Mitazalimab (CD40 agonist) in combination with mFOLFIRINOX in patients with metastatic pancreatic ductal adenocarcinoma: Safety data and recommended dose for phase 2 from OPTIMIZE-1, a phase 1b/2 study

Jean-Luc van Laethem[1], Ivan Borbath[2], Hans Prenen[3], Aurélien Lambert[4], Karen Geboes[5], Jean-Frédéric Blanc[6], Yago Pico de Coaña[7], Karin Enell Smith[7], Lena Schultz[7], Karin Nordbladh[7], Peter Ellmark[7], Sumeet Ambarkhane[7], Malin Carlsson[7], Philippe Cassier[8]

1. Hôspital Erasme, Brussels, Belgium; 2. Cliniques Universitaires St-Luc, Brussels, Belgium; 3. UUniversitair Ziekenhuis Antwerpen, Edegem, Belgium; 4. Institut de Cancérologie de Lorraine, Nancy, France; 5. Universitair Ziekenhuis Gent , Ghent, Belgium; 6. Centre Hospitalier Universitaire (CHU) de Bordeaux - Hôpital Haut-Lévêque, Bordeaux, France; Alligator Bioscience AB, Lund, Sweden; 8. Centre Léon Bérard Lyon, France

Mitazalimab, a CD40 agonist with best-in-class profile

- Mitazalimab is a **FcγR crosslinking dependent CD40 agonistic** antibody (IgG1) with a tumor-directed immune activation
- Mitazalimab binds with **high affinity** to a unique binding epitope on the CD40 receptor allowing for high efficacy and potency
- Mitazalimab has the **potential for superior clinical activity**, since mitazalimab is the only FcγR-dependent CD40 agonist with tumor-directed activity that can be dosed >1 mg/kg

Mitazalimab, a human CD40 agonistic antibody developed for cancer immunotherapy

- Targeting CD40 kickstarts the cancer-immunity cycle, priming and activating tumor-specific T cells[1,2].
- Pancreatic ductal adenocarcinoma (PDAC) is characterized by a desmoplastic tumor stroma that creates a mechanical barrier that limits exposure to chemotherapy, hosts immune-suppressive macrophages and leads to poor immune infiltration.
- CD40 agonists promote degradation of the tumor stroma by myeloid cells, improving the influx of T cells and chemotherapeutic agents into the tumor[1].

Mode of action

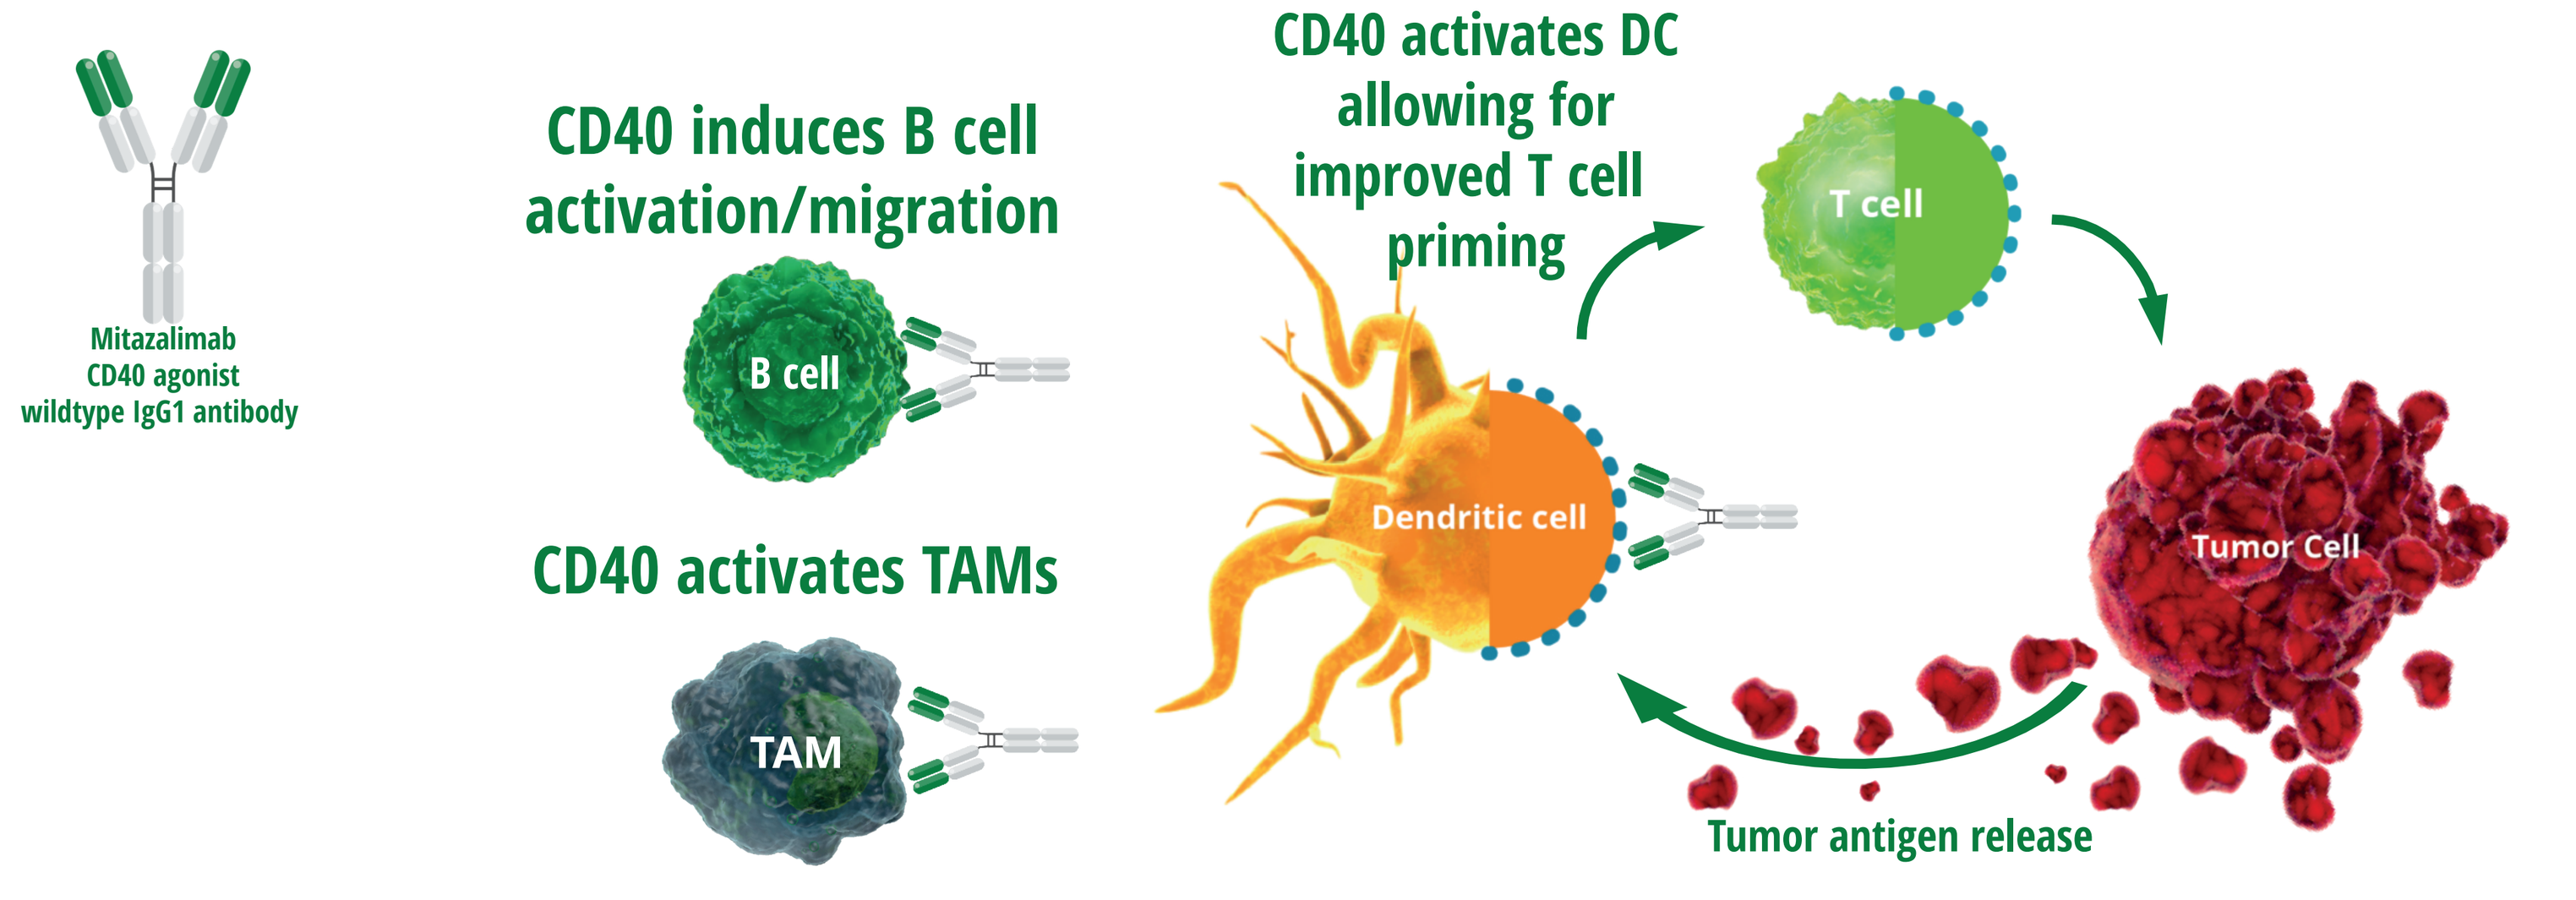
Mitazalimab
CD40 agonist
wildtype IgG1 antibody
CD40 induces B cell activation/migration
B cell
CD40 activates TAMs
TAM
CD40 activates DC allowing for improved T cell priming
Dendritic cell
T cell
Tumor Cell
Tumor antigen release

OPTIMIZE-1 study overview

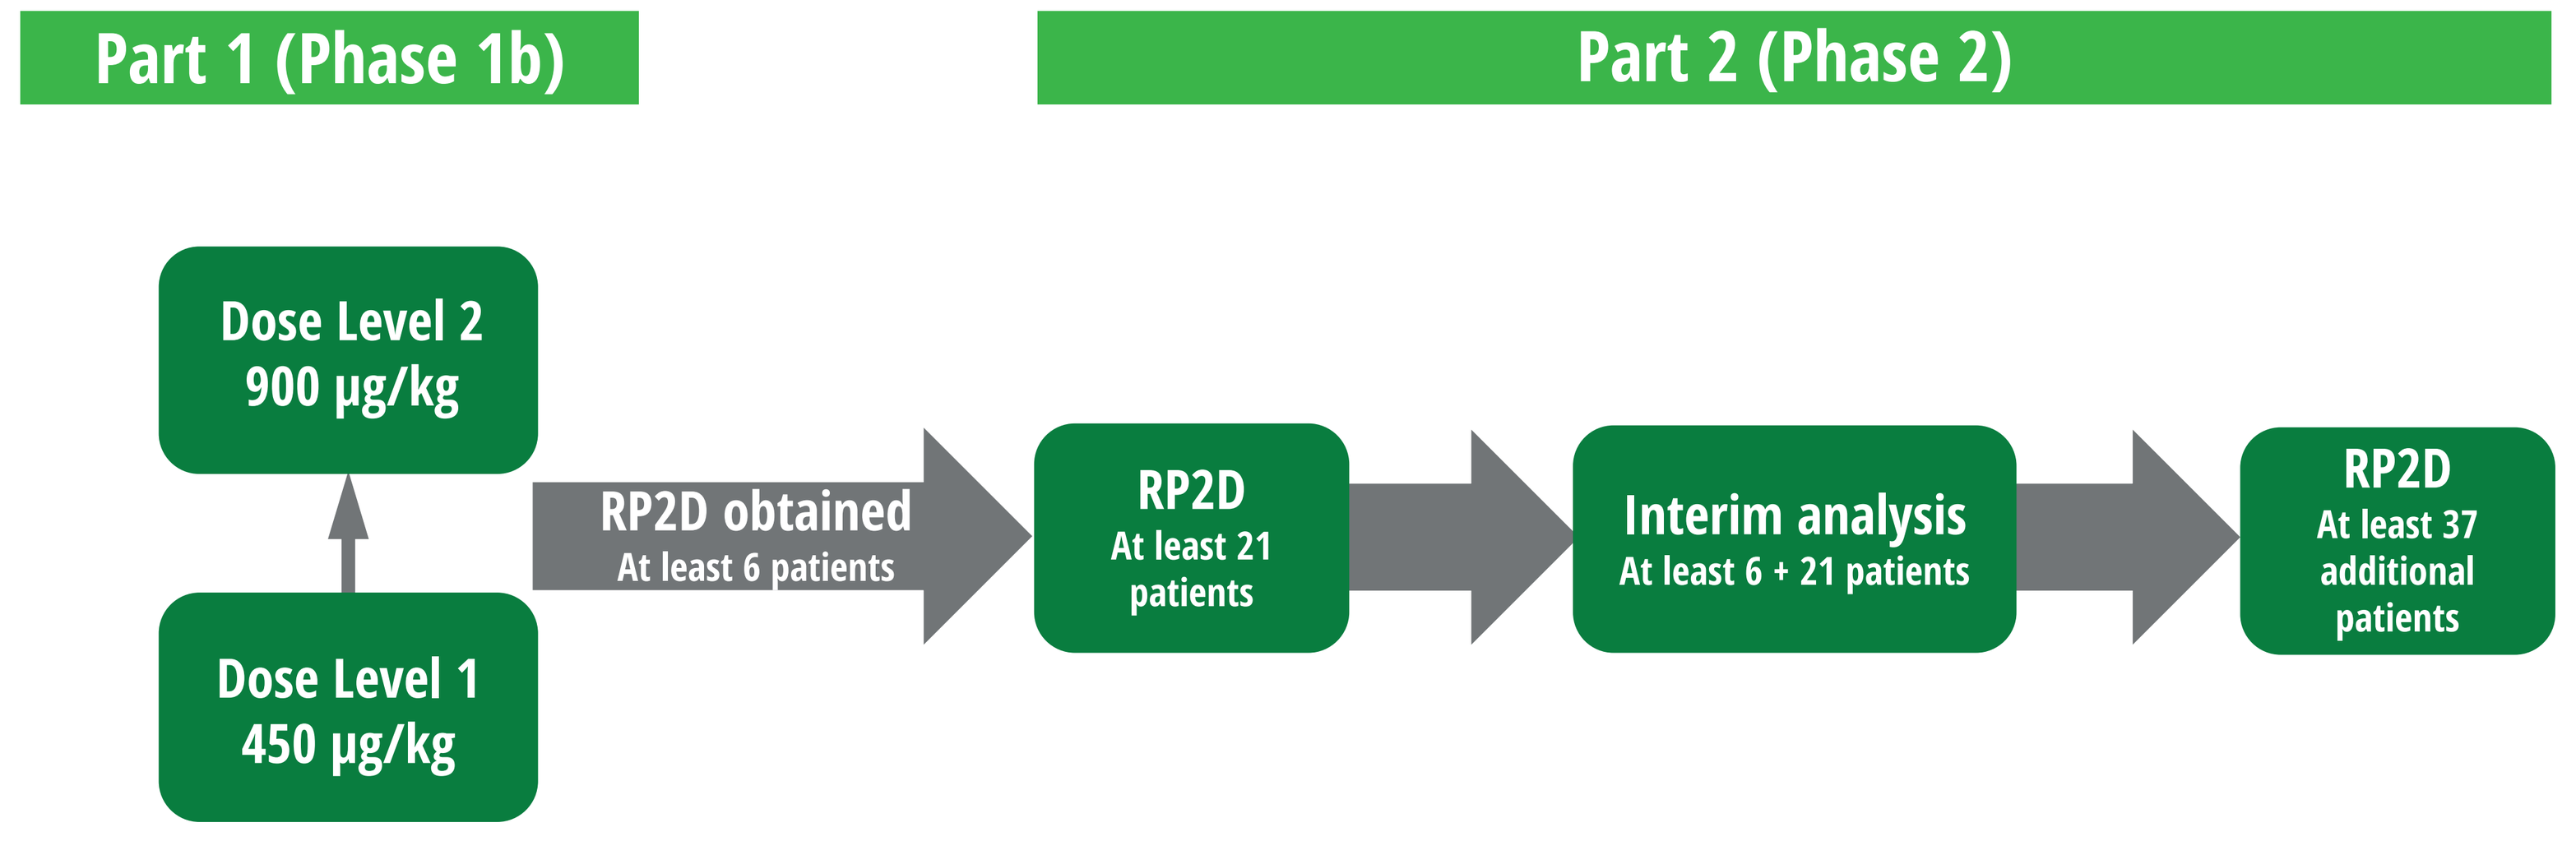
Part 1 (Phase 1b)
Part 2 (Phase 2)
Dose Level 2
900 µg/kg
Dose Level 1
450 µg/kg
RP2D obtained
At least 6 patients
RP2D
At least 21 patients
Interim analysis
At least 6 + 21 patients
RP2D
At least 37 additional patients

RP2D (recommended phase 2 dose)

OPTIMIZE-1 study design

OPTIMIZE-1 (NCT04888312) is a phase 1b/2, open-label, multicenter study designed to evaluate safety, tolerability, and efficacy of mitazalimab in combination with chemotherapy (mFOLFIRINOX) in adults diagnosed with previously untreated metastatic PDAC.

Mitazalimab and mFOLFIRINOX are administered by intravenous infusions. The first cycle lasts 21 days: mitazalimab is administered on Day 1 and 10 and mFOLFIRINOX infusion starts on Day 8. The second and subsequent cycles follow a 14-day cycle schedule where mitazalimab is administered 2 days after mFOLFIRINOX.

Dosing regimen

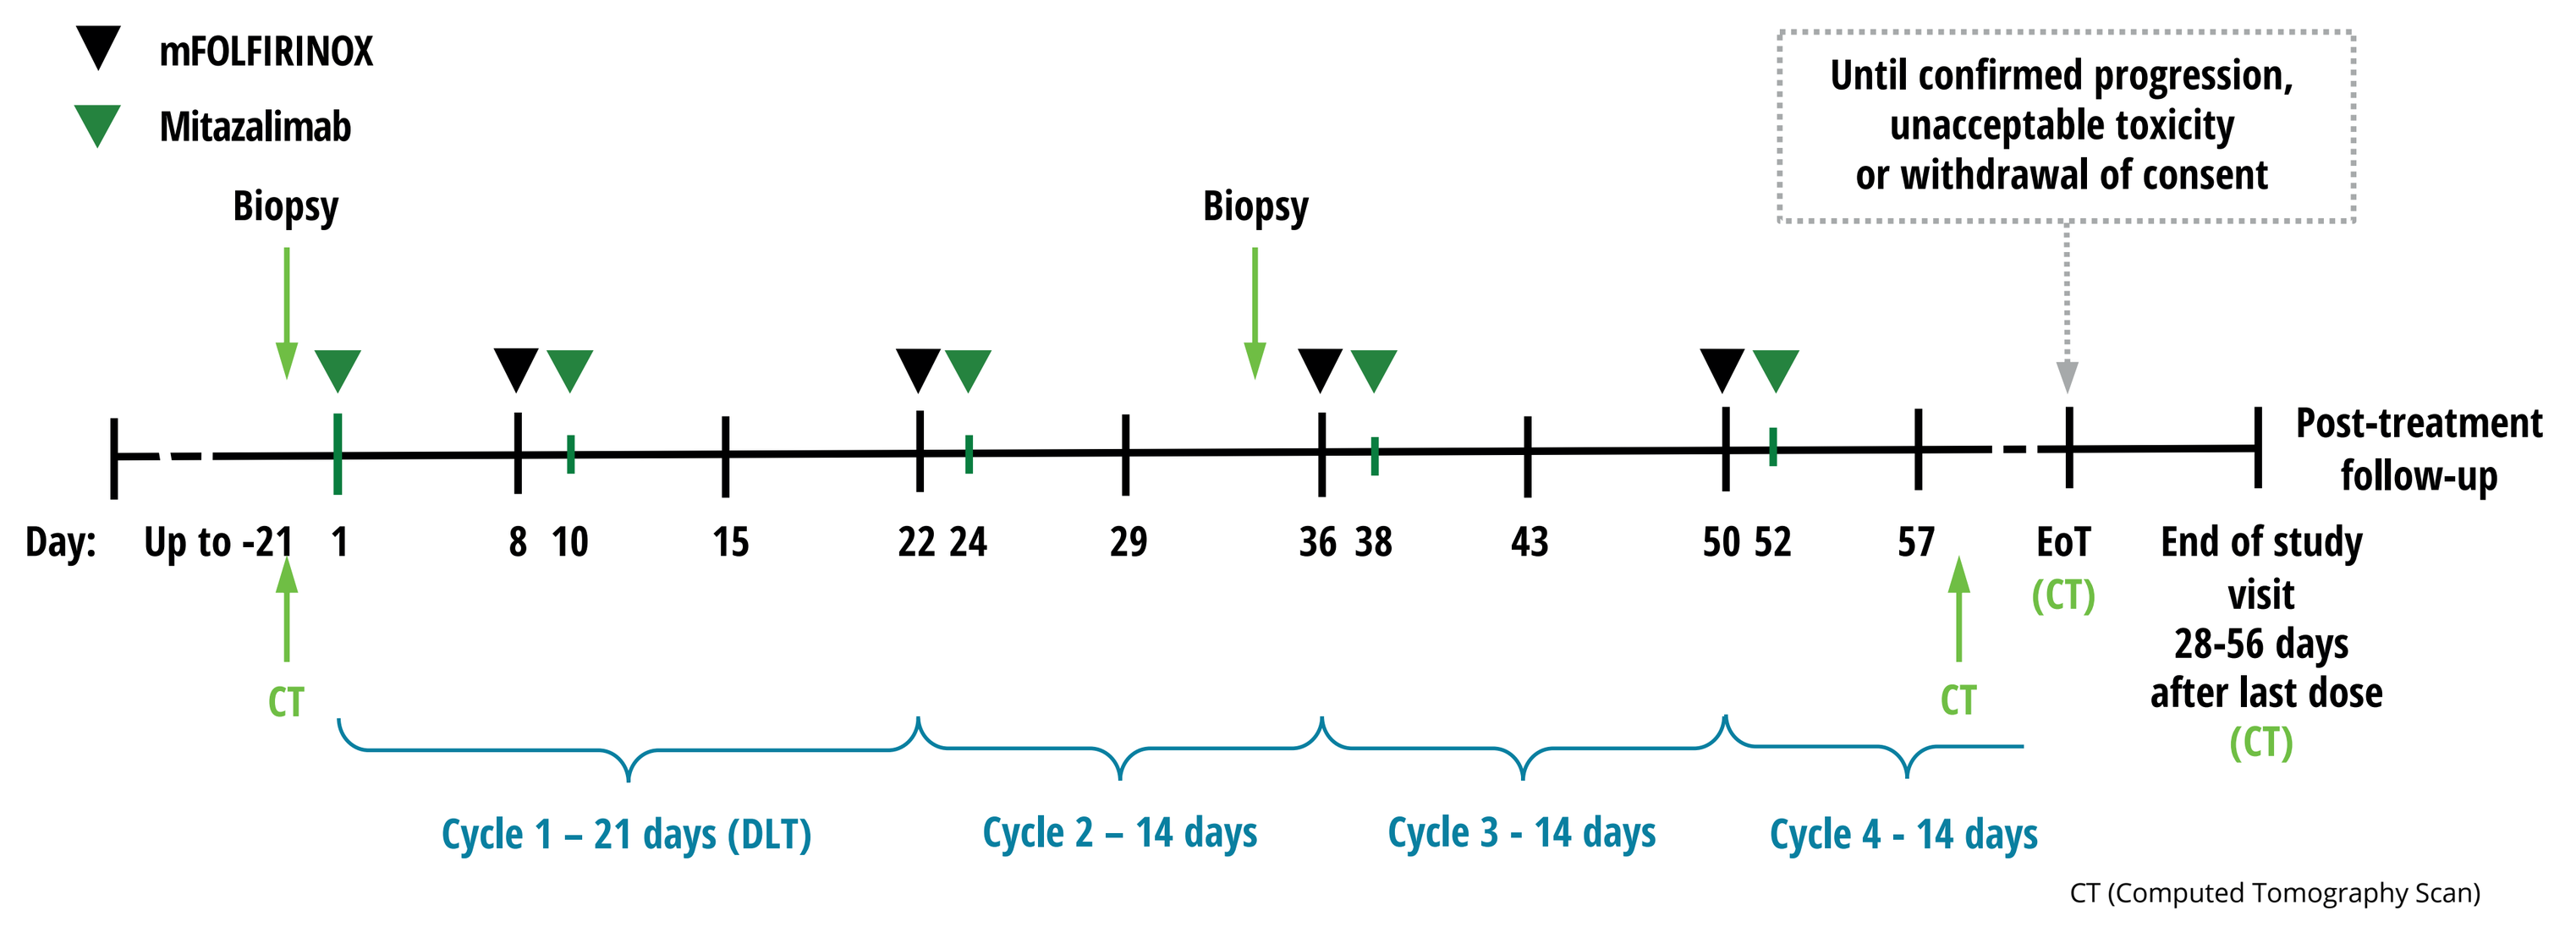
mFOLFIRINOX
Mitazalimab
Biopsy
Biopsy
Until confirmed progression, unacceptable toxicity or withdrawal of consent
Post-treatment follow-up
Day: Up to -21 1 8 10 15 22 24 29 36 38 43 50 52 57
EoT (CT)
End of study visit 28-56 days after last dose (CT)
CT
CT
Cycle 1 – 21 days (DLT)
Cycle 2 – 14 days
Cycle 3 - 14 days
Cycle 4 - 14 days
CT (Computed Tomography Scan)

Study Objectives

Phase 1b

Primary objective: To determine the recommended Phase 2 dose (RP2D).
Secondary objectives include: Assesment of clinical activity (overall response rate (ORR)), disease control rate and time to next anti-cancer therapy) and survival outcome.

Phase 2

Primary objective: To assess the clinical activity of mitazalimab in combination with chemotherapy as determined by ORR.

Secondary objectives include: survival outcomes, Best Overall Response (BOR), Duration of response (DoR), Duration of SD, Disease control rate, and time to next anti-cancer therapy.

Key Eligibility Criteria

Inclusion:
- Diagnosis of metastatic PDAC
- ECOG status of 0 or 1
- No previous chemotherapy for PDAC
- No prior abdominal radiotherapy
- Life expectancy ≥ 3 months
- Acceptable hematologic and clinical laboratory chemistry values

Exclusion:
- Other types of non-ductal tumor of the pancreas
- Known CNS metastases or carcinomatous meningitis
- Has other current cancer or history of cancer in the prior 3 years
- Receipt of attenuated vaccine within 28 days before the first dose of study treatment

Results from part 1 (Dose Escalation)

- Part 1 recruited 11 patients in two dose cohorts: 450 and 900 µg/kg
- Five patients in each cohort received at least one complete treatment cycle
- Data cutoff for RP2D determination was March 9 2022

Patient Demographics

	450 µg/kg	900 µg/kg	Overall
Number of patients	5	6	11
Median (range) age, years	65 (60-66)	60 (57-70)	62 (57-70)
Age Group, n (%)			
18 – 64	2(40)	4(67)	7(63.6)
65 +	3(60)	2(33)	4(36.4)
ECOG Performance status, n (%)			
1	3(60)	3(50)	6(54.5) 5(45.5)
0	2(40)	3(50)	
Median time since diagnosis, days	21	25	25

Time on mitazalimab treatment

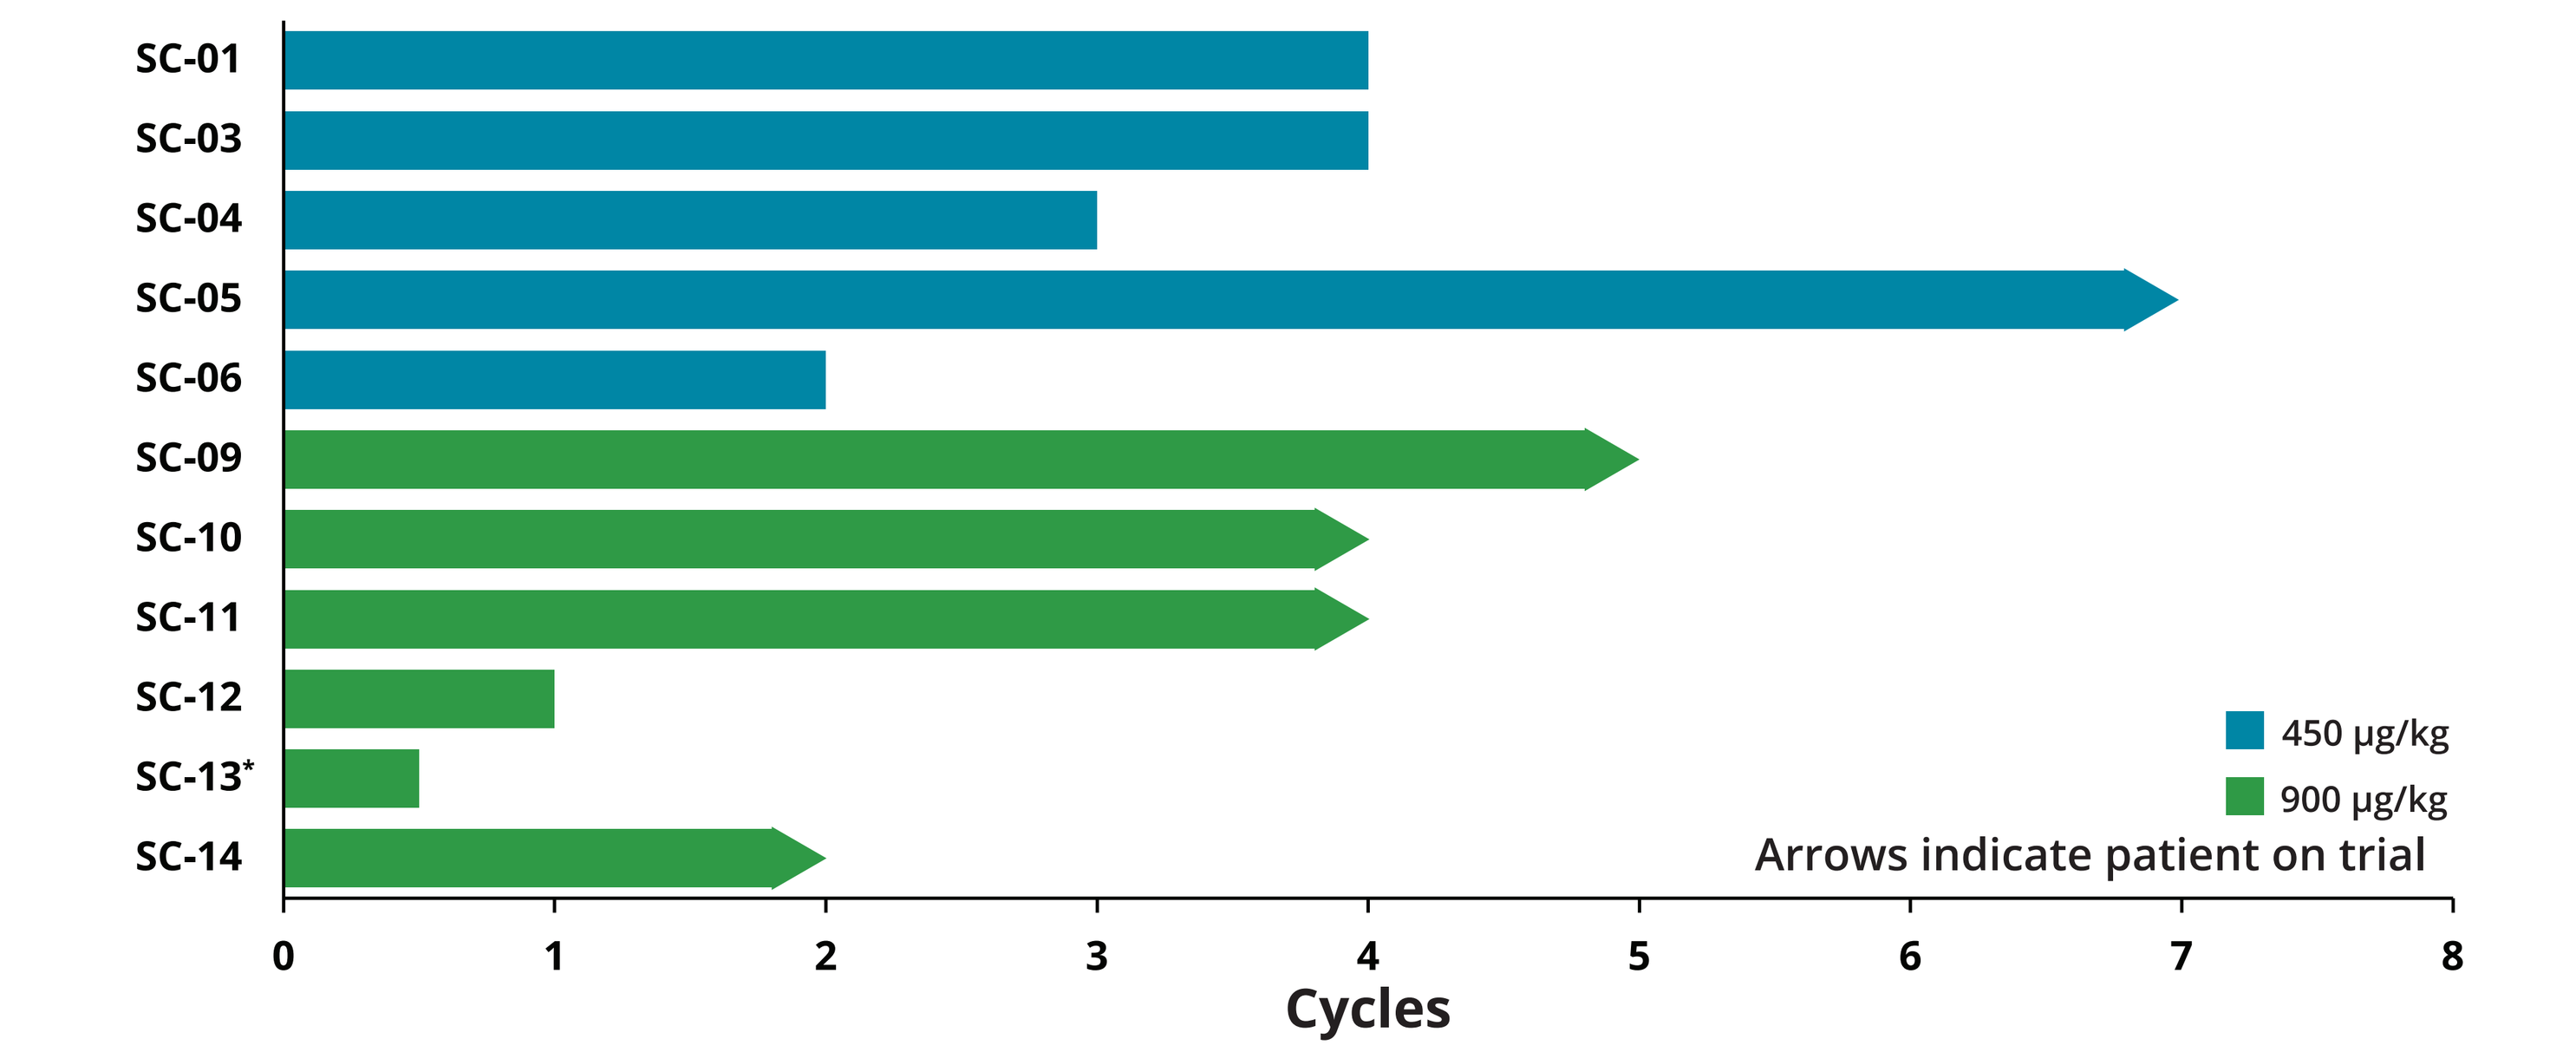
SC-01
SC-03
SC-04
SC-05
SC-06
SC-09
SC-10
SC-11
SC-12
SC-13*
SC-14
0 1 2 3 4 5 6 7 8
Cycles
450 µg/kg
900 µg/kg
Arrows indicate patient on trial

*Patient SC-13 withdrew from the trial for administrative reasons after the first mitazalimab infusion, prior to receiving mFOLFIRINOX and was not included in RP2D determination

Treatment Emergent Adverse Events

	Mitazalimab dose level (µg/kg)		
	450	900	Overall
Number of patients	5	6	11
Patients with any TEAEs, n (%)	5 (100)	4 (66.7)	9 (81.8)
Grade 1 or 2 TEAEs, n (%)	5 (100)	4 (66.7)	9 (81.8)
Grade 3 TEAEs, n (%)	3 (60)	1 (16.7)	4 (36.4)
Grade ≥4 TEAEs, n (%)	0	0	0

- Grade 1-2 mitazalimab related AEs included fatigue, fever, muscle pain and loss of appetite
- None of the grade 3 AEs in the 450 µg/kg cohort (abdominal pain, anorexia, dysphagia, denutrition) were classified as related to mitazalimab
- The patient with grade 3 AEs in the 900 µg/kg cohort developed fatigue and a headache that were classified as mitazalimab-related leading to treatment discontinuation

Peripheral blood markers align with mitazalimab mode of action

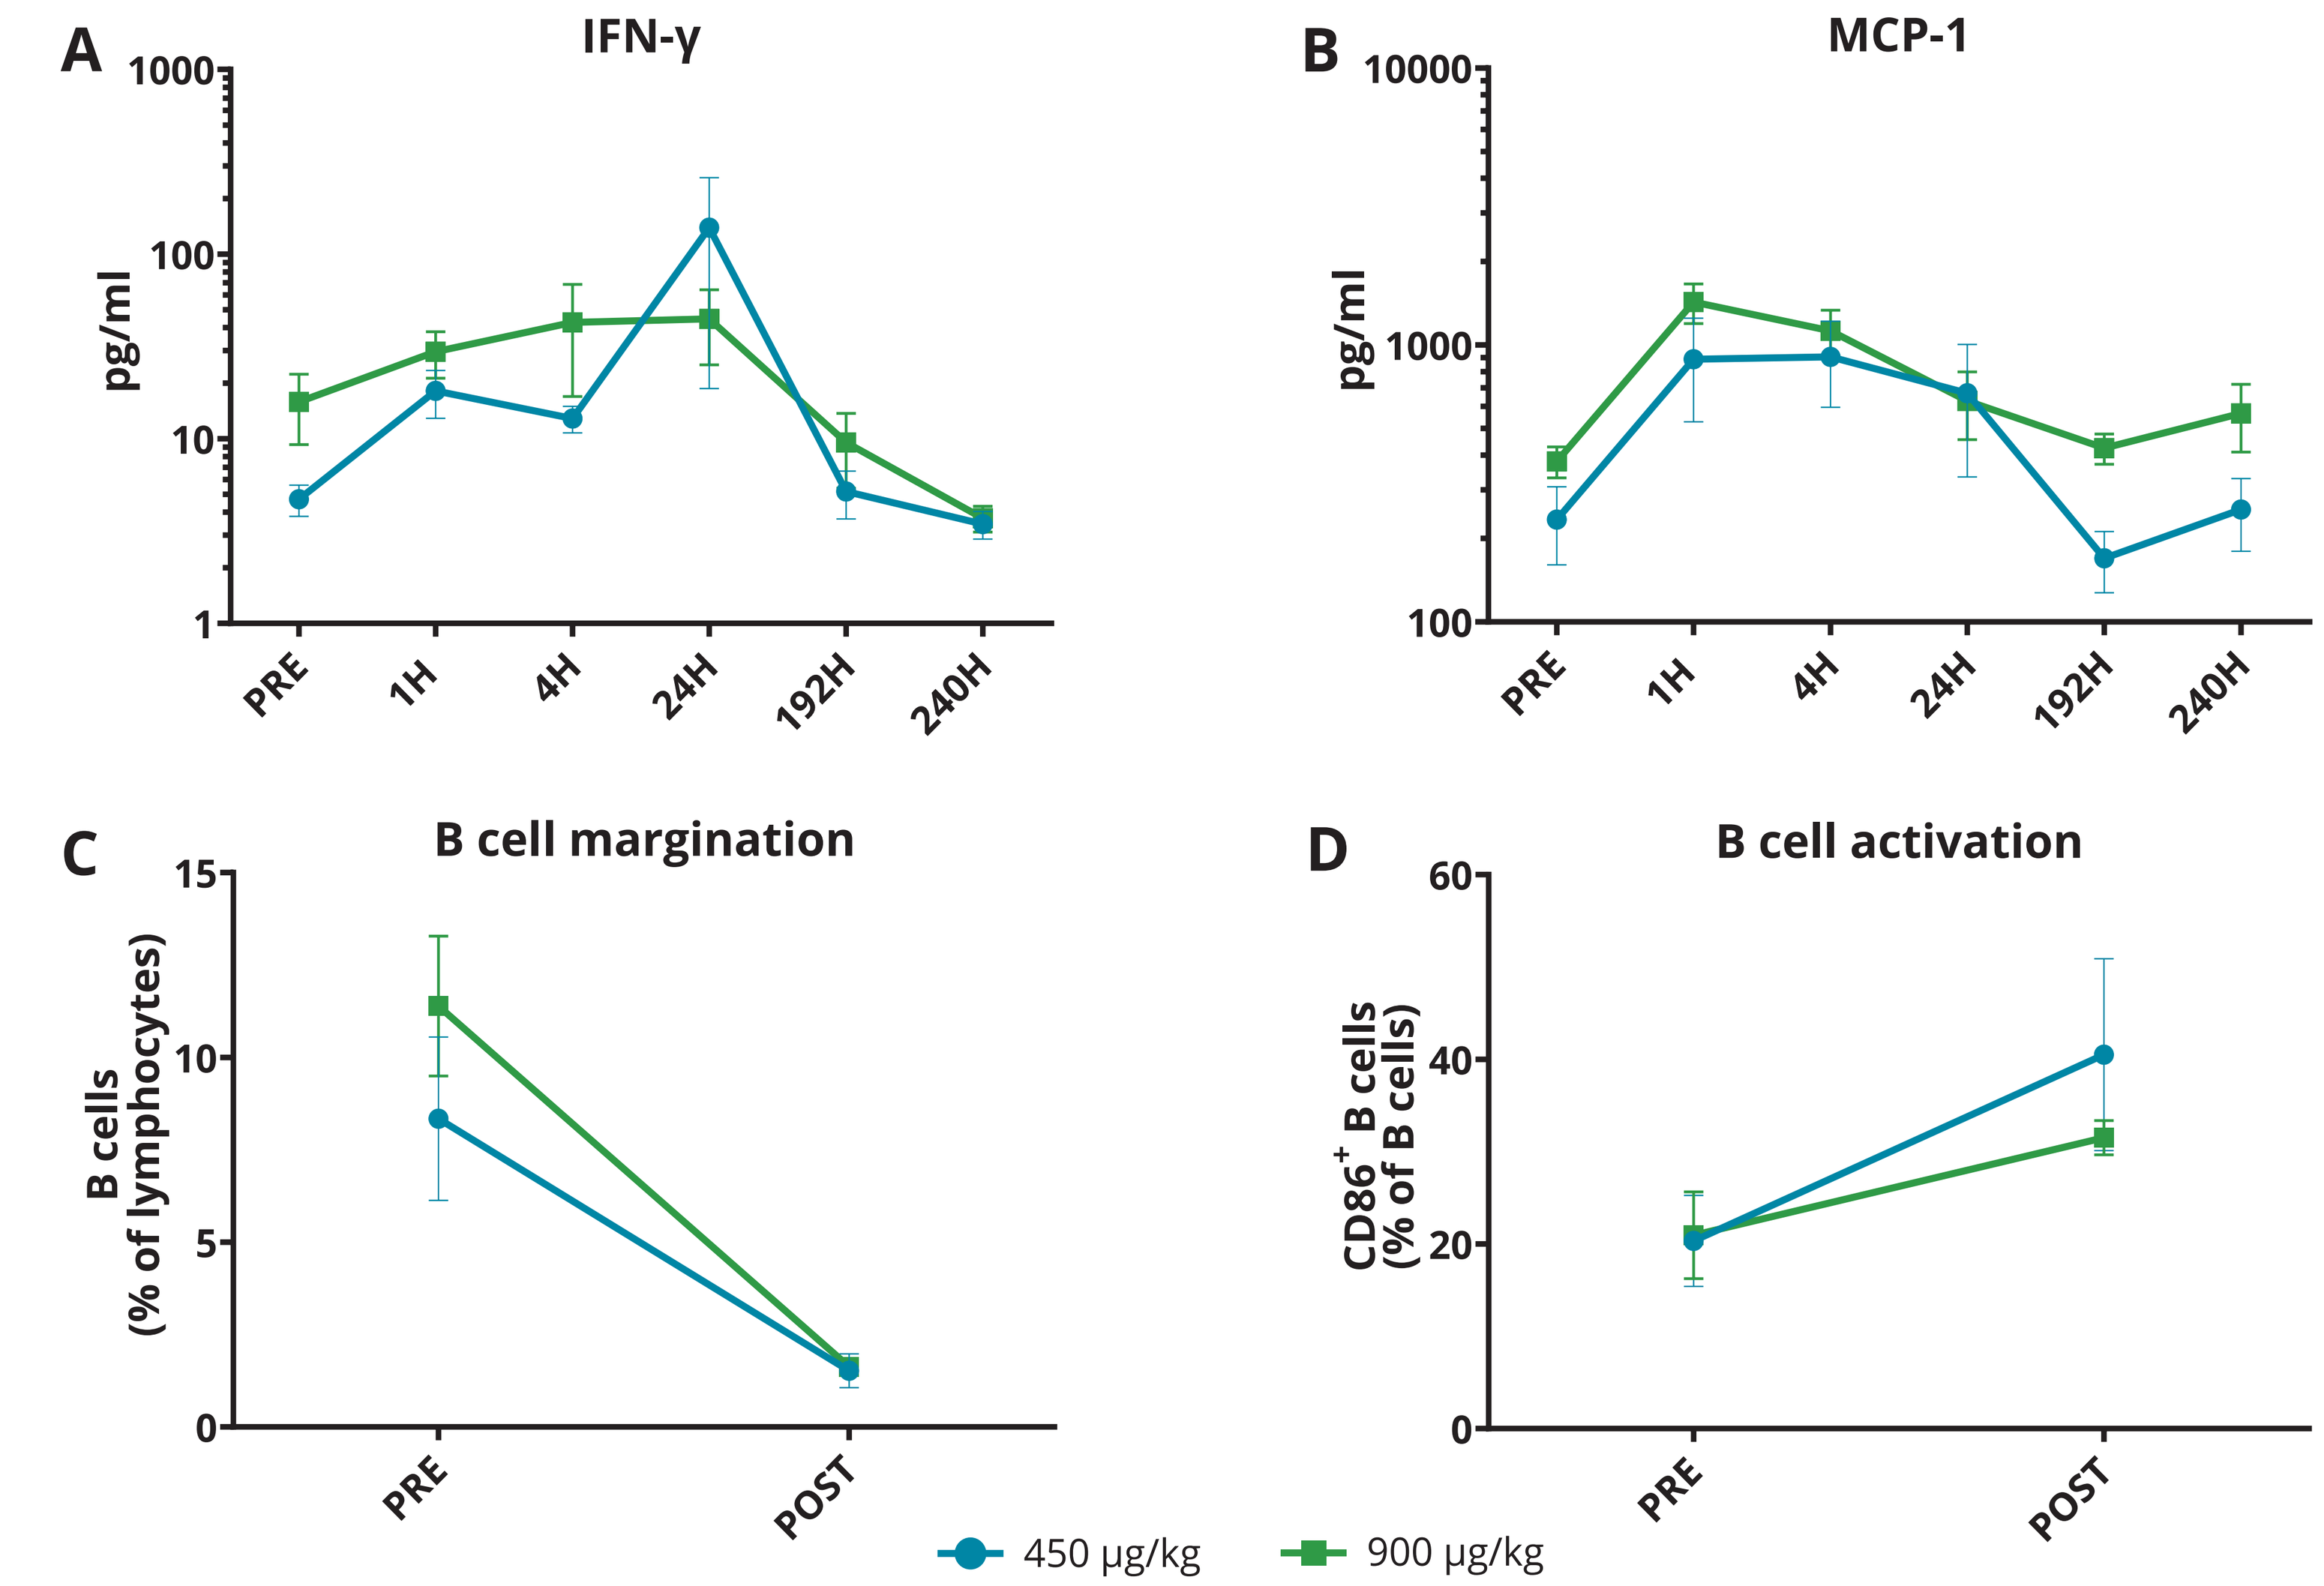
A
IFN-γ
pg/ml
1000
100
10
1
B
MCP-1
pg/ml
10000
1000
100
PRE 1H 4H 24H 192H 240H
C
B cell margination
B cells (% of lymphocytes)
15
10
5
0
D
B cell activation
CD86+ B cells (% of B cells)
60
40
20
0
PRE POST
450 µg/kg
900 µg/kg

Modulation of cytokine/chemokines and immune cell populations analyzed in peripheral blood following infusion with mitazalimab. (A) IFN-γ; (B) MCP-1; (C) Circulating B cells; (D) B cell activation.

- Cytokine and chemokine increases in peripheral blood confirm activation of T cells and myeloid cells
- B cell margination and B cell activation for remaining B cells was observed in peripheral blood post-treatment with mitazalimab

Conclusions

- Mitazalimab combined with mFOLFIRINOX is safe and well tolerated.
- Mitazalimab related AEs at both dose levels (450 and 900 µg/kg) occurring in >1 patient were fever, muscle pain loss of apetite and fatigue
- Peripheral blood markers confirm immune activation, in accordance with mitazalimab's mode of action
- 900 µg/kg dose of mitazalimab was selected as the RP2D
- Phase 2 of the OPTIMIZE-1 trial is currently enrolling patients

ACKNOWLEDGEMENTS:
- **The patients and their families**
- **The study investigators and clinical research staff**

References

1. Enell Smith, K., Deronic, A., Hägerbrand, K., Norlén, P. & Ellmark, P. Rationale and clinical development of CD40 agonistic antibodies for cancer immunotherapy. Exp Op Biological Therapy, 2021
2. Ellmark, P., Mangsbo, S.M., Furebring, C., Totterman, T.H. & Norlen, P. Kick-starting the cancer-immunity cycle by targeting CD40. Oncoimmunology, 2015

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from the author of this poster.

ALLIGATOR bioscience