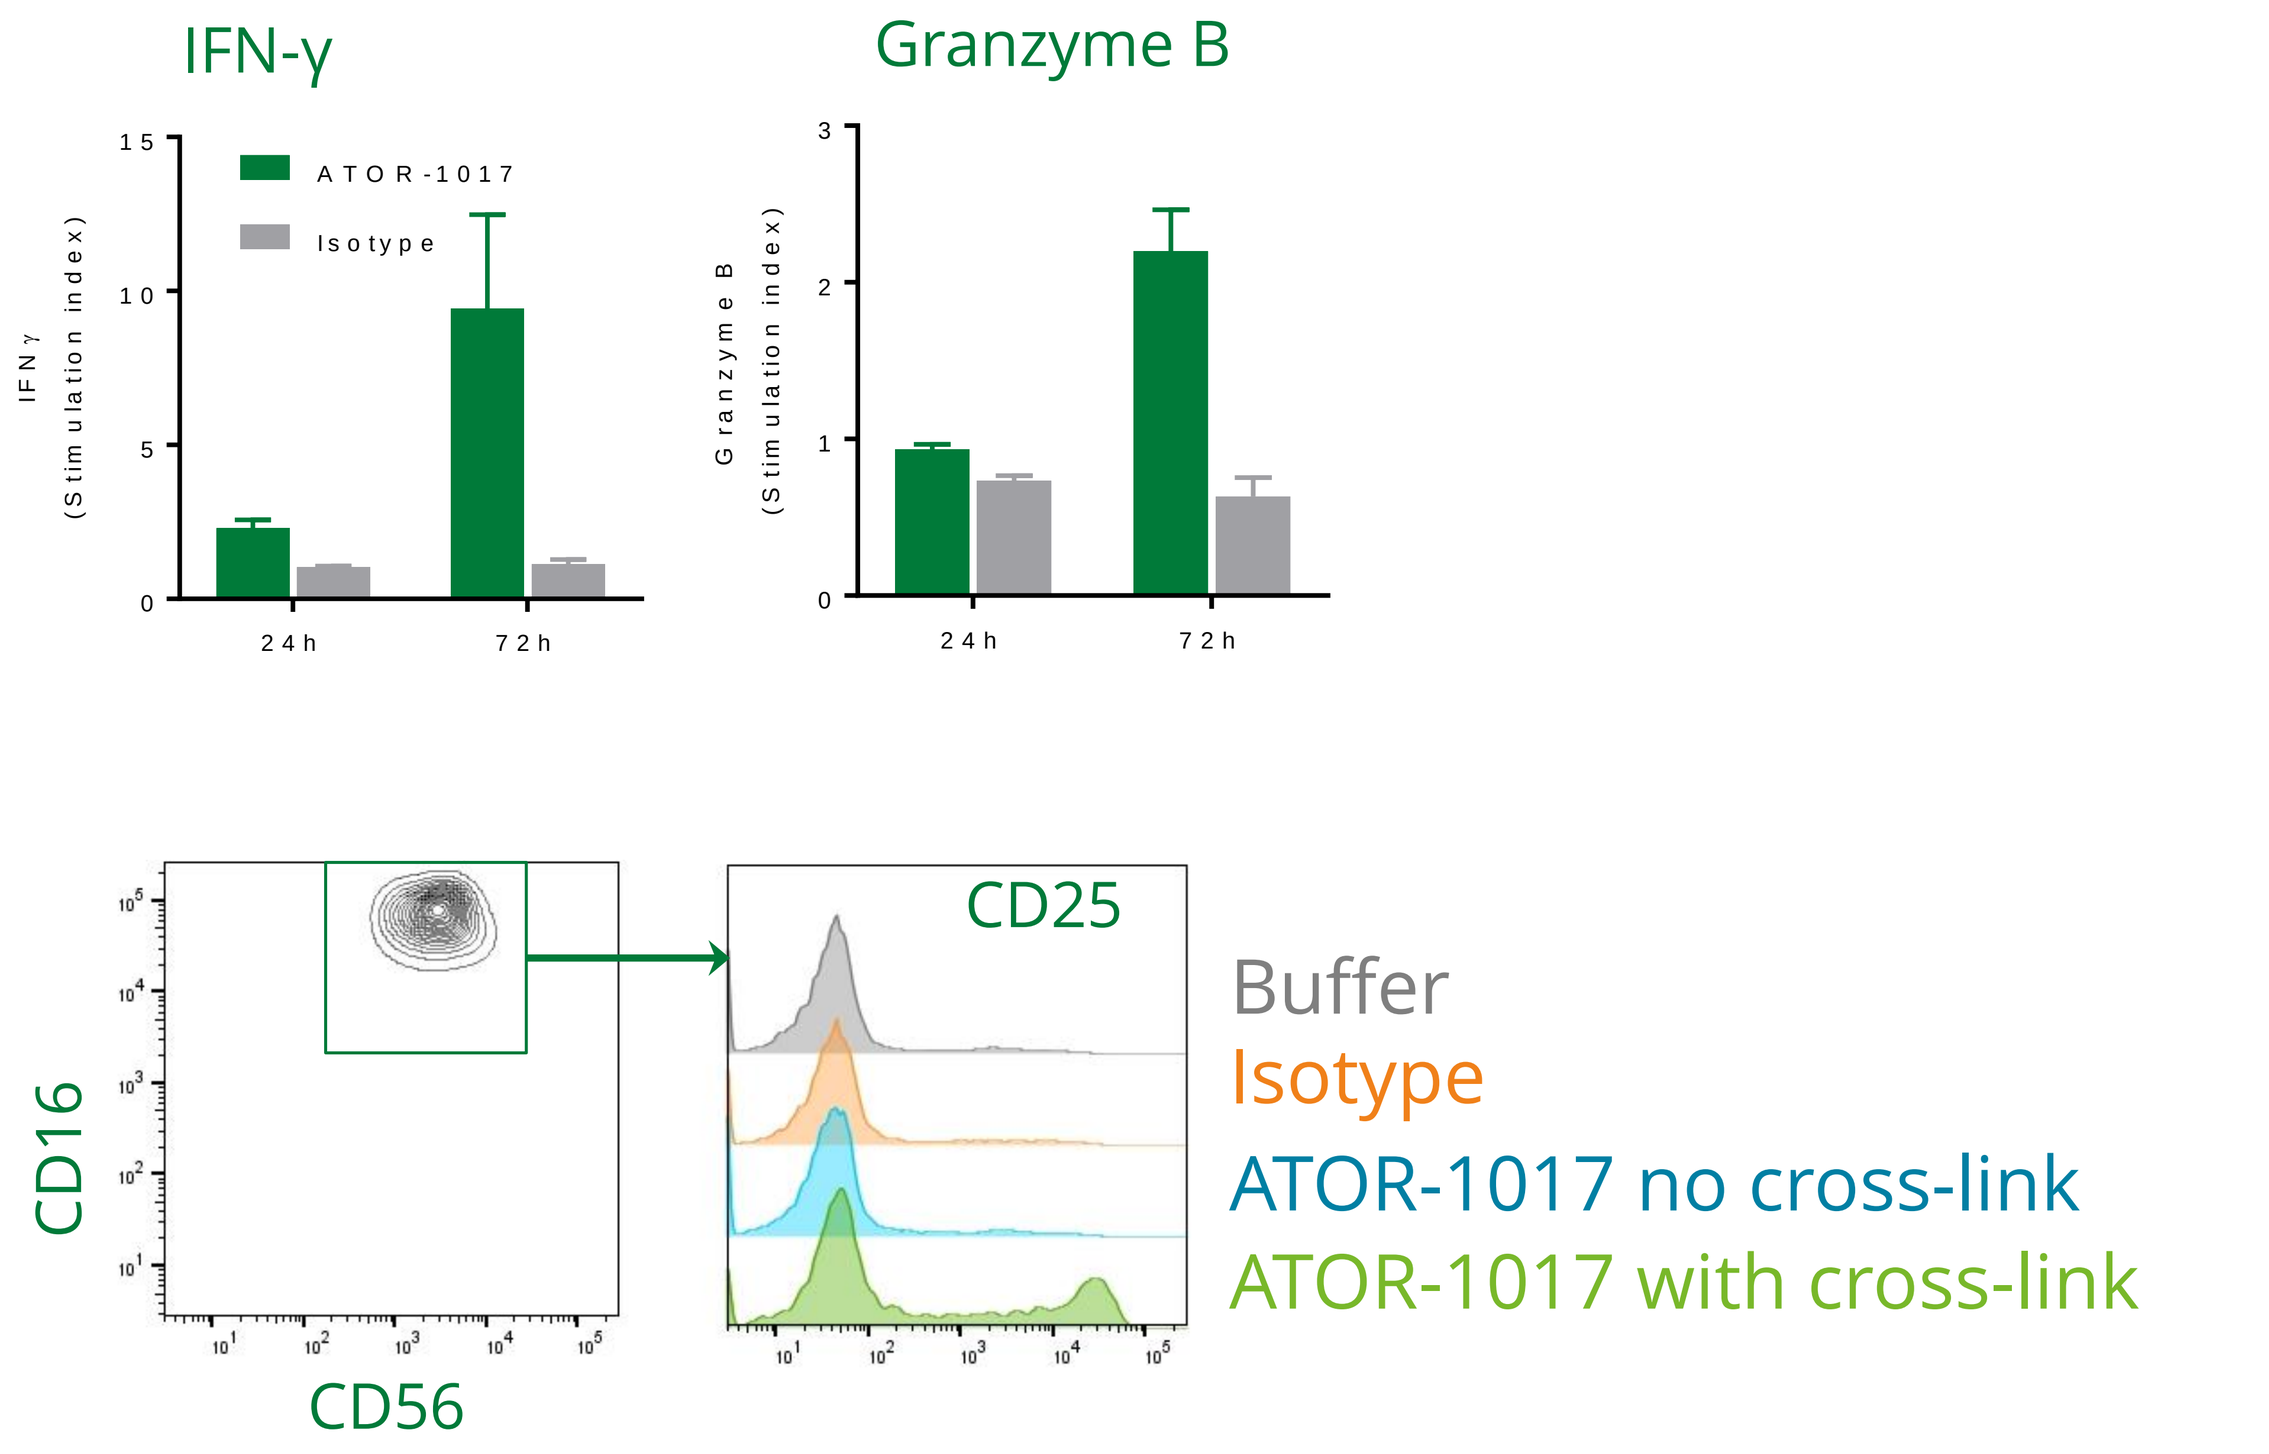
IFN-γ
Granzyme B
ATOR-1017
Isotype
24h
72h
CD16
CD56
CD25
Buffer
Isotype
ATOR-1017 no cross-link
ATOR-1017 with cross-link

ATOR-1017 activates cytotoxic NK cells. NK cells were purified from human peripheral blood (n=6), pre-activated with 200 IU (10 ng/ml) IL-2 over night and activated with ATOR-1017 cross-linked with FcγRI expressing cells. Following 24, 48 or 72 h incubation, supernatants were harvested and IFN-γ and Granzyme B quantified by ELISA and CD25 expression of the NK cells ($CD3^-CD56^+CD16^+$) was analyzed with flow cytometry.

Summary and Conclusions

- ATOR-1017 is an agonistic 4-1BB antibody activating cytotoxic CD8+ T cells and NK cells
- The agonistic function of ATOR-1017 is dependent on cross-linking with FcγRI and FcγRIIb
- 4-1BB is co-expressed and co-localized with FcγRI and FcγRII in several tumors
- The immune activation will be directed to tumors co-expressing both specific FcγRs and 4-1BB, which are potential biomarkers for patients and tumor indication selection
- ATOR-1017 is in pre-clinical development with initiation of phase I in 2019