ATOR-1144 is a tumor-directed CTLA-4 x GITR bispecific antibody that acts by depleting Tregs and activating effector T cells and NK cells

Sara Fritzell, Mattias Levin, Anna Dahlman, Ida Åberg, Maria Johansson, Magnus Winnerstam, Karin Enell Smith, Peter Ellmark, Christina Furebring, Per Norlén and Anne Månsson Kvarnhammar

Alligator Bioscience AB, Lund, Sweden

Background

Rationale

- CTLA-4 is a checkpoint receptor highly expressed on tumor-infiltrating T cells, particularly T regulatory cells (Tregs)[1, 2]
- GITR is a TNFR superfamily member highly expressed on tumor-infiltrating T cells, in particular Tregs, but also on NK cells and tumor cells[1, 3-7]
- ATOR-1144 combines targeting of CTLA-4 and GITR to achieve tumor-directed immune activation

About ATOR-1144

- ATOR-1144 is a CTLA-4 x GITR bispecific IgG1 antibody that binds to both targets with high affinity (nM range)
- ATOR-1144 was developed for treatment of solid tumors and hematological malignancies

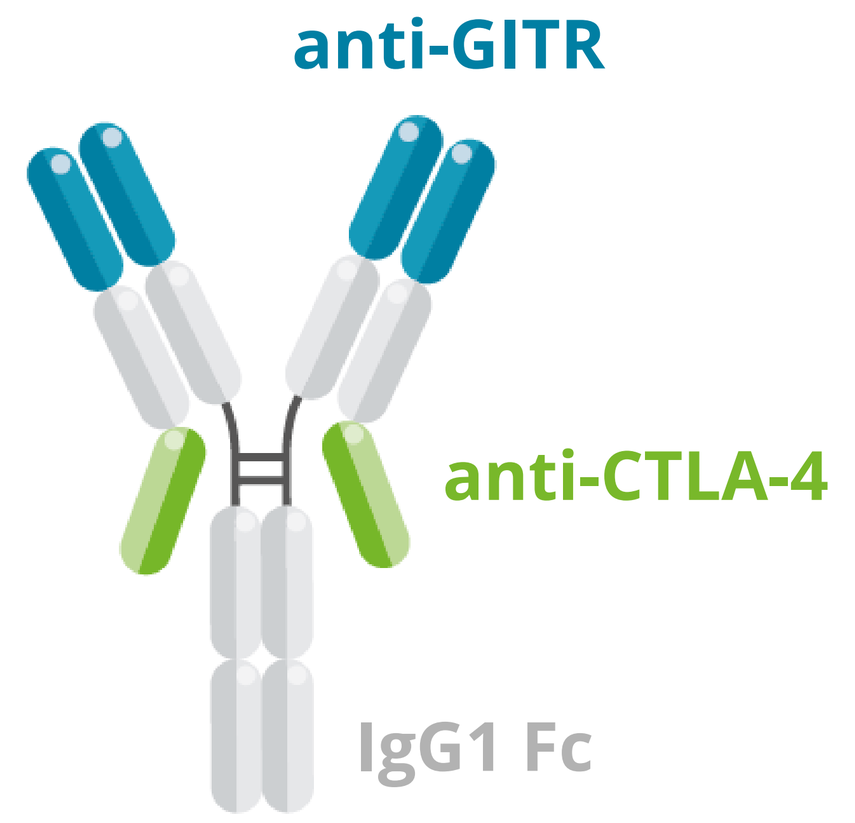
anti-GITR
anti-CTLA-4
IgG1 Fc

References

1. Arce Vargas et al. Cancer Cell. 2018; 33: 1-15. 2. Montler et al. Clin Trans Immunol. 2016; 5: e70. 3. Sukumar et al. Cancer Res. 2018; 77(16): 4378-88. 4. van Beek et al. Int J Cancer. 2019; Feb 4. 5. Argast et al. Proceedings: AACR Annual Meeting 2018 (abstract #3826). 6. Wang et al. Proceedings: AACR Annual Meeting 2017 (abstract #5621). 7. Holland et al. Proceedings: AACR Annual Meeting 2018 (abstract #3813).

GITR is expressed on TILs and tumor cells

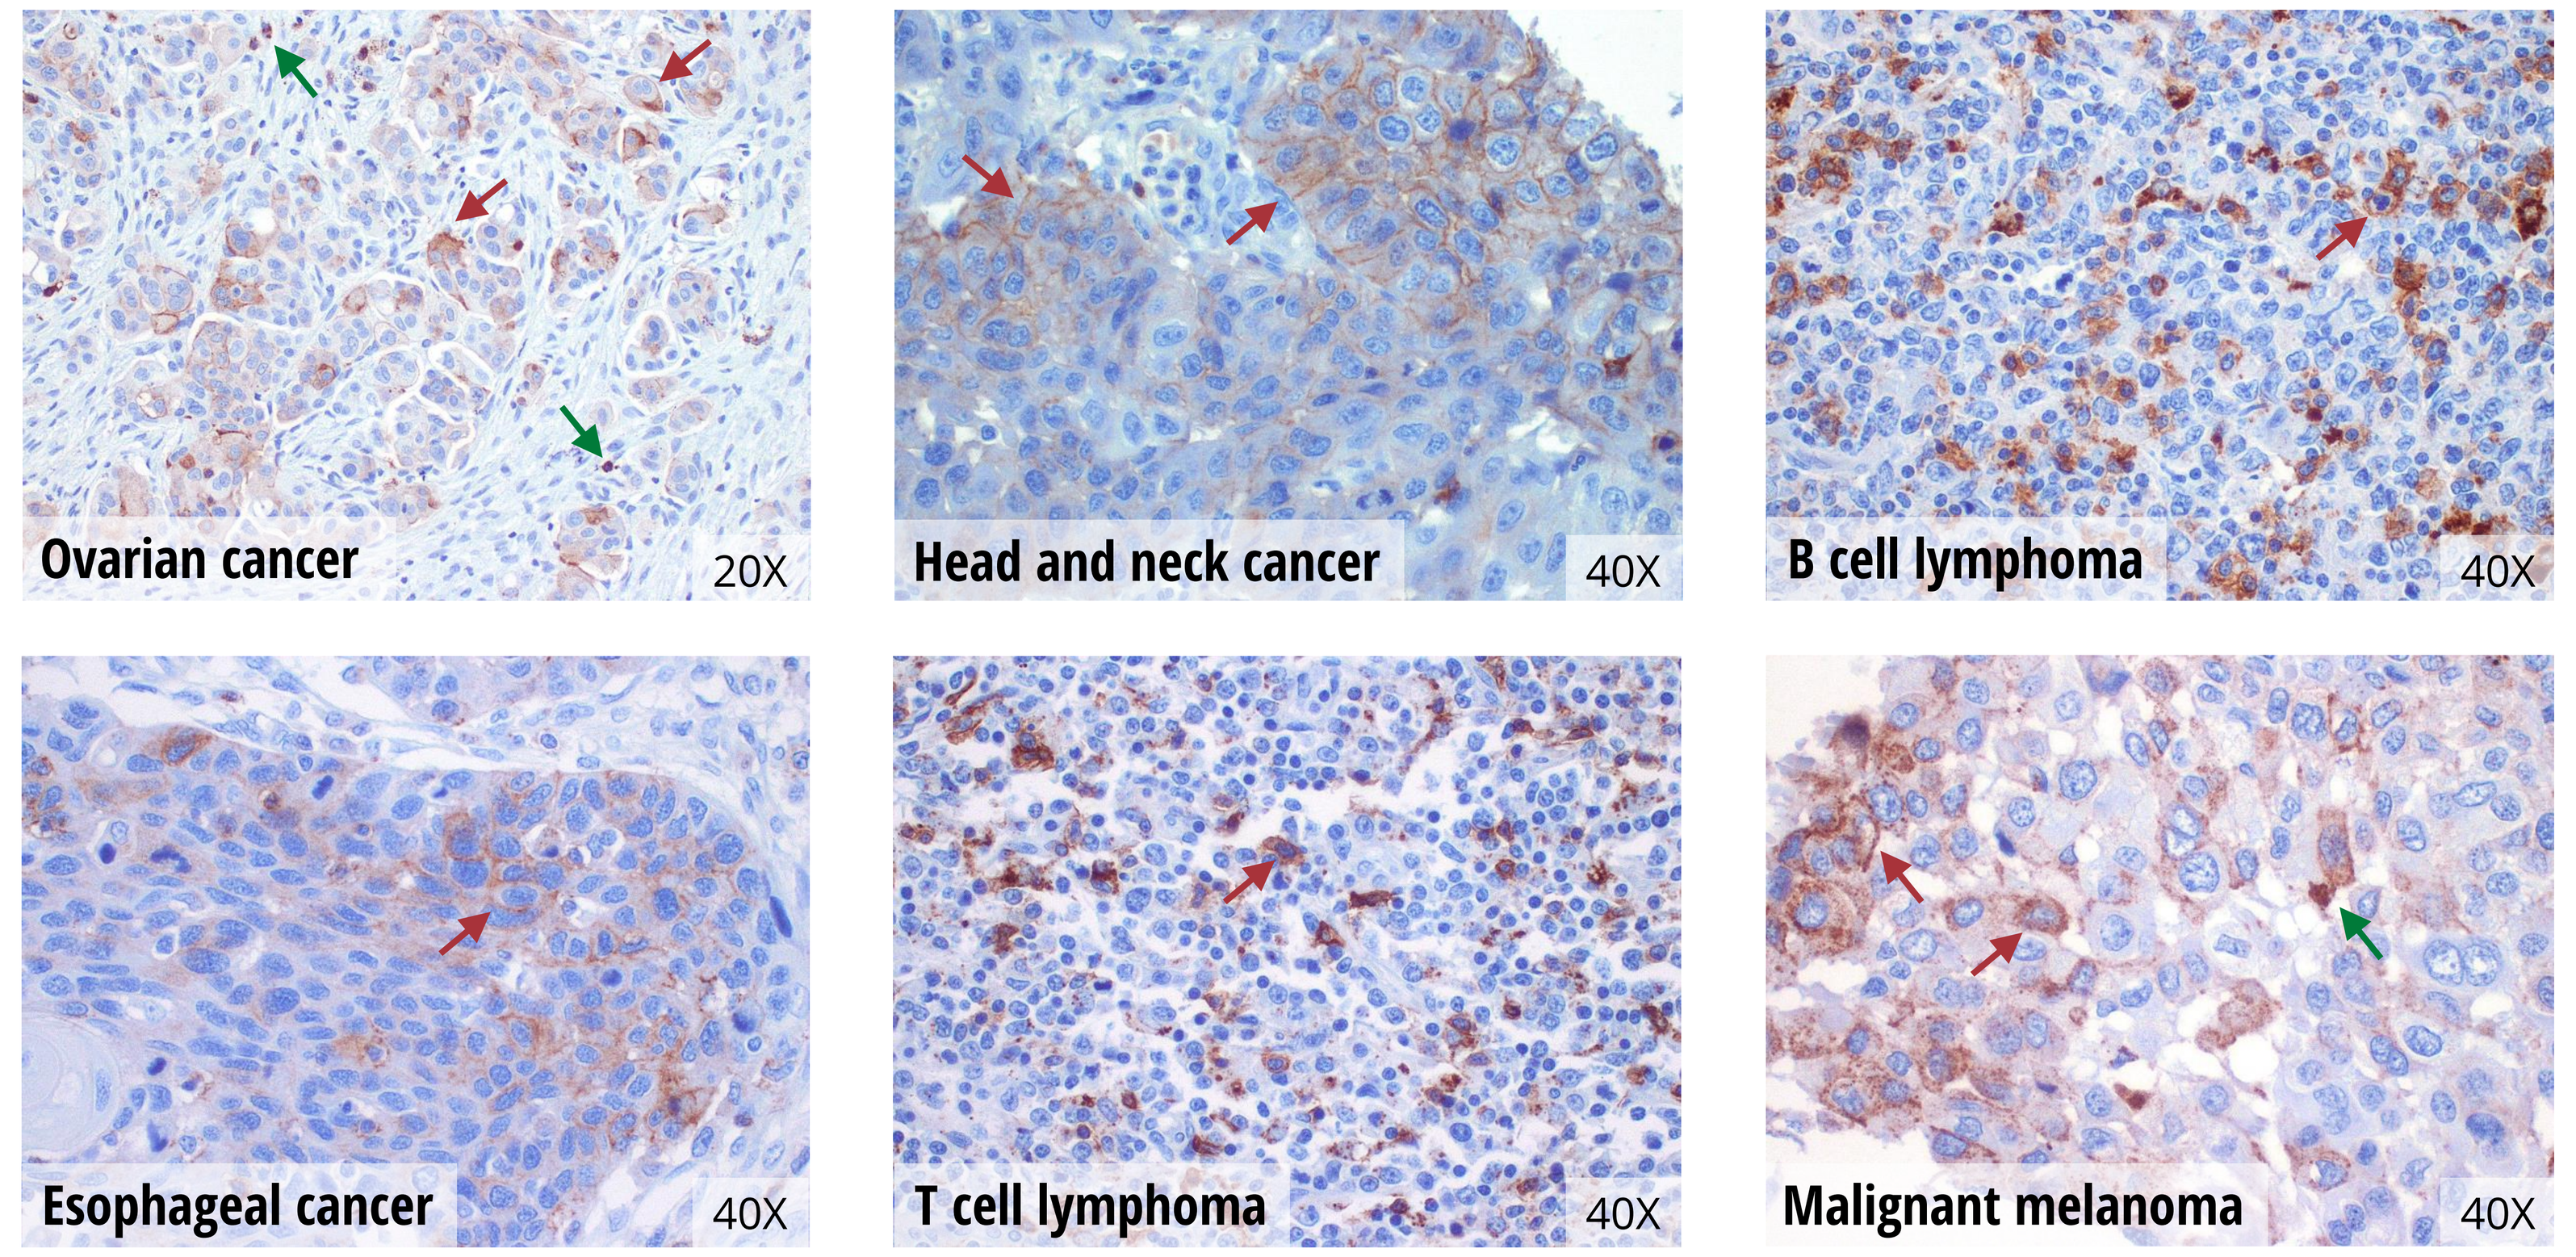
Ovarian cancer
20X
Head and neck cancer
40X
B cell lymphoma
40X
Esophageal cancer
40X
T cell lymphoma
40X
Malignant melanoma
40X

Figure 1. GITR expression (brown) was assessed in formalin-fixed, paraffin embedded tissue microarrays from various cancer indications using a rabbit anti-human GITR antibody (ab237713, Abcam). **Green** arrows indicate staining of infiltrating mononuclear cells. **Red** arrows indicate staining of neoplastic cells.

ATOR-1144 induces formation of cell complexes

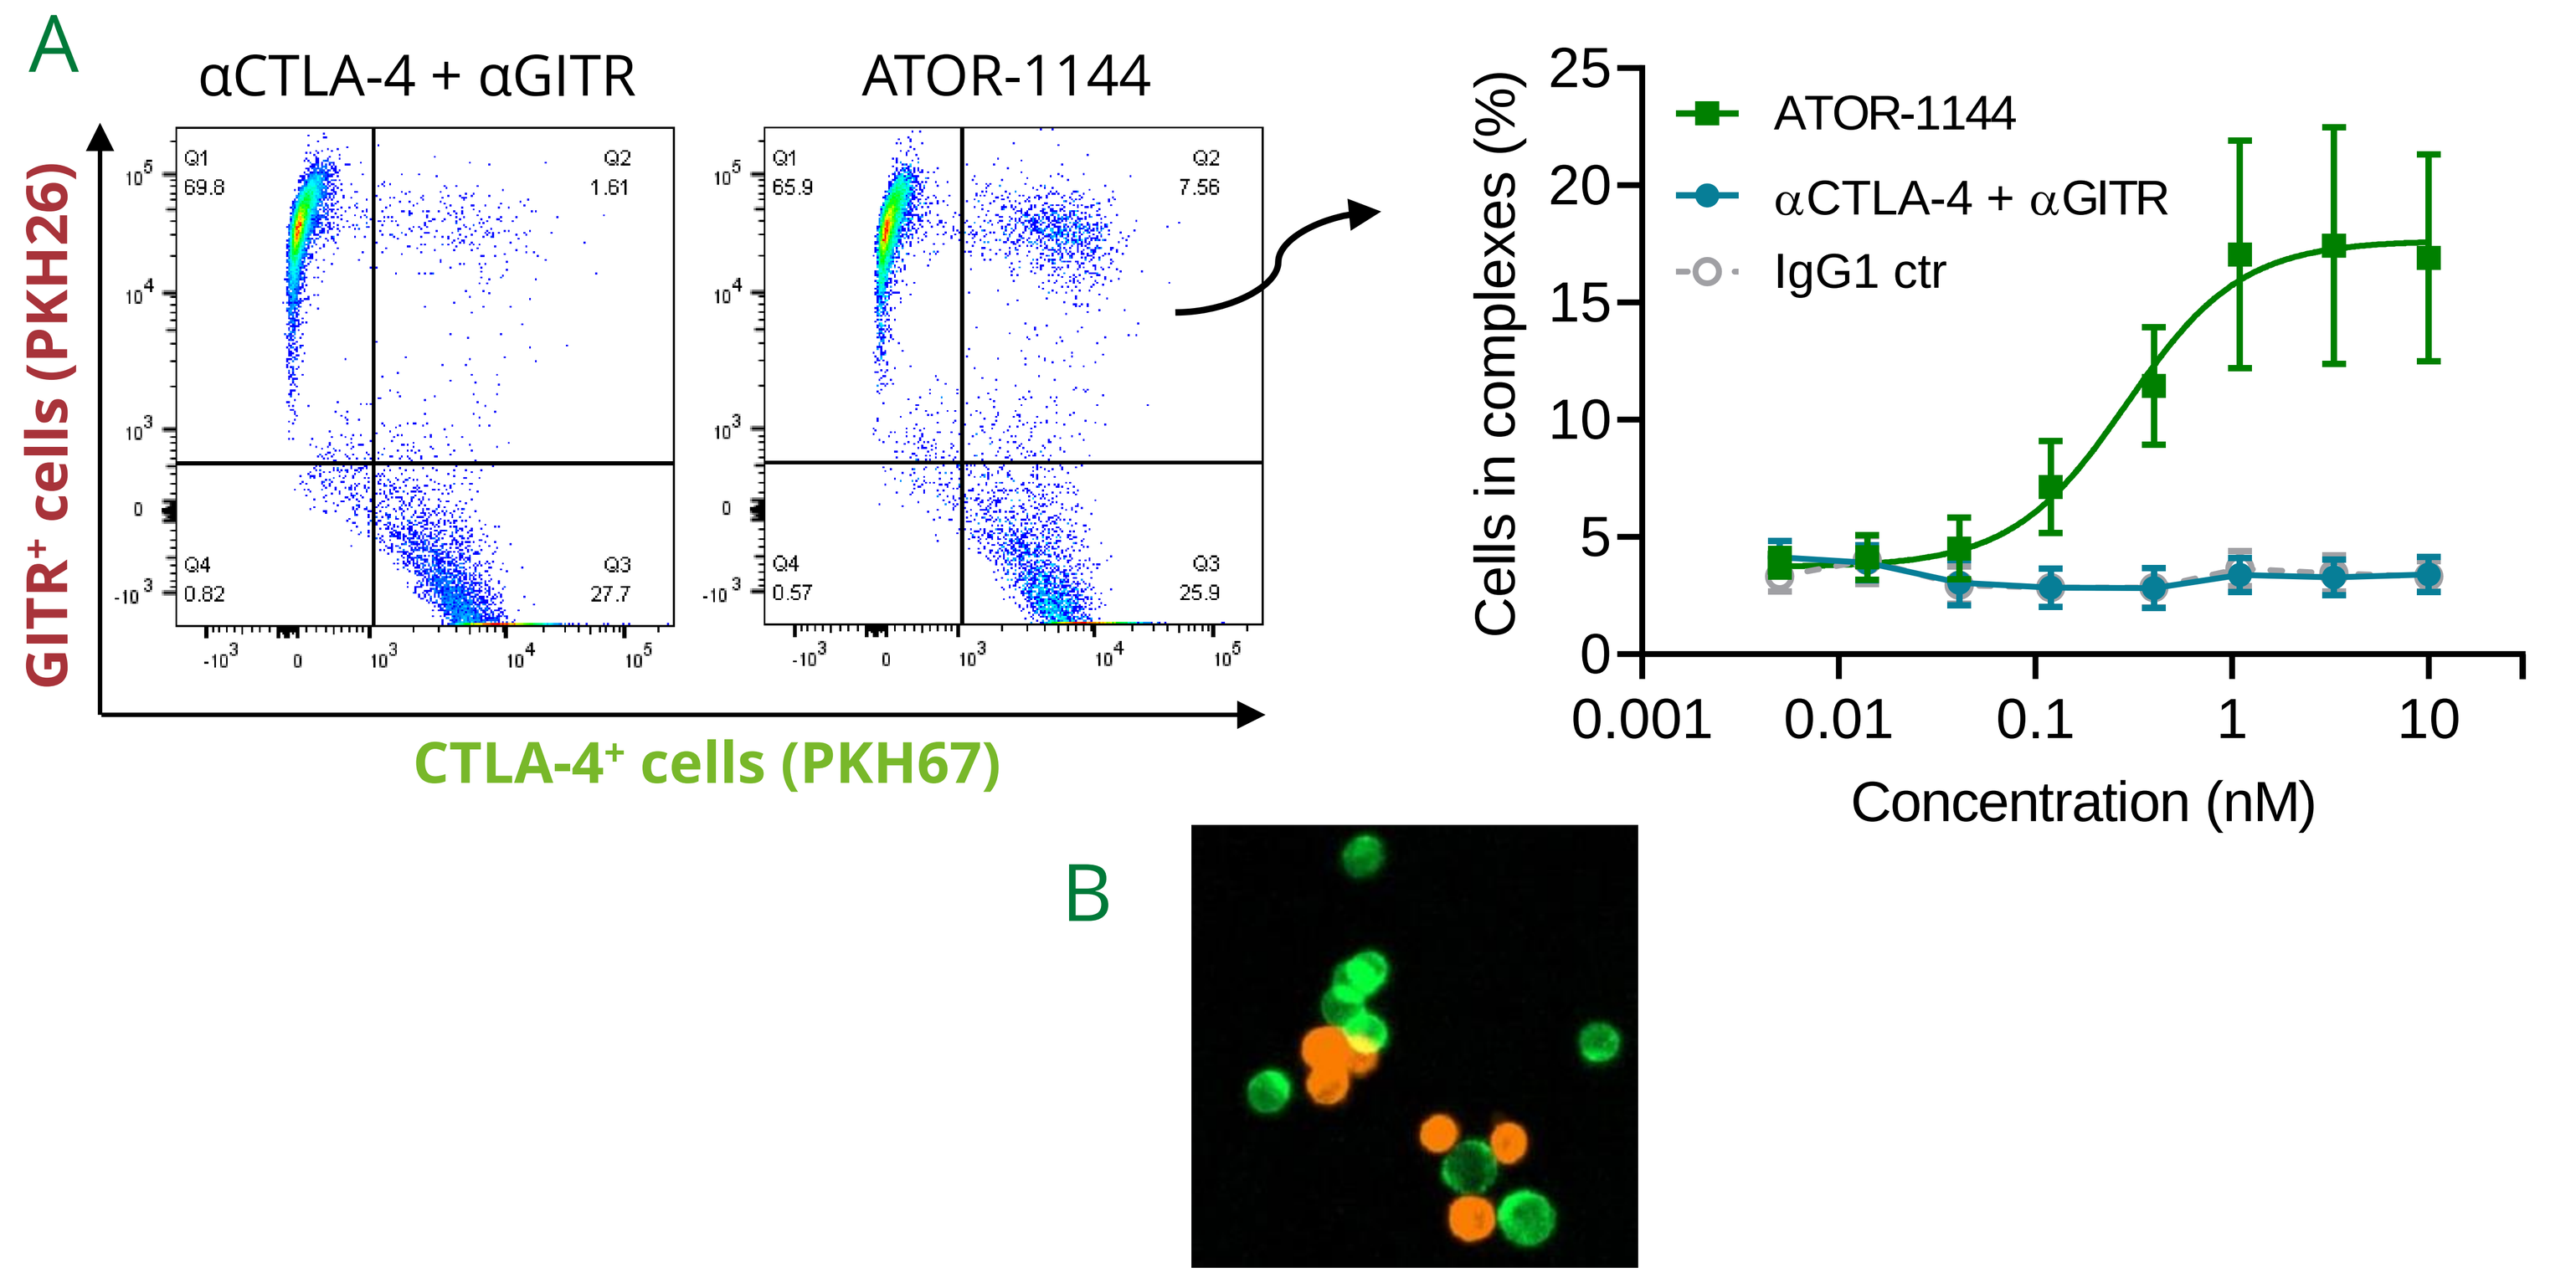
A
αCTLA-4 + αGITR
ATOR-1144
GITR⁺ cells (PKH26)
CTLA-4⁺ cells (PKH67)
Q1 69.8
Q2 1.61
Q4 0.82
Q3 27.7
Q1 65.9
Q2 7.56
Q4 0.57
Q3 25.9
Cells in complexes (%)
ATOR-1144
αCTLA-4 + αGITR
IgG1 ctr
0.001
0.01
0.1
1
10
Concentration (nM)
B

Figure 2. GITR⁺ and CTLA-4⁺ CHO cells were labelled with PKH26 (red dye) and PKH67 (green dye), respectively. Cells were mixed 1:1 and antibodies were added. **(A)** The percentage of cells in complexes was quantified by flow cytometry (n=4). **(B)** The formation of cell complexes was visualized using a Cytation 5 Cell Imaging reader.

ATOR-1144 activates human T cells

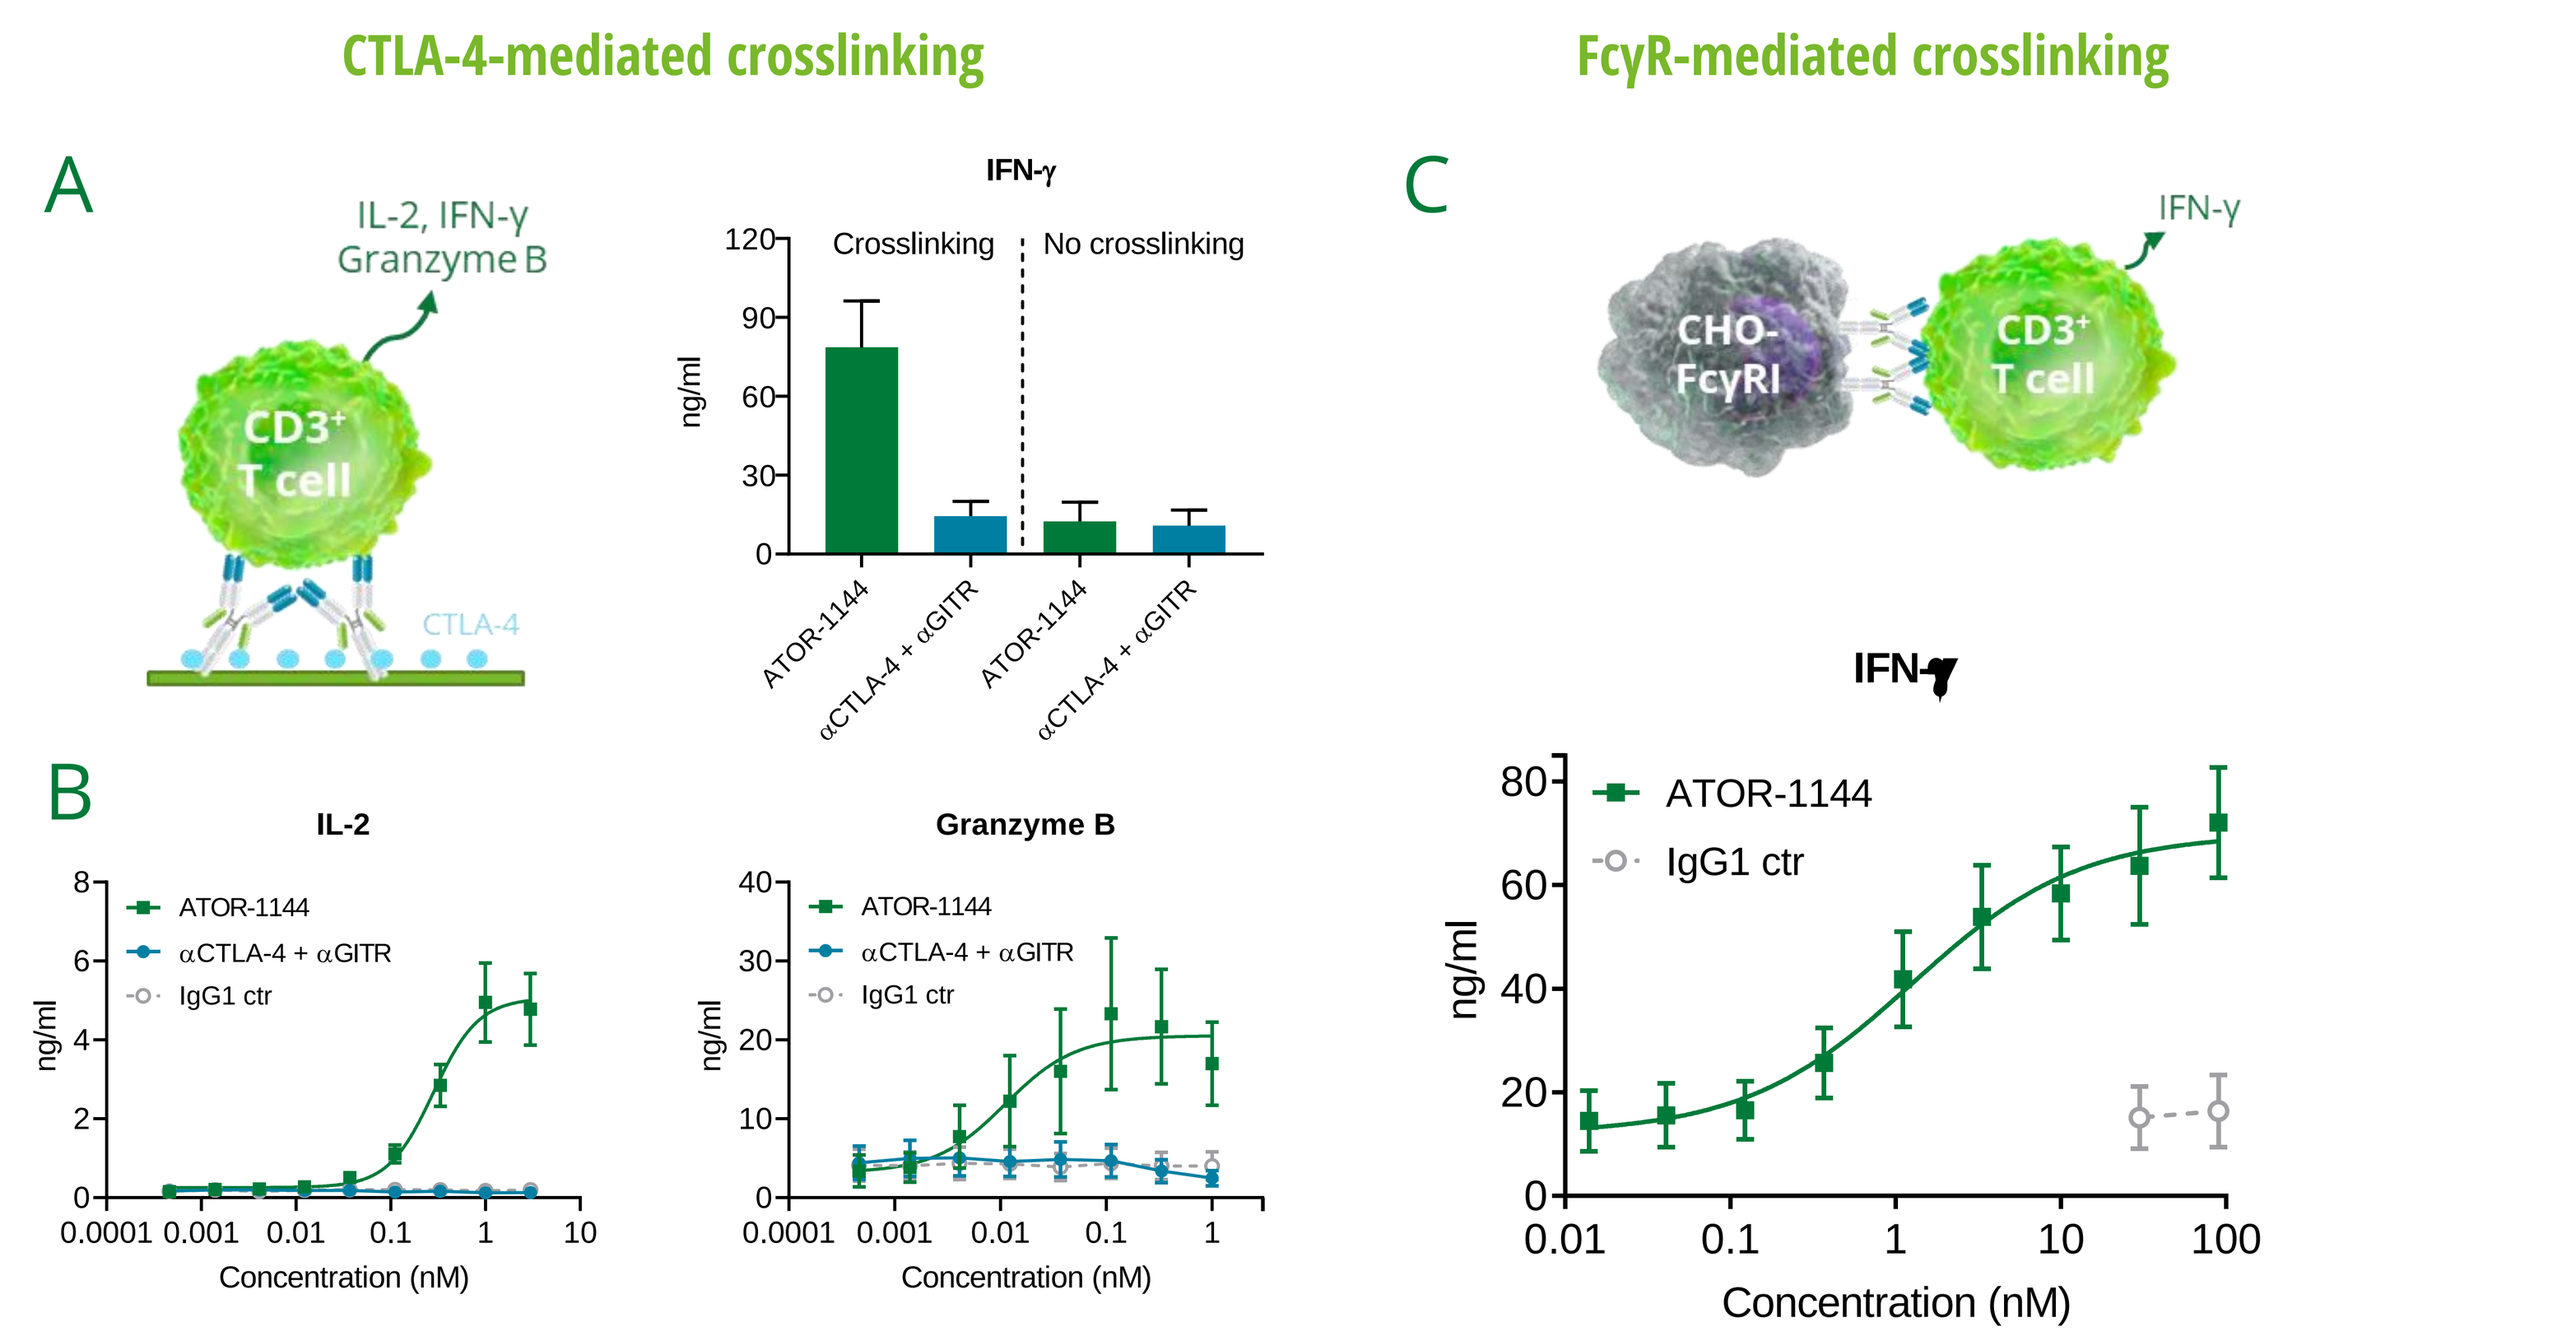
CTLA-4-mediated crosslinking
FcγR-mediated crosslinking
A
IL-2, IFN-γ
Granzyme B
CD3⁺ T cell
CTLA-4
IFN-γ
Crosslinking
No crosslinking
ng/ml
ATOR-1144
αCTLA-4 + αGITR
B
IL-2
Granzyme B
ATOR-1144
αCTLA-4 + αGITR
IgG1 ctr
Concentration (nM)
C
CHO-FcγRI
CD3⁺ T cell
IFN-γ
ATOR-1144
IgG1 ctr
Concentration (nM)

Figure 3. (A) CD3⁺ T cells cultured with or without immobilized CTLA-4, anti-CD3 and antibodies (n=2). **(B)** CD3⁺ T cells cultured with immobilized CTLA-4, anti-CD3 and antibodies (n=13). **(C)** CD3⁺ T cells cultured with irradiated CHO-FcγRI cells, anti-CD3 and antibodies (n=8). IL-2, IFN-γ and granzyme B were measured by ELISA.

ATOR-1144 activates human NK cells and enhances cytotoxicity

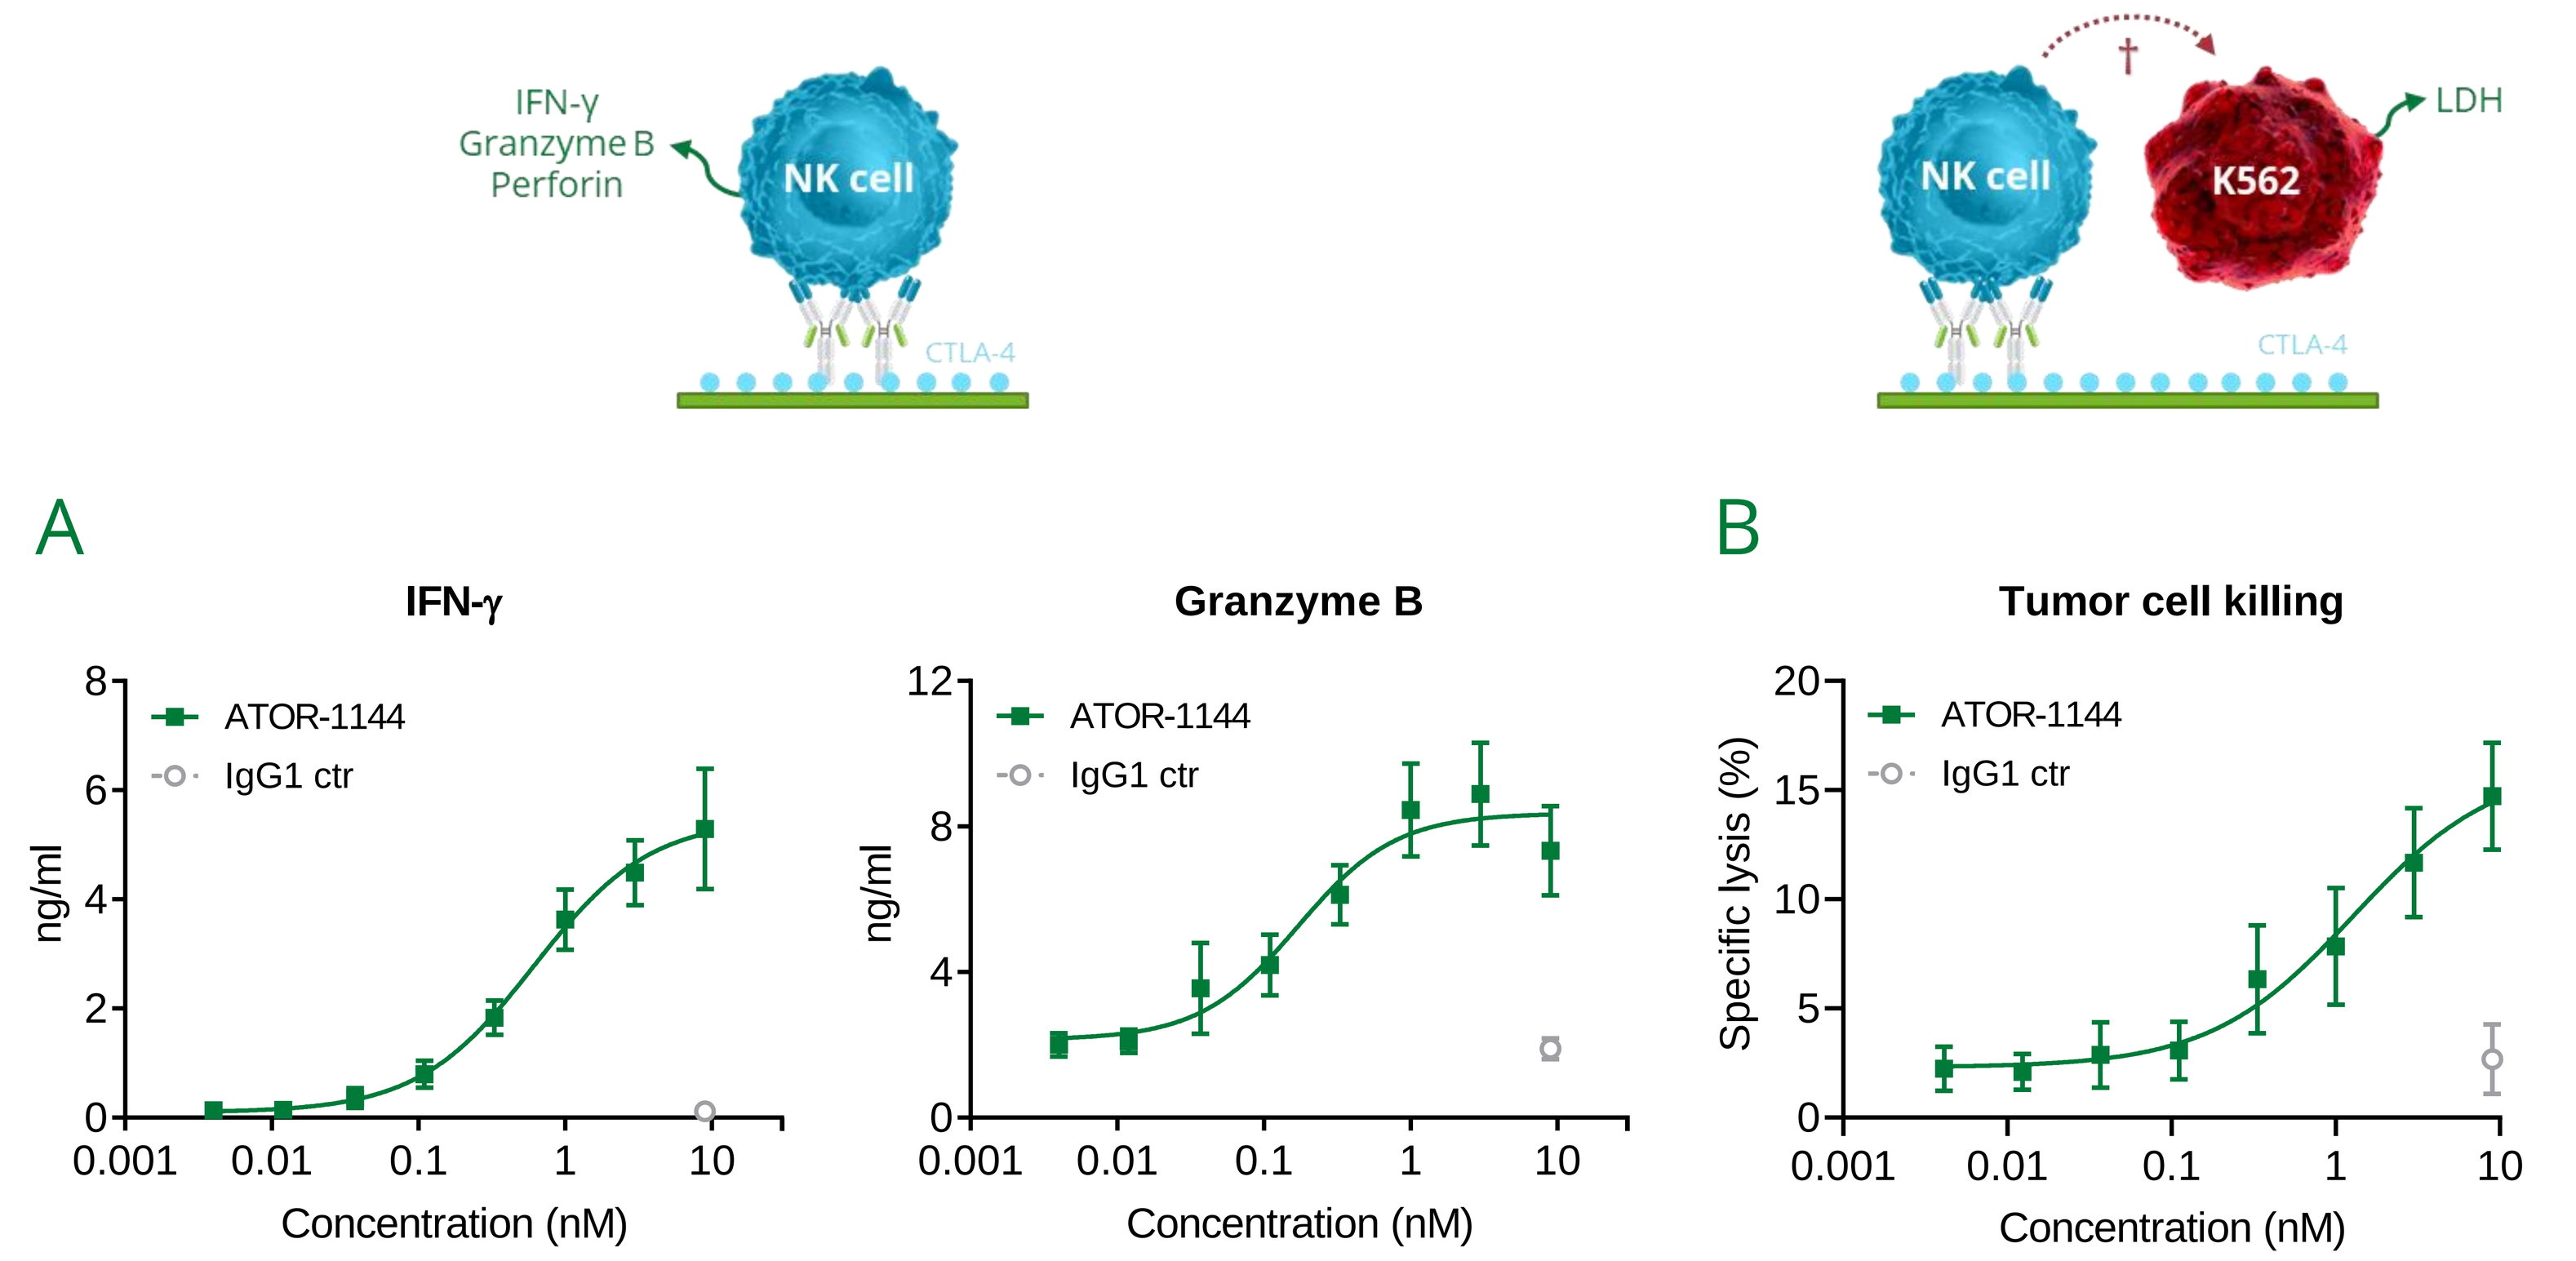
IFN-γ
Granzyme B
Perforin
NK cell
CTLA-4
NK cell
K562
LDH
A
IFN-γ
Granzyme B
ng/ml
ATOR-1144
IgG1 ctr
Concentration (nM)
B
Tumor cell killing
Specific lysis (%)
ATOR-1144
IgG1 ctr
Concentration (nM)

Figure 4. NK cells were cultured in plates with immobilized CTLA-4 in the presence of IL-2 and with or without ATOR-1144 and IgG1 control for 48 h. **(A)** IFN-γ and granzyme B release was measured in the supernatants by ELISA (n=8-10). **(B)** K562 cells were added, and after an additional 4-h culture period, specific lysis after background subtraction was determined as LDH release (n=10).

ATOR-1144 induces ADCC of Tregs

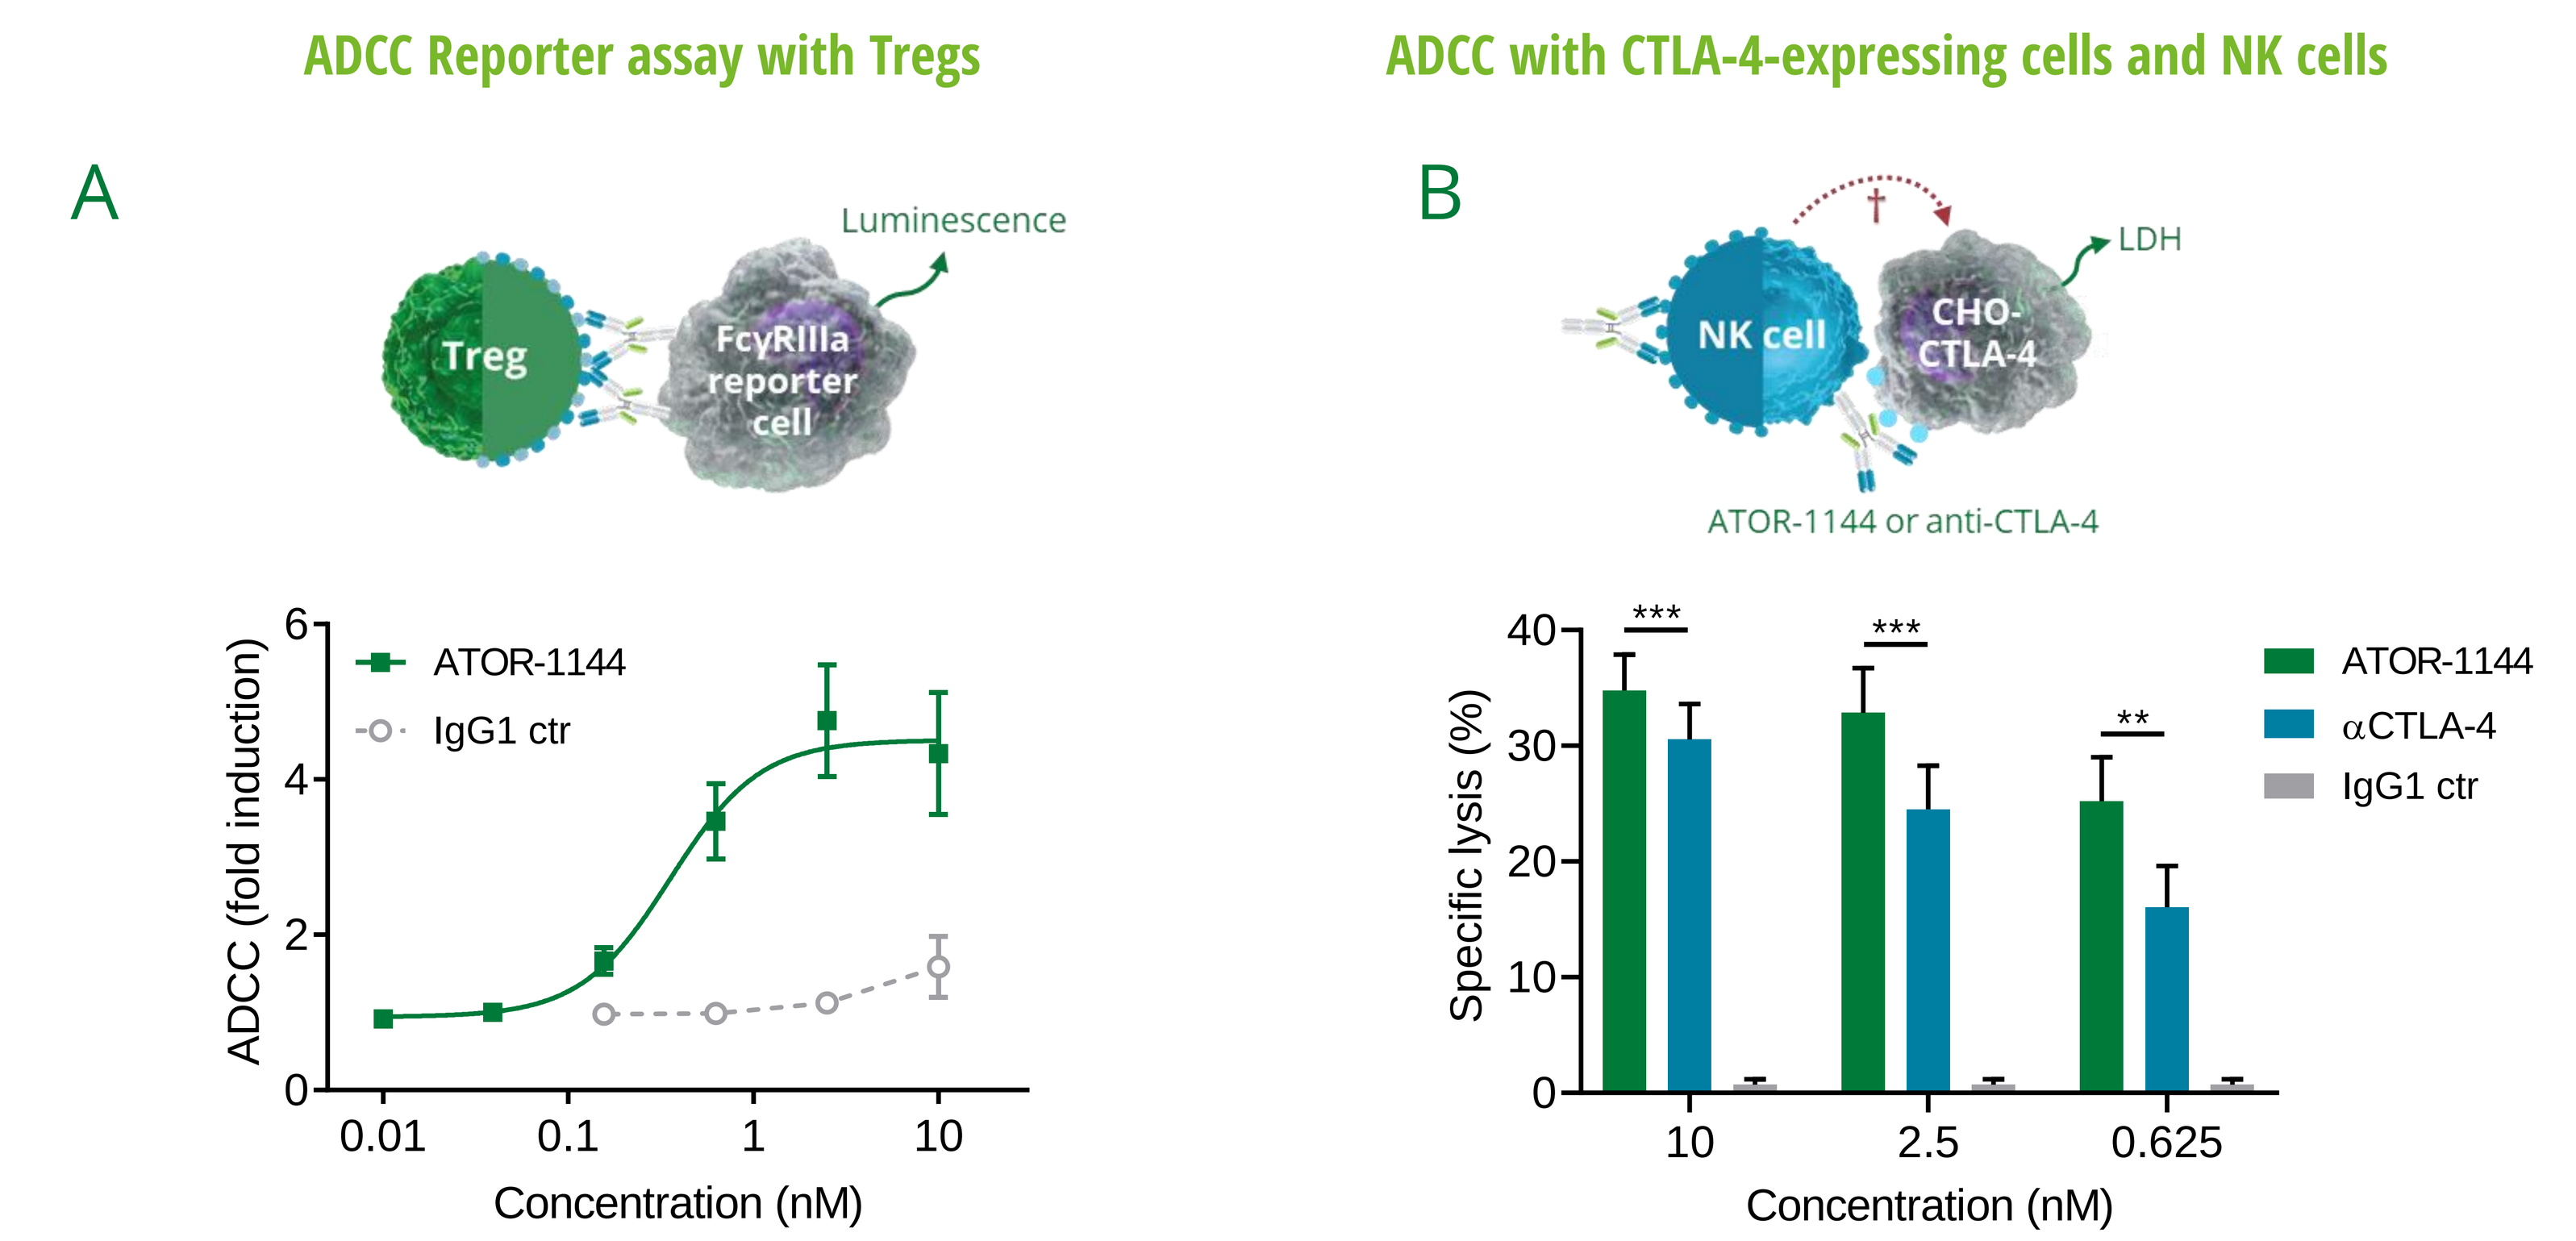
ADCC Reporter assay with Tregs
ADCC with CTLA-4-expressing cells and NK cells
A
Treg
FcγRIIIa reporter cell
Luminescence
ADCC (fold induction)
ATOR-1144
IgG1 ctr
Concentration (nM)
B
NK cell
CHO-CTLA-4
LDH
ATOR-1144 or anti-CTLA-4
Specific lysis (%)
ATOR-1144
αCTLA-4
IgG1 ctr
10
2.5
0.625
Concentration (nM)

Figure 5. (A) Human in vitro activated Tregs were incubated with antibodies for 6 h. Treg depletion was measured in an ADCC (FcγRIIIa) reporter assay (n=5). **(B)** CTLA-4⁺ CHO cells were cultured together with NK cells at a 1:10 ratio in the presence of ATOR-1144, anti-CTLA-4 antibody or isotype control. Specific lysis was determined as LDH release (n=8).

ATOR-1144 induces ADCC of GITR⁺ tumor cells

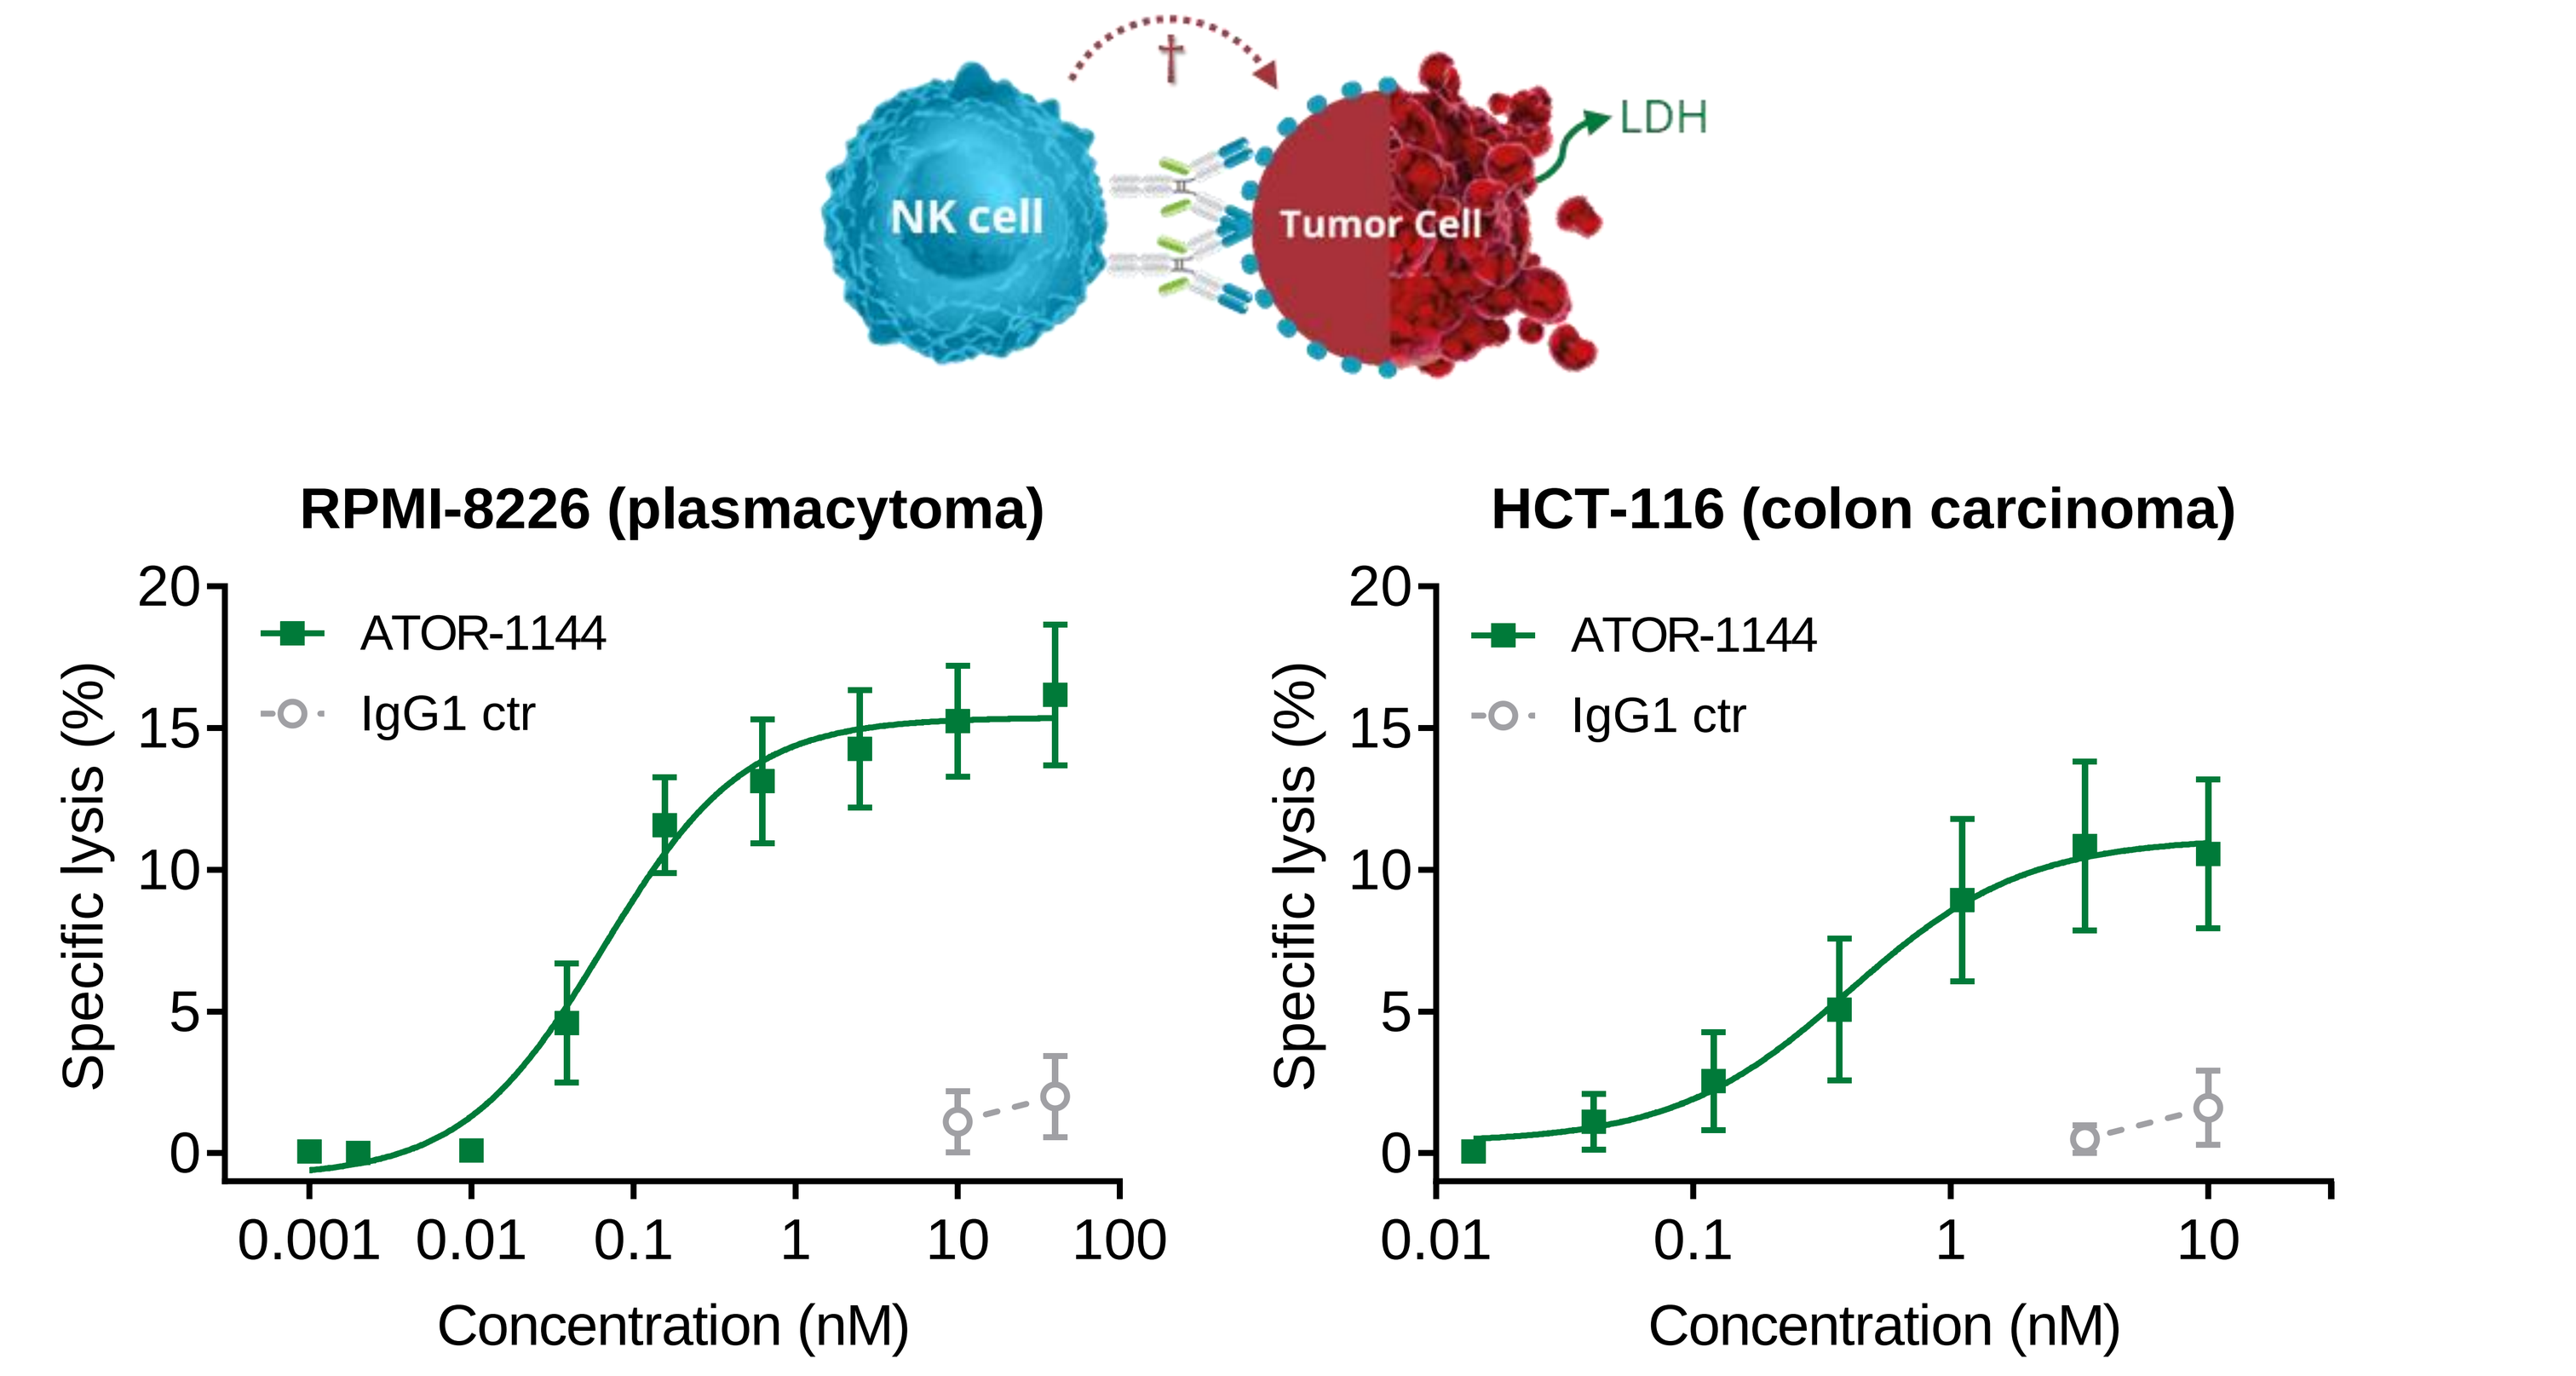
NK cell
Tumor Cell
LDH
RPMI-8226 (plasmacytoma)
HCT-116 (colon carcinoma)
Specific lysis (%)
ATOR-1144
IgG1 ctr
Concentration (nM)

Figure 6. Tumor cells were cultured overnight with NK cells at a 1:10 ratio in the presence of ATOR-1144 or isotype control. Specific lysis was determined as LDH release (n=4-6).

Summary and Conclusions

Summary

- ATOR-1144 acts through several mechanisms:
 - Activation of effector T cells
 - Depletion of Tregs and tumor cells
 - Activation of NK cells for enhanced tumor cell killing

Conclusions

- ATOR-1144 is a next generation CTLA-4 targeting antibody with enhanced Treg depletion and direct anti-tumor activity
- Dual targeting of CTLA-4 and GITR is expected to direct the effect to the tumor area

Contact information

Contact the author for more information: amk@alligatorbioscience.com

AACR Annual Meeting 2019, Poster # 4077
Alligator Bioscience AB, Lund, Sweden

ALLIGATOR bioscience