Combination treatment with ATOR-4066, a Neo-X-Prime™ bispecific antibody targeting CD40 and CEACAM5, and anti-PD-1 reverses T cell exhaustion *in vitro*

Ida Uddbäck[1*], Sara Fritzell[1], Lill Ljung[1], Anneli Nilsson[1], Mona Celander[1], Amulya Krishna Shetty[1], Maria Johansson[1], Hampus Andersson[1,2], David Gomez Jimenez[1], Mattias Levin[1], Malin Lindstedt[1,2], Karin Hägerbrand[1], Anette Sundstedt[1] and Peter Ellmark[1,2]

[1] Alligator Bioscience AB, Medicon Village, Lund, Sweden; [2] Department of Immunotechnology, Lund University, Lund, Sweden *Presenting author

Abstract 837

INTRODUCTION

- ATOR-4066 is a preclinical stage bispecific antibody targeting CD40 and CEACAM5, developed using Alligator's novel Neo-X-Prime™ platform, which induces neoantigen specific T-cell response by activating antigen presenting cells (APCs)
- ATOR-4066 binds to CD40 on dendritic cells (DCs) and CEACAM5, a tumor-associated antigen (TAA), expressed on tumor cells and on tumor-derived material (such as exosomes or tumor debris containing neoantigen), leading to tumor directed activation of the DCs, enhanced uptake of tumor-derived material, cross-presentation of neoantigen, priming of neoantigen-specific T cells and killing of tumor cells.
- We have previously demonstrated potent anti-tumor efficacy of ATOR-4066 treatment *in vivo*
- Moreover, ATOR-4066 induces strong activation of CD40 expressing cells *in vitro* using CEACAM5 expressing tumor material from patients
- Here we present further preclinical data strengthening the potential of ATOR-4066 as an anti-tumor treatment both as a stand-alone therapy but also in combination with anti-PD-1 treatment

ATOR-4066: CD40 x CEACAM5 targeting bispecific antibody

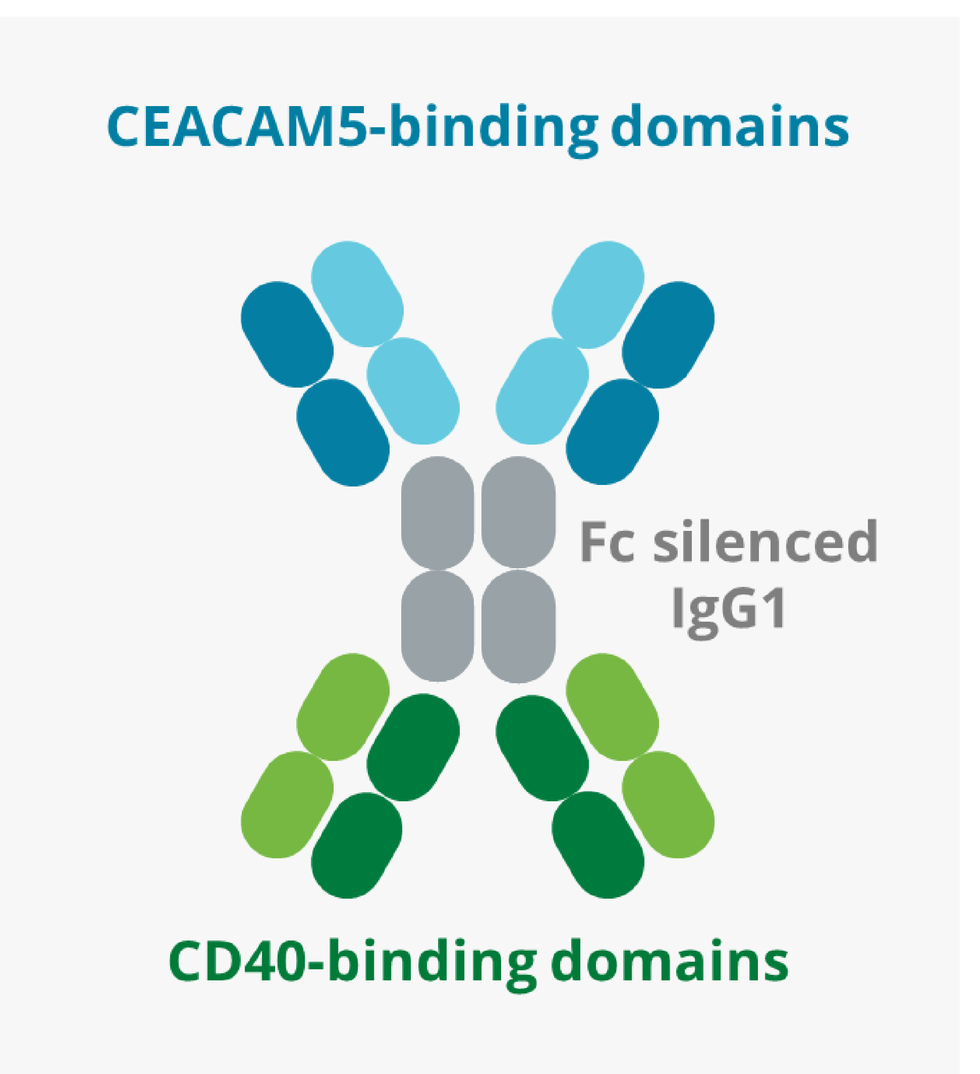
CEACAM5-binding domains
Fc silenced IgG1
CD40-binding domains

- ATOR-4066 is a human Fc-silenced IgG1 in the bispecific tetravalent RUBY® format, comprising two sets of binders targeting CD40 and the TAA carcinoembryonic antigen 5 (CEACAM5) developed for immunotherapy for patients with advanced solid cancers expressing CEACAM5.
- ATOR-4066 binds to both targets simultaneously and subsequently activates CD40-expressing cells only when binding to CEACAM5-expressing tumor cells. The CEACAM5-conditional activation is anticipated to limit toxicity due to systemic immune activation.

MODE OF ACTION

MoA of ATOR-4066:

A. CEAMCAM5-conditional CD40 activation of DCs and macrophages

B. Novel mechanism for cross-priming of neoantigen specific T cells through enhanced DC update of CEACAM5-expressing tumor-derived material

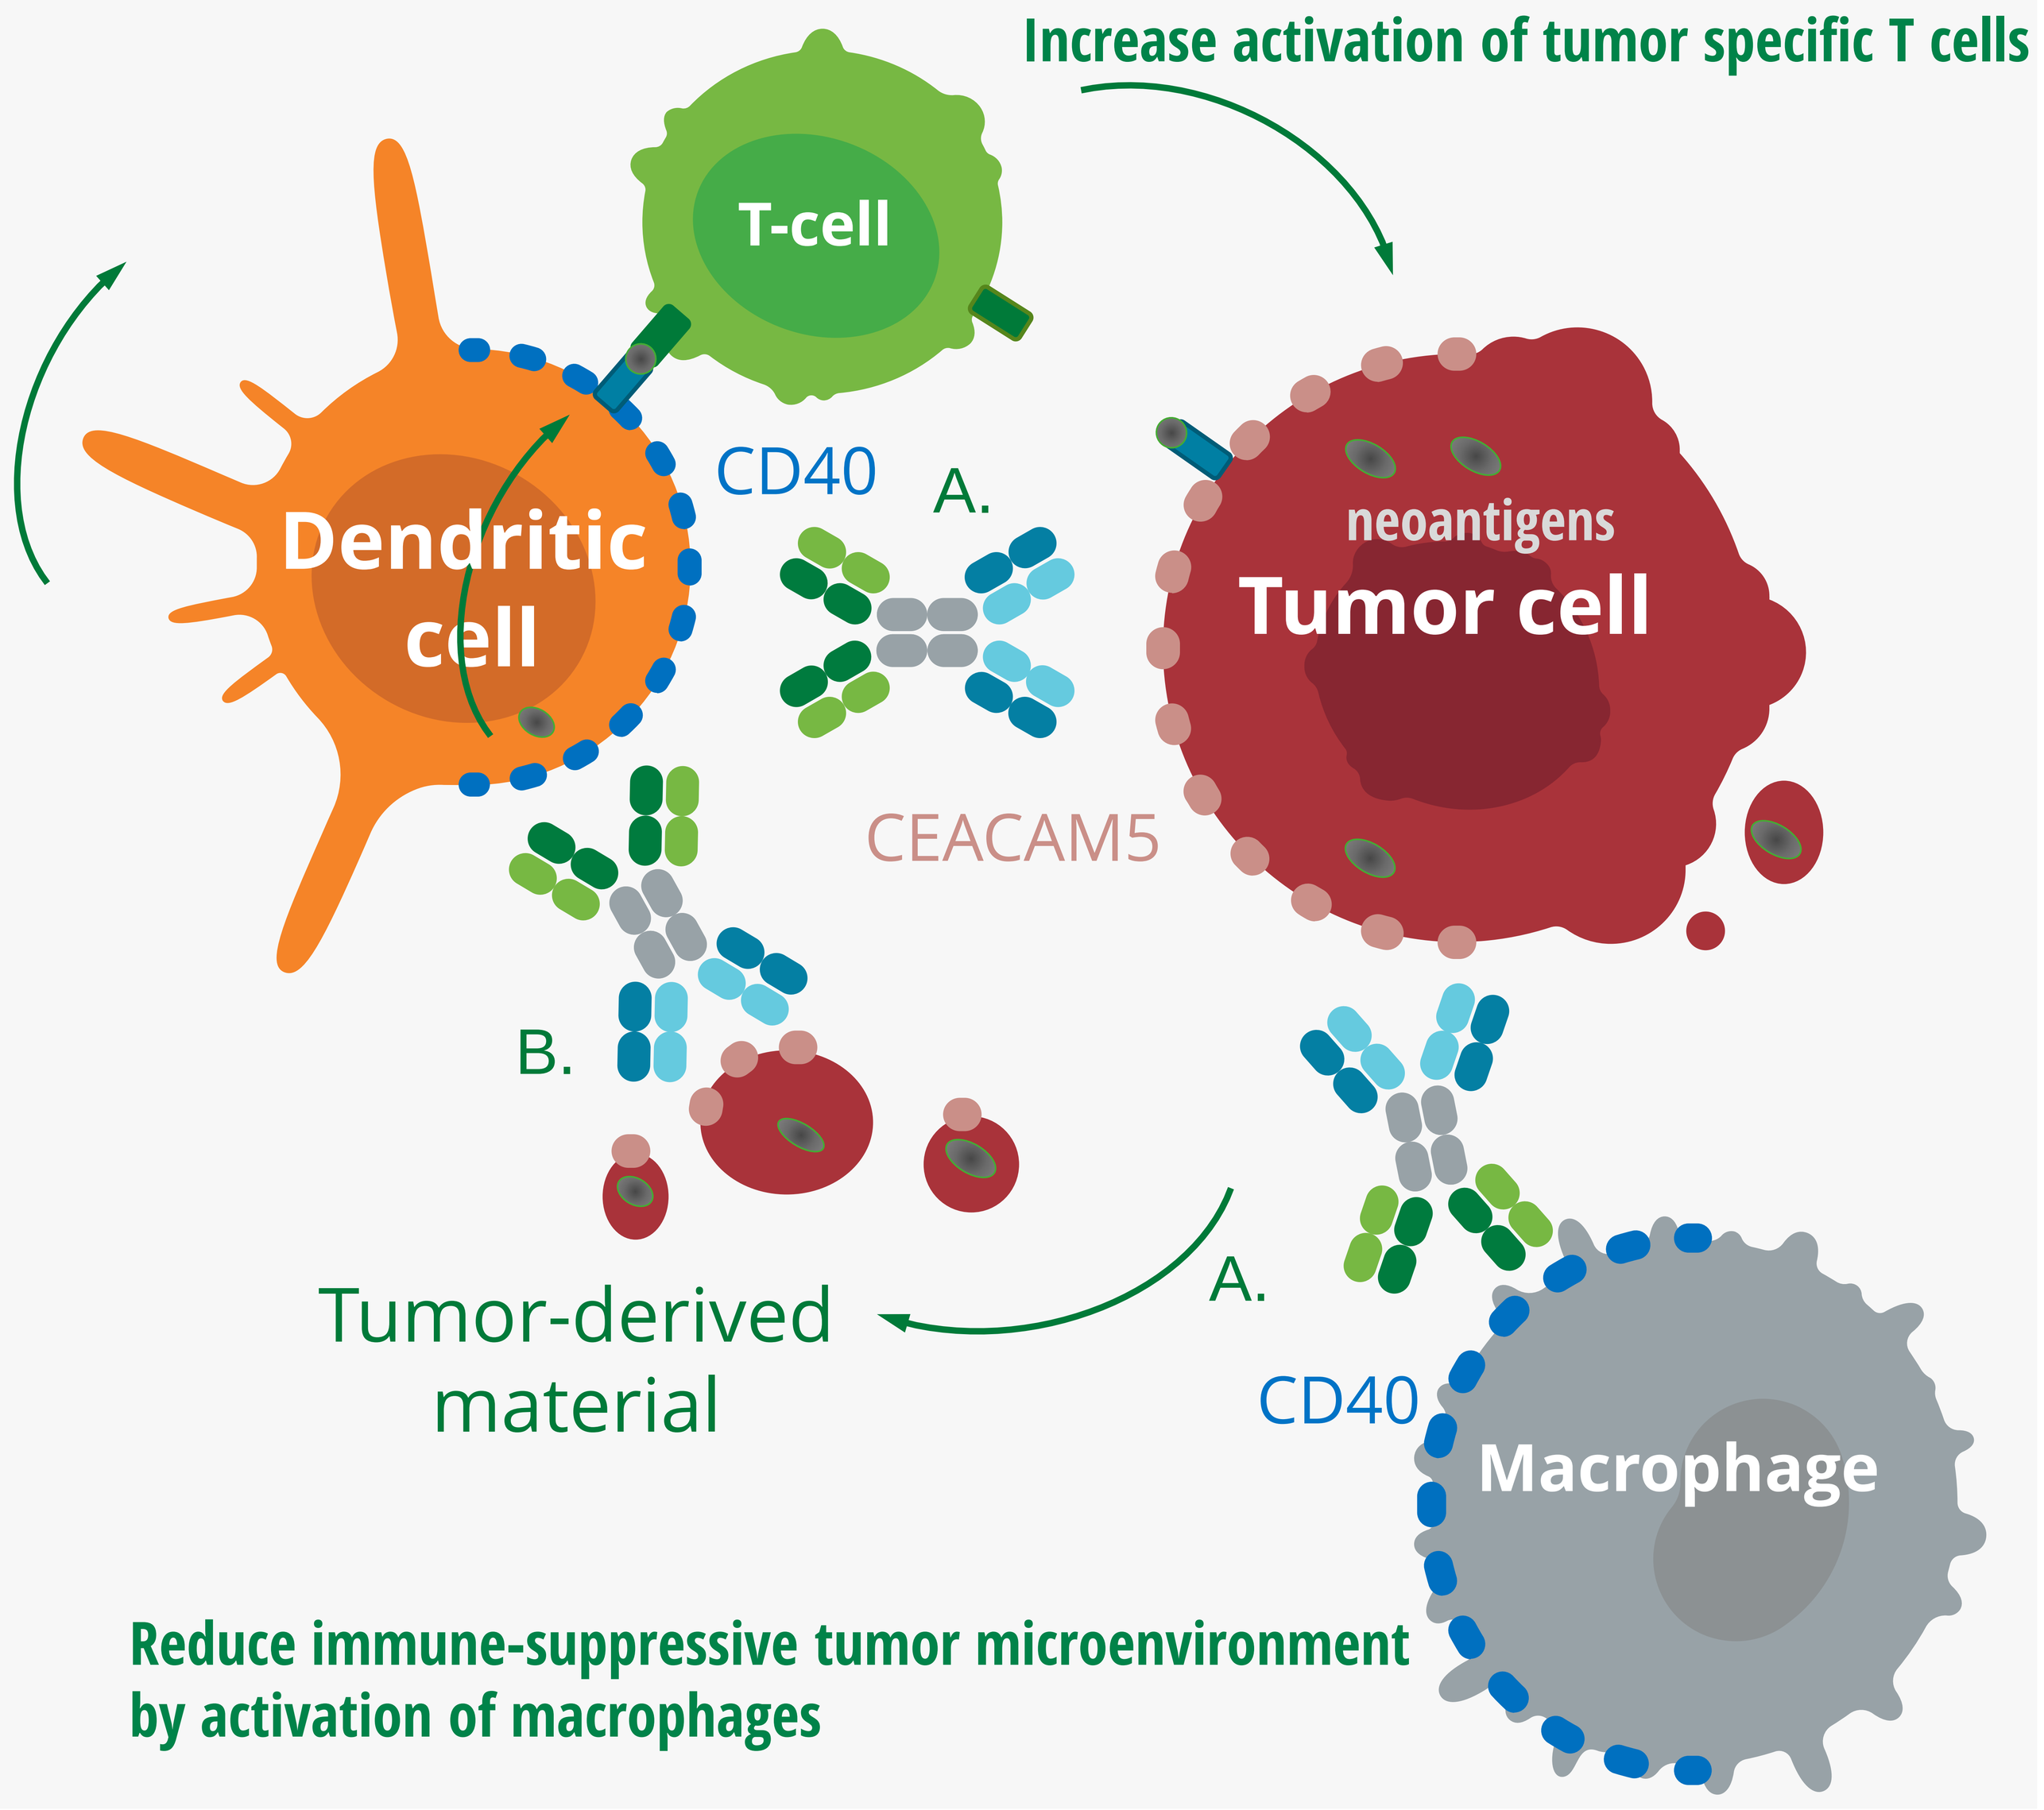
Increase activation of tumor specific T cells
T-cell
Dendritic cell
CD40
A.
neoantigens
Tumor cell
CEACAM5
B.
Tumor-derived material
A.
CD40
Macrophage
Reduce immune-suppressive tumor microenvironment by activation of macrophages

[1]Hägerbrand K, Varas L, Deronic A, *et al*. Bispecific antibodies targeting CD40 and tumor-associated antigens promote cross-priming of T cells resulting in an antitumor response superior to monospecific antibodies. *Journal for ImmunoTherapy of Cancer* 2022;**10:**e005018. doi: 10.1136/jitc-2022-005018.

RESULTS

CEACAM5-conditional activation of primary human B cells and monocyte-derived DCs by ATOR-4066

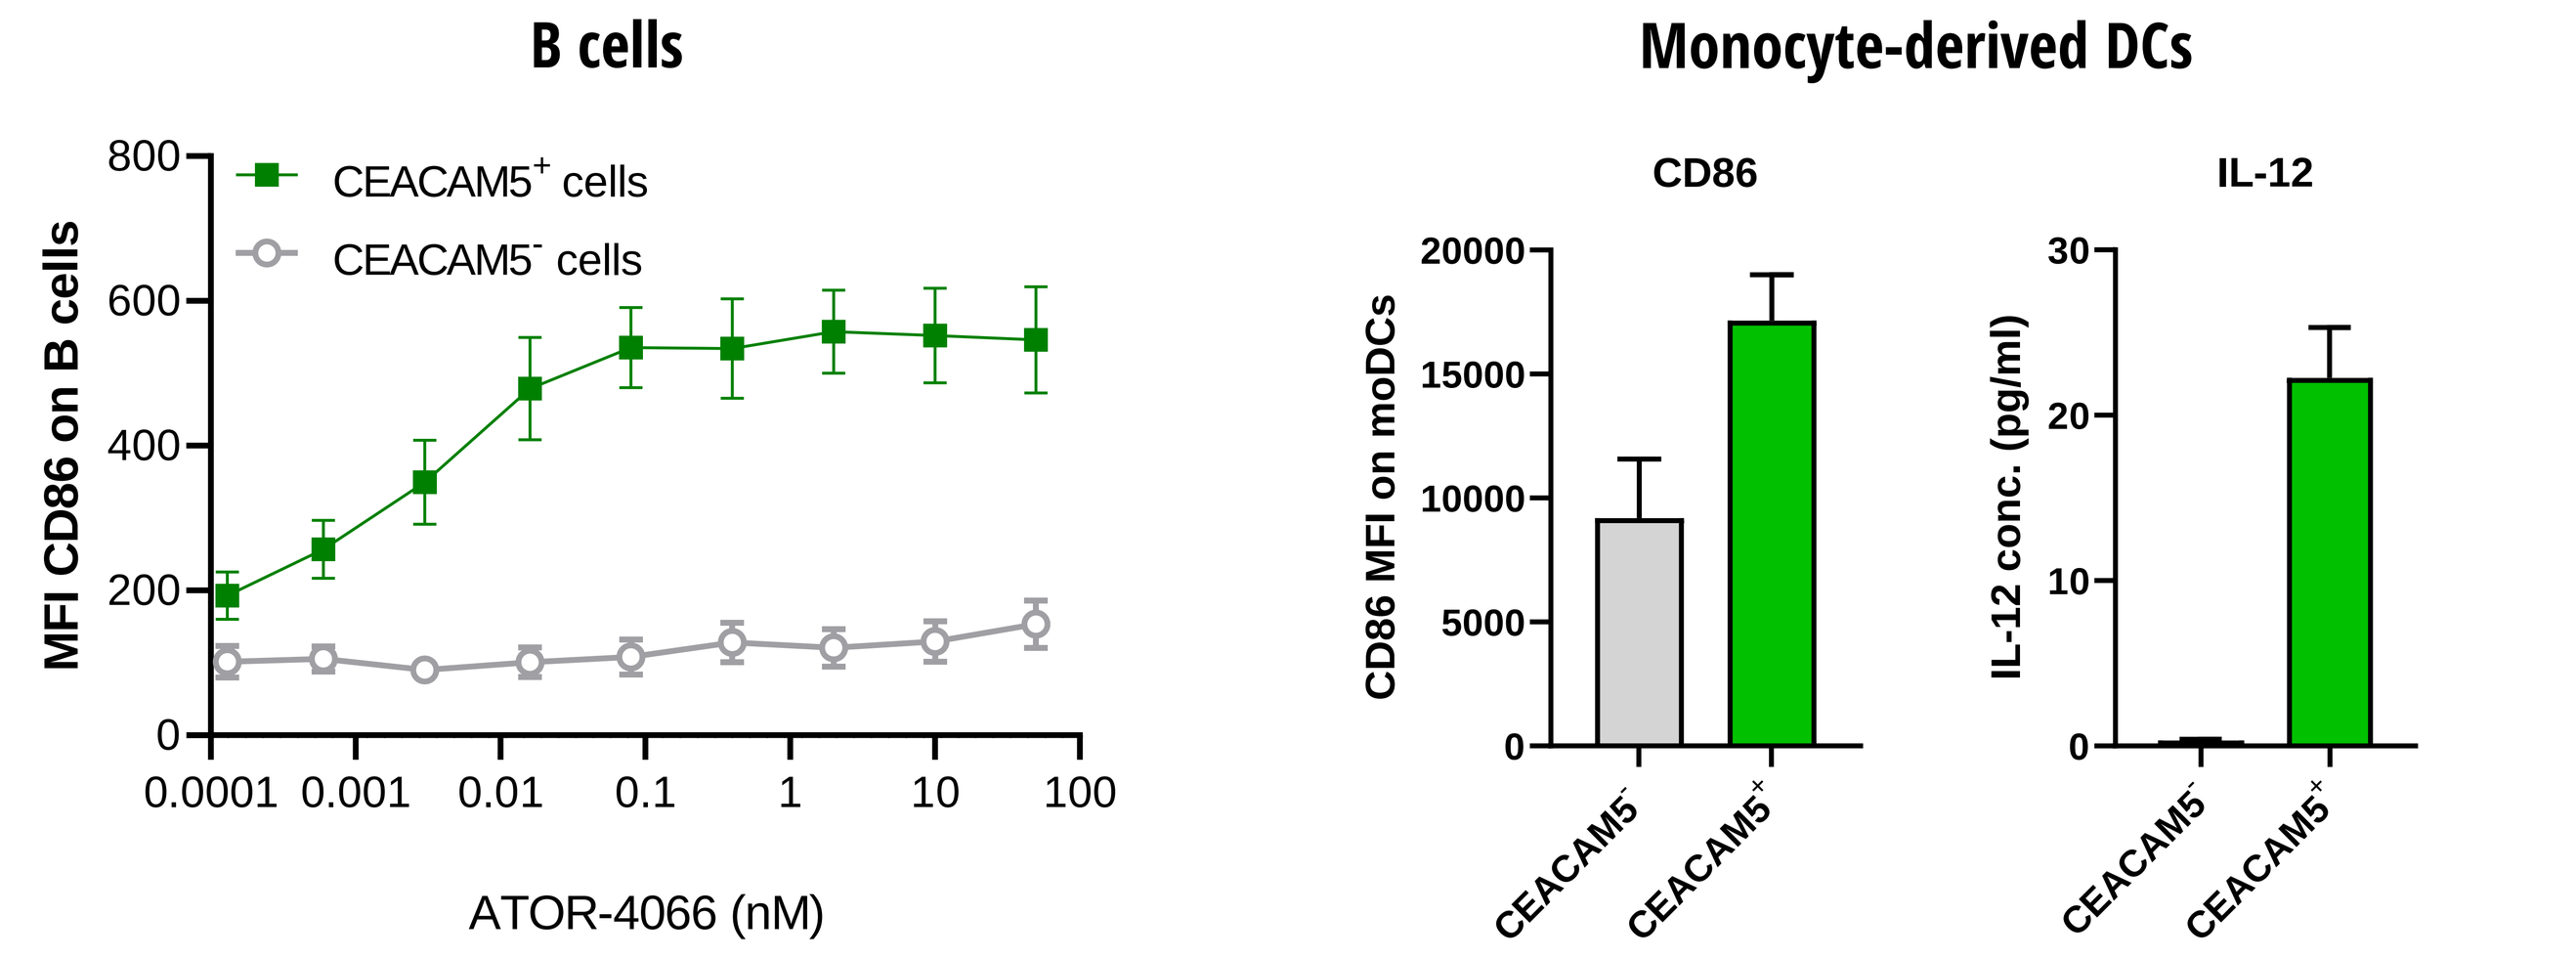
B cells
Monocyte-derived DCs
CEACAM5+ cells
CEACAM5- cells
MFI CD86 on B cells
ATOR-4066 (nM)
CD86
IL-12
CD86 MFI on moDCs
IL-12 conc. (pg/ml)
CEACAM5-
CEACAM5+

Figure 1. Primary human B cells were cultured in the presence of CEACAM5-transfected or wildtype CHO cells and activated by ATOR-4066 for 48 hours (hrs). The expression of CD86 was analyzed by flow cytometry (n=5; left graph). Monocyte-derived DCs (moDCs) were cultured in the presence of CEACAM5-coated or uncoated beads and ATOR-4066 for 48 hrs. The expression of CD86 was analyzed by flow cytometry and IL-12p40 levels in supernatants were analyzed by ELISA (n=3; right graphs).

ATOR-4066 mediates clustering of CEACAM5-expressing tumor debris with CD40-expressing APCs

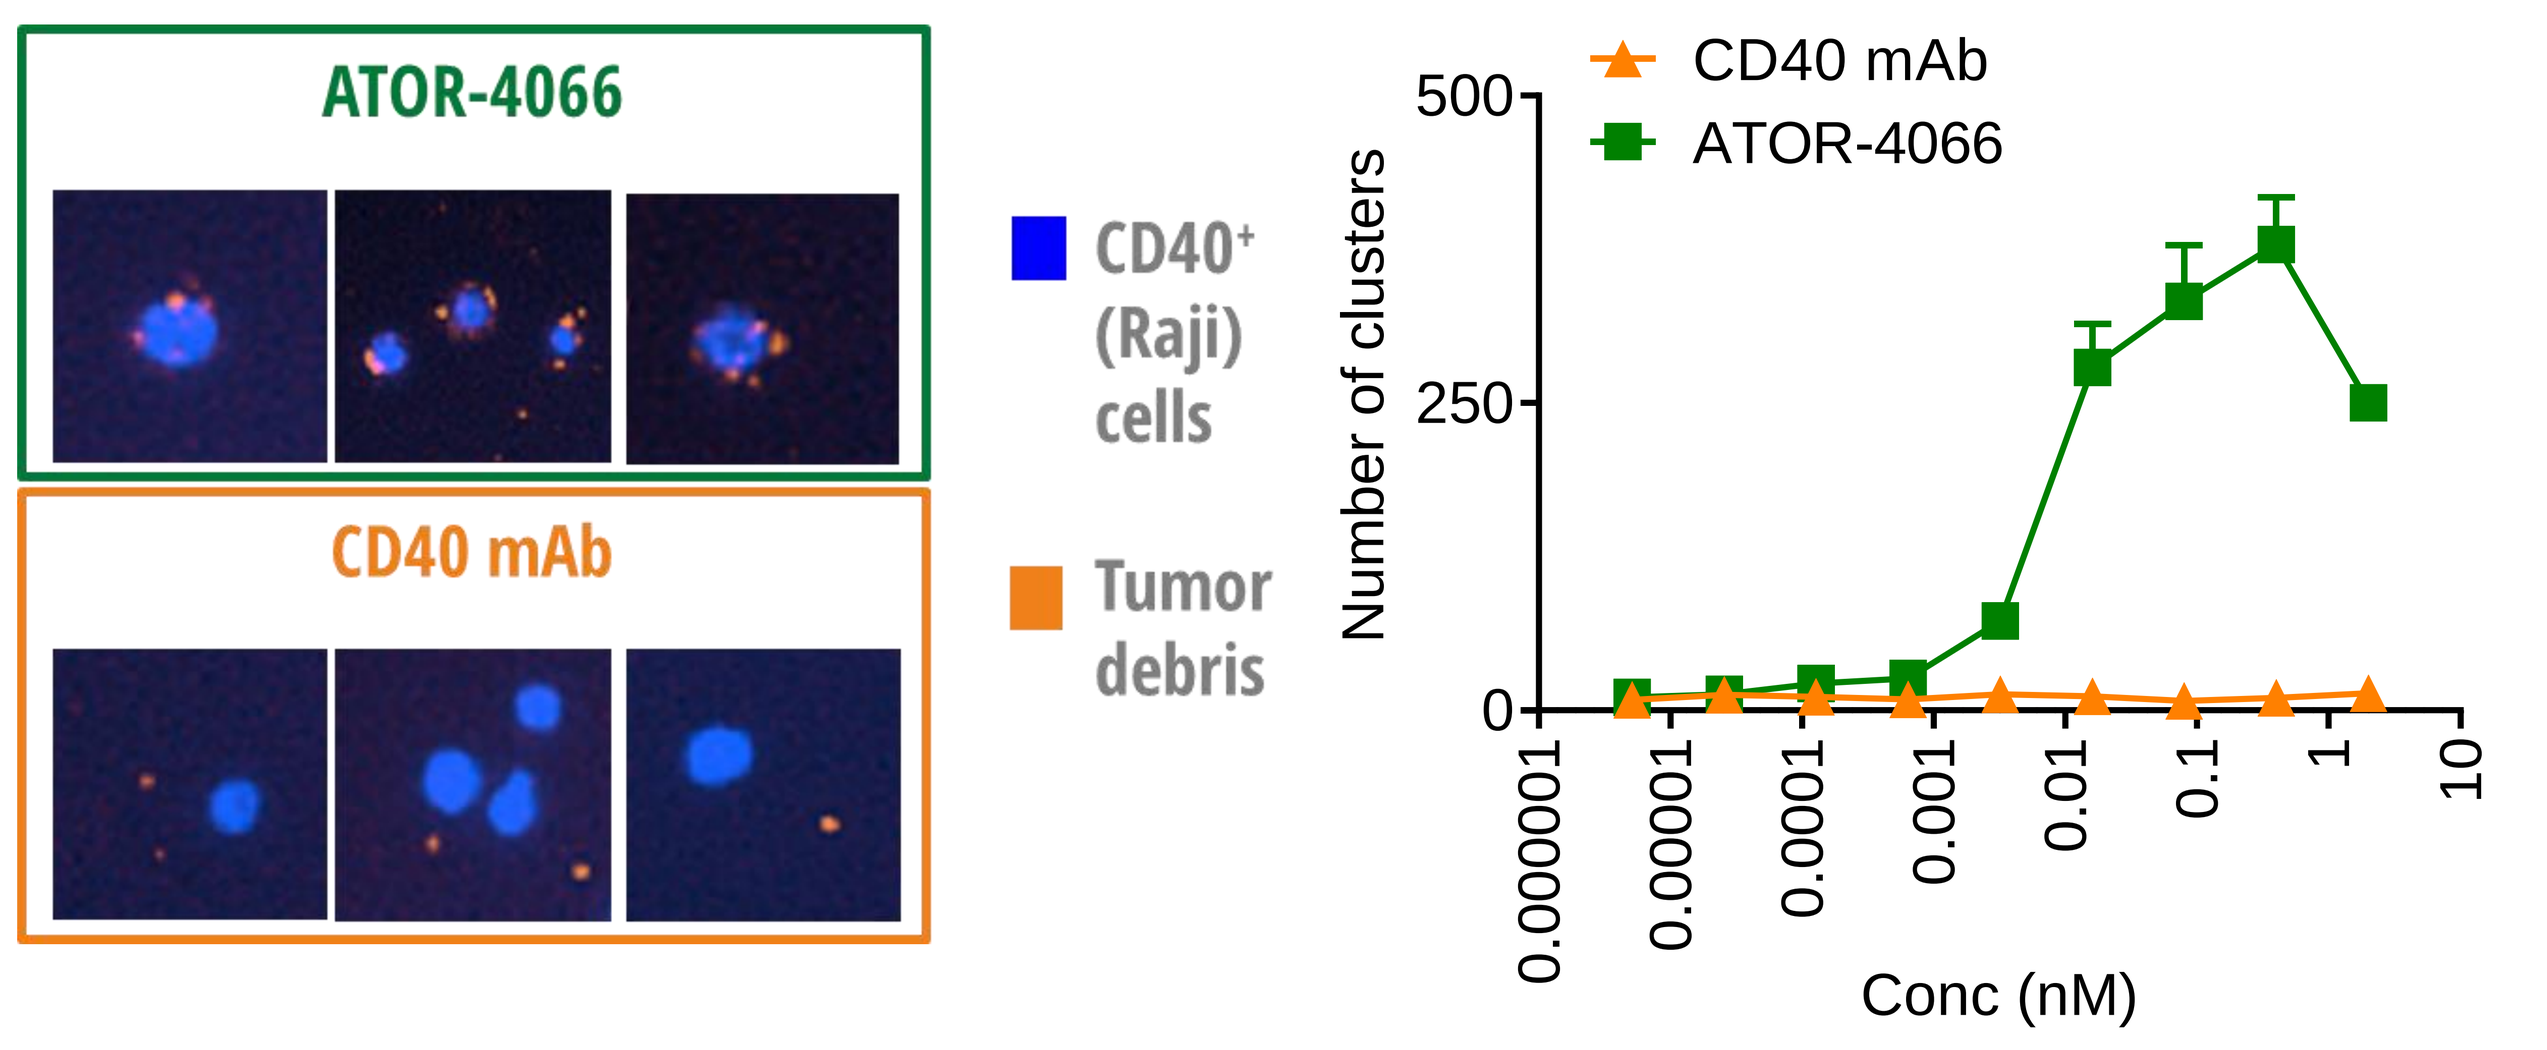
ATOR-4066
CD40 mAb
CD40+ (Raji) cells
Tumor debris
CD40 mAb
ATOR-4066
Number of clusters
Conc (nM)

Figure 2. Fluorescently labeled CD40⁺ Raji cells were cultured with fluorescently labeled CEACAM5⁺ tumor debris and titrating concentrations of CD40 mAb or ATOR-4066. Images were captured using a live cell imaging system and clusters of CD40⁺ cells co-localized with tumor debris were quantified after 8 hrs of culture.

Strong anti-tumor efficacy and induction of immunological memory after ATOR-4066 treatment *in vivo*

Experimental setup in human CD40 transgenic mice

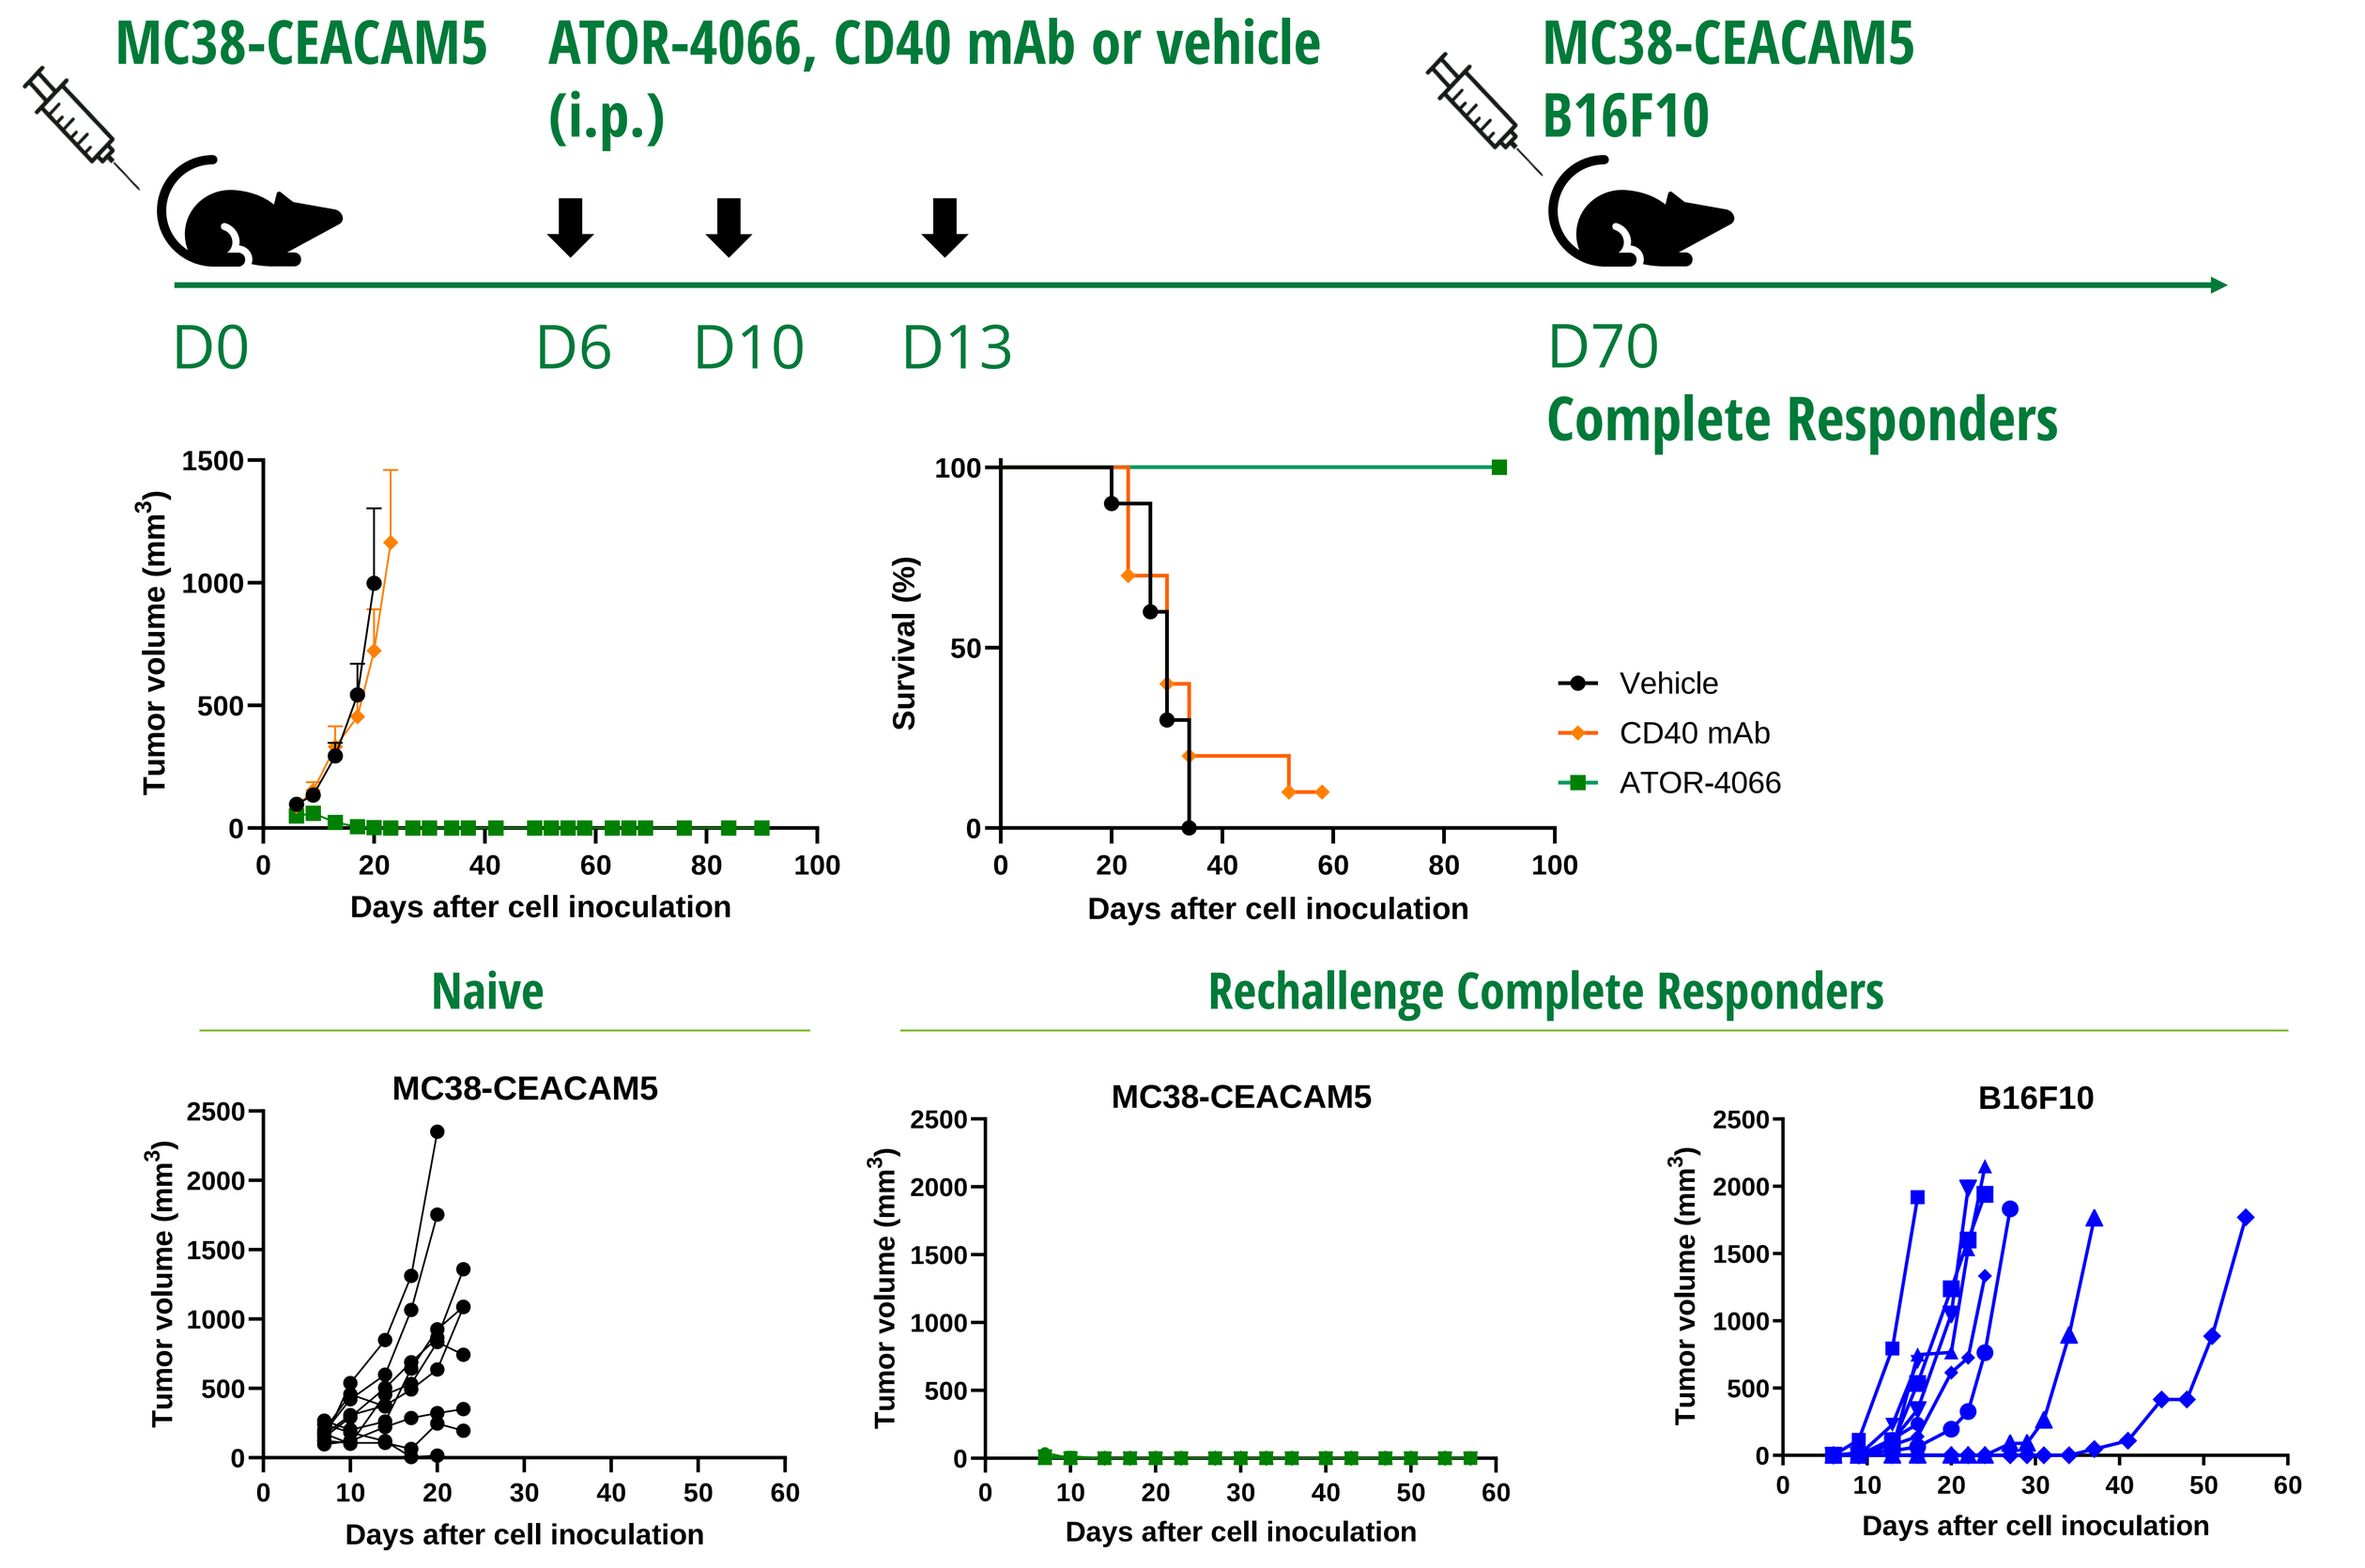
MC38-CEACAM5
ATOR-4066, CD40 mAb or vehicle (i.p.)
MC38-CEACAM5
B16F10
D0
D6
D10
D13
D70
Complete Responders
Tumor volume (mm3)
Days after cell inoculation
Survival (%)
Vehicle
CD40 mAb
ATOR-4066
Naive
Rechallenge Complete Responders
MC38-CEACAM5
MC38-CEACAM5
B16F10

Figure 3. Human CD40 transgenic (hCD40tg) mice were inoculated subcutaneously (s.c.) with MC38-CEACAM5 cells and 100 µg CD40 mAb or a molar equivalent dose (167 µg) of ATOR-4066 or vehicle control were administered intraperitoneally (i.p.) on days 6, 10 and 13 (top row) and tumor volume and survival was monitored. After 70 days, complete responders, mice cured from their primary tumors by ATOR-4066 treatment, were rechallenged with homologous (MC38-CEACAM5) or irrelevant (B16) tumors (bottom row). Naïve hCD40tg mice served as tumor growth control. Tumor volume mean±SEM (n=10/group) is presented.

ATOR-4066 displays strong anti-tumor effect *in vivo* also in larger tumors with heterogenous CEACAM5 expression

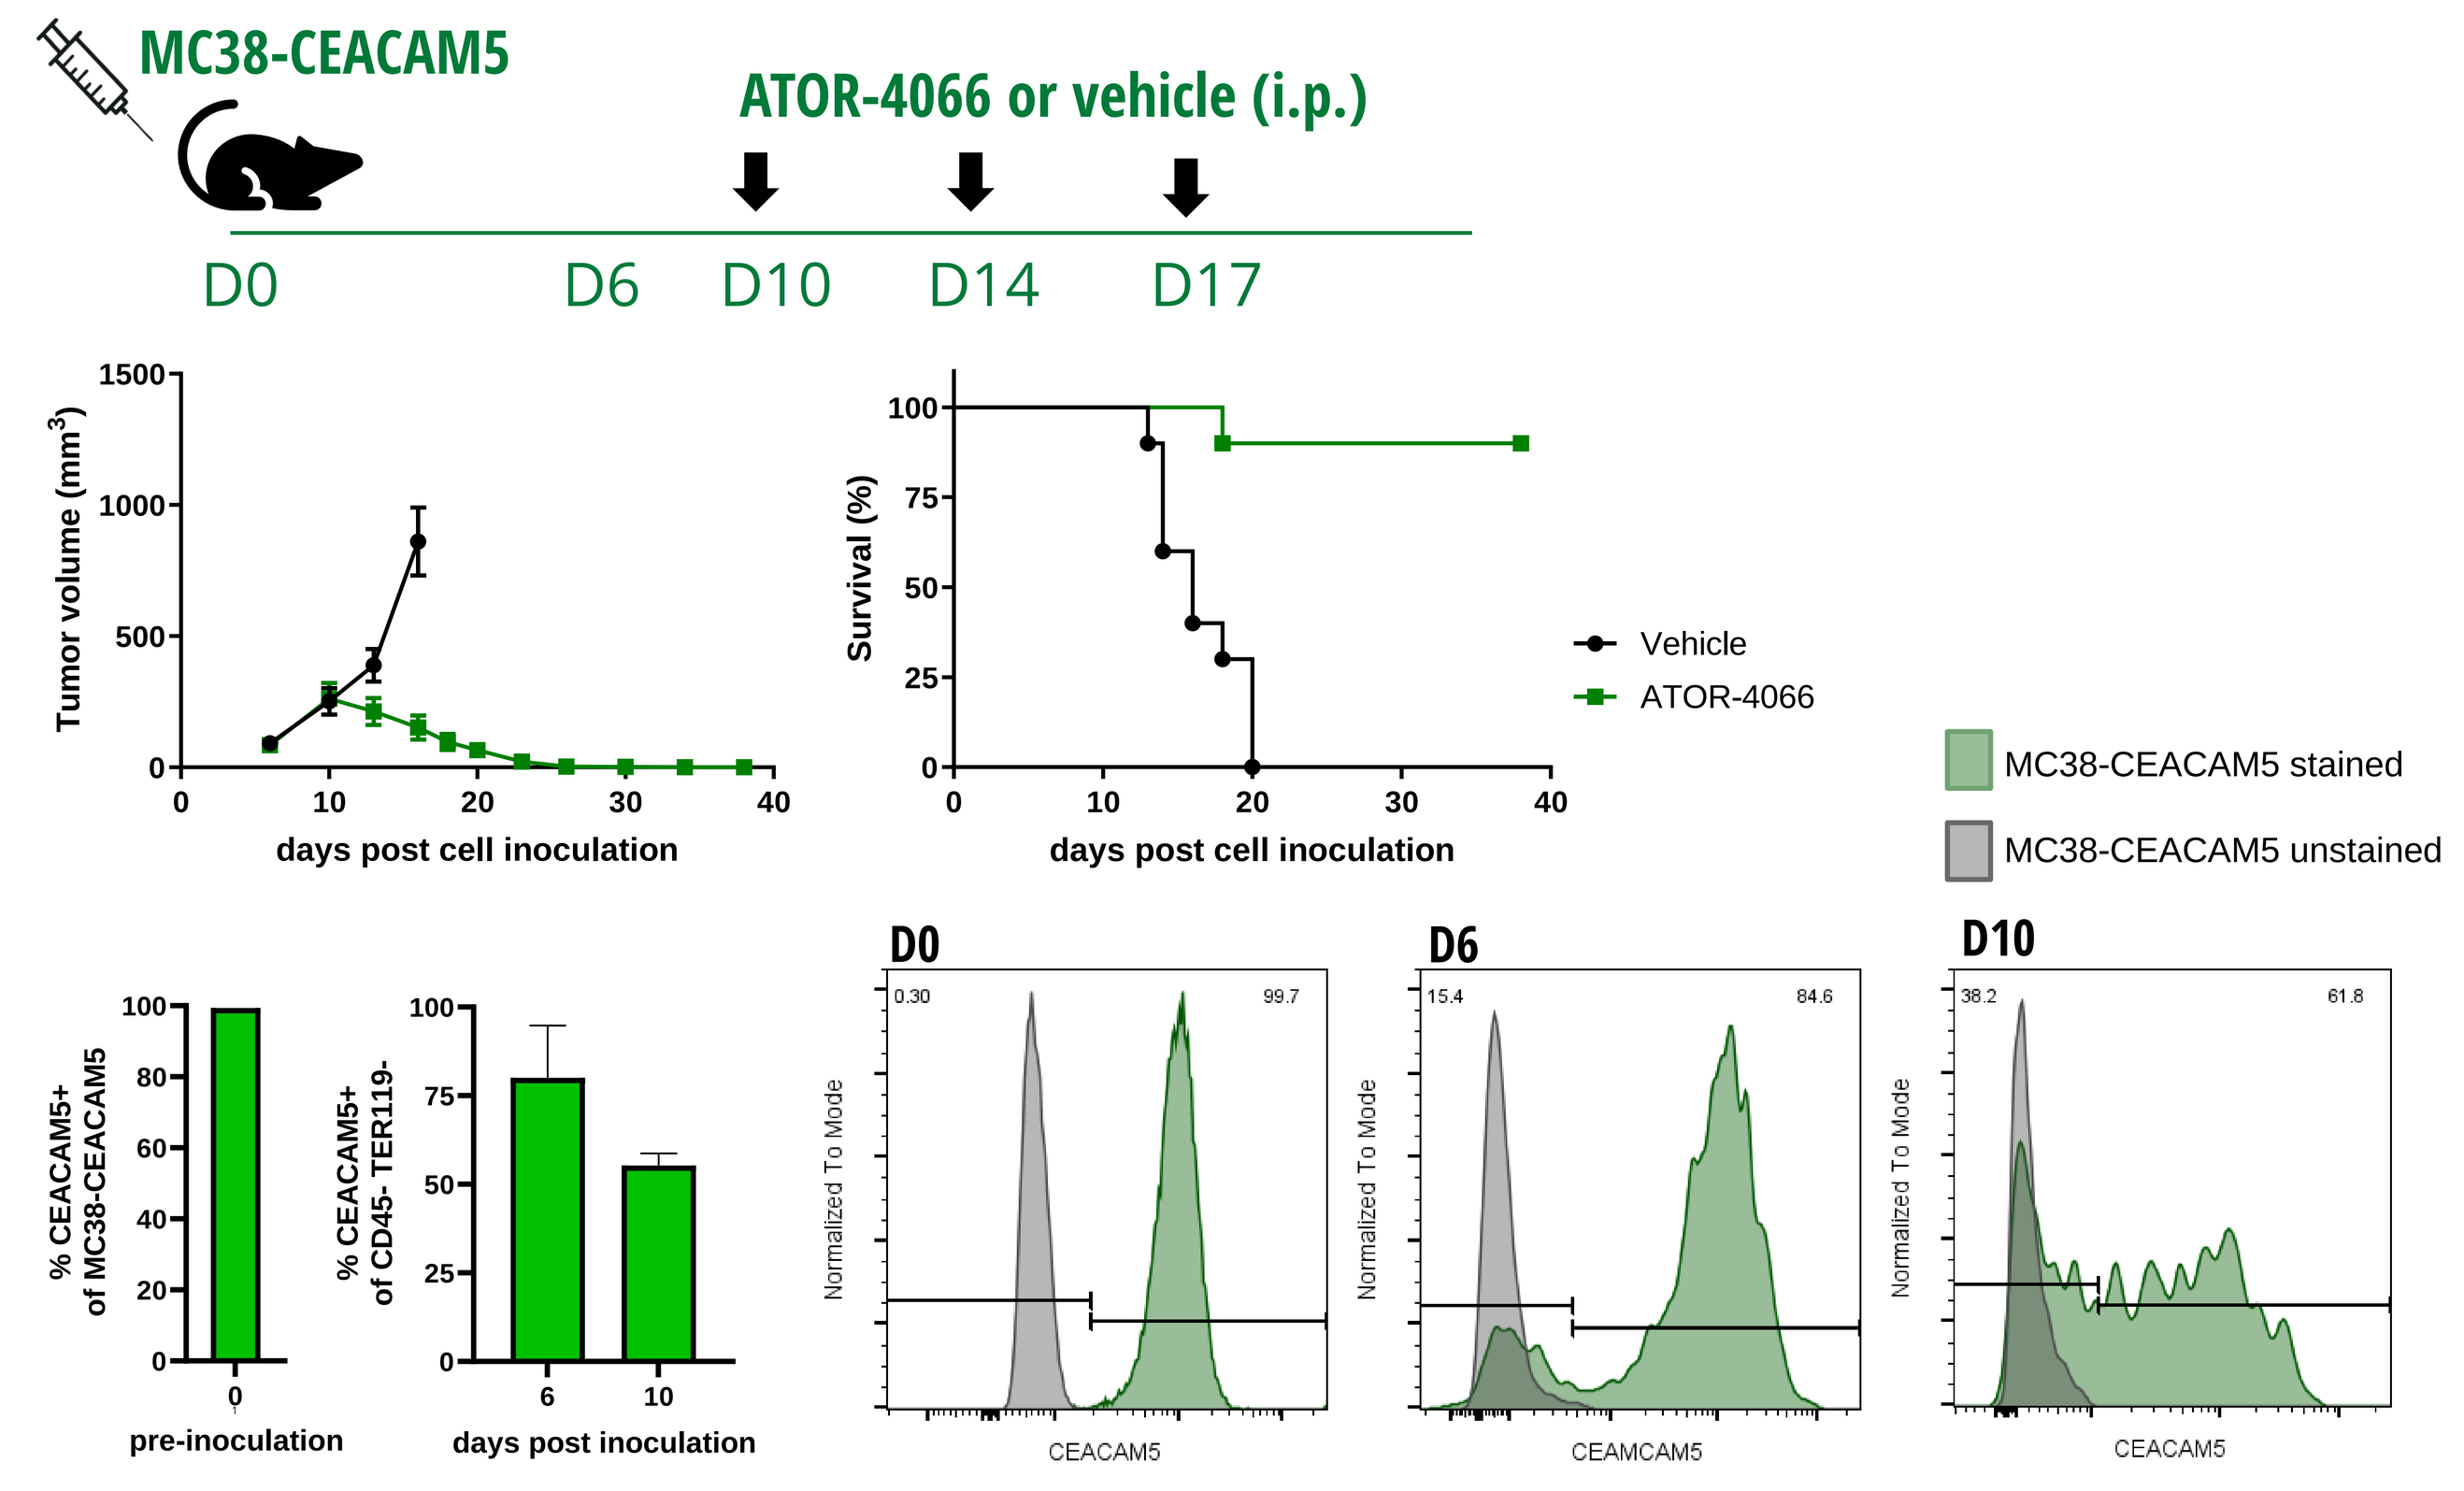
MC38-CEACAM5
ATOR-4066 or vehicle (i.p.)
D0
D6
D10
D14
D17
Tumor volume (mm3)
days post cell inoculation
Survival (%)
Vehicle
ATOR-4066
MC38-CEACAM5 stained
MC38-CEACAM5 unstained
% CEACAM5+ of MC38-CEACAM5
pre-inoculation
% CEACAM5+ of CD45- TER119-
days post inoculation
D0
D6
D10
Normalized To Mode
CEACAM5

Figure 4. hCD40tg mice were inoculated with MC38-CEACAM5 cells s.c. and 167 µg of ATOR-4066 or vehicle control were administered i.p. on days 10, 14 and 17 (top row). CEACAM5 expression was evaluated in tumor cells prior inoculation (bottom left graph) and ex vivo from tumors in untreated mice days 6 and 10 after inoculation (bottom right graph). Representative histograms of CEACAM5 expression to the right. Tumor volume mean±SEM (n=10/group).

Induction of exhausted T cells by repeated T cell stimulation *in vitro*

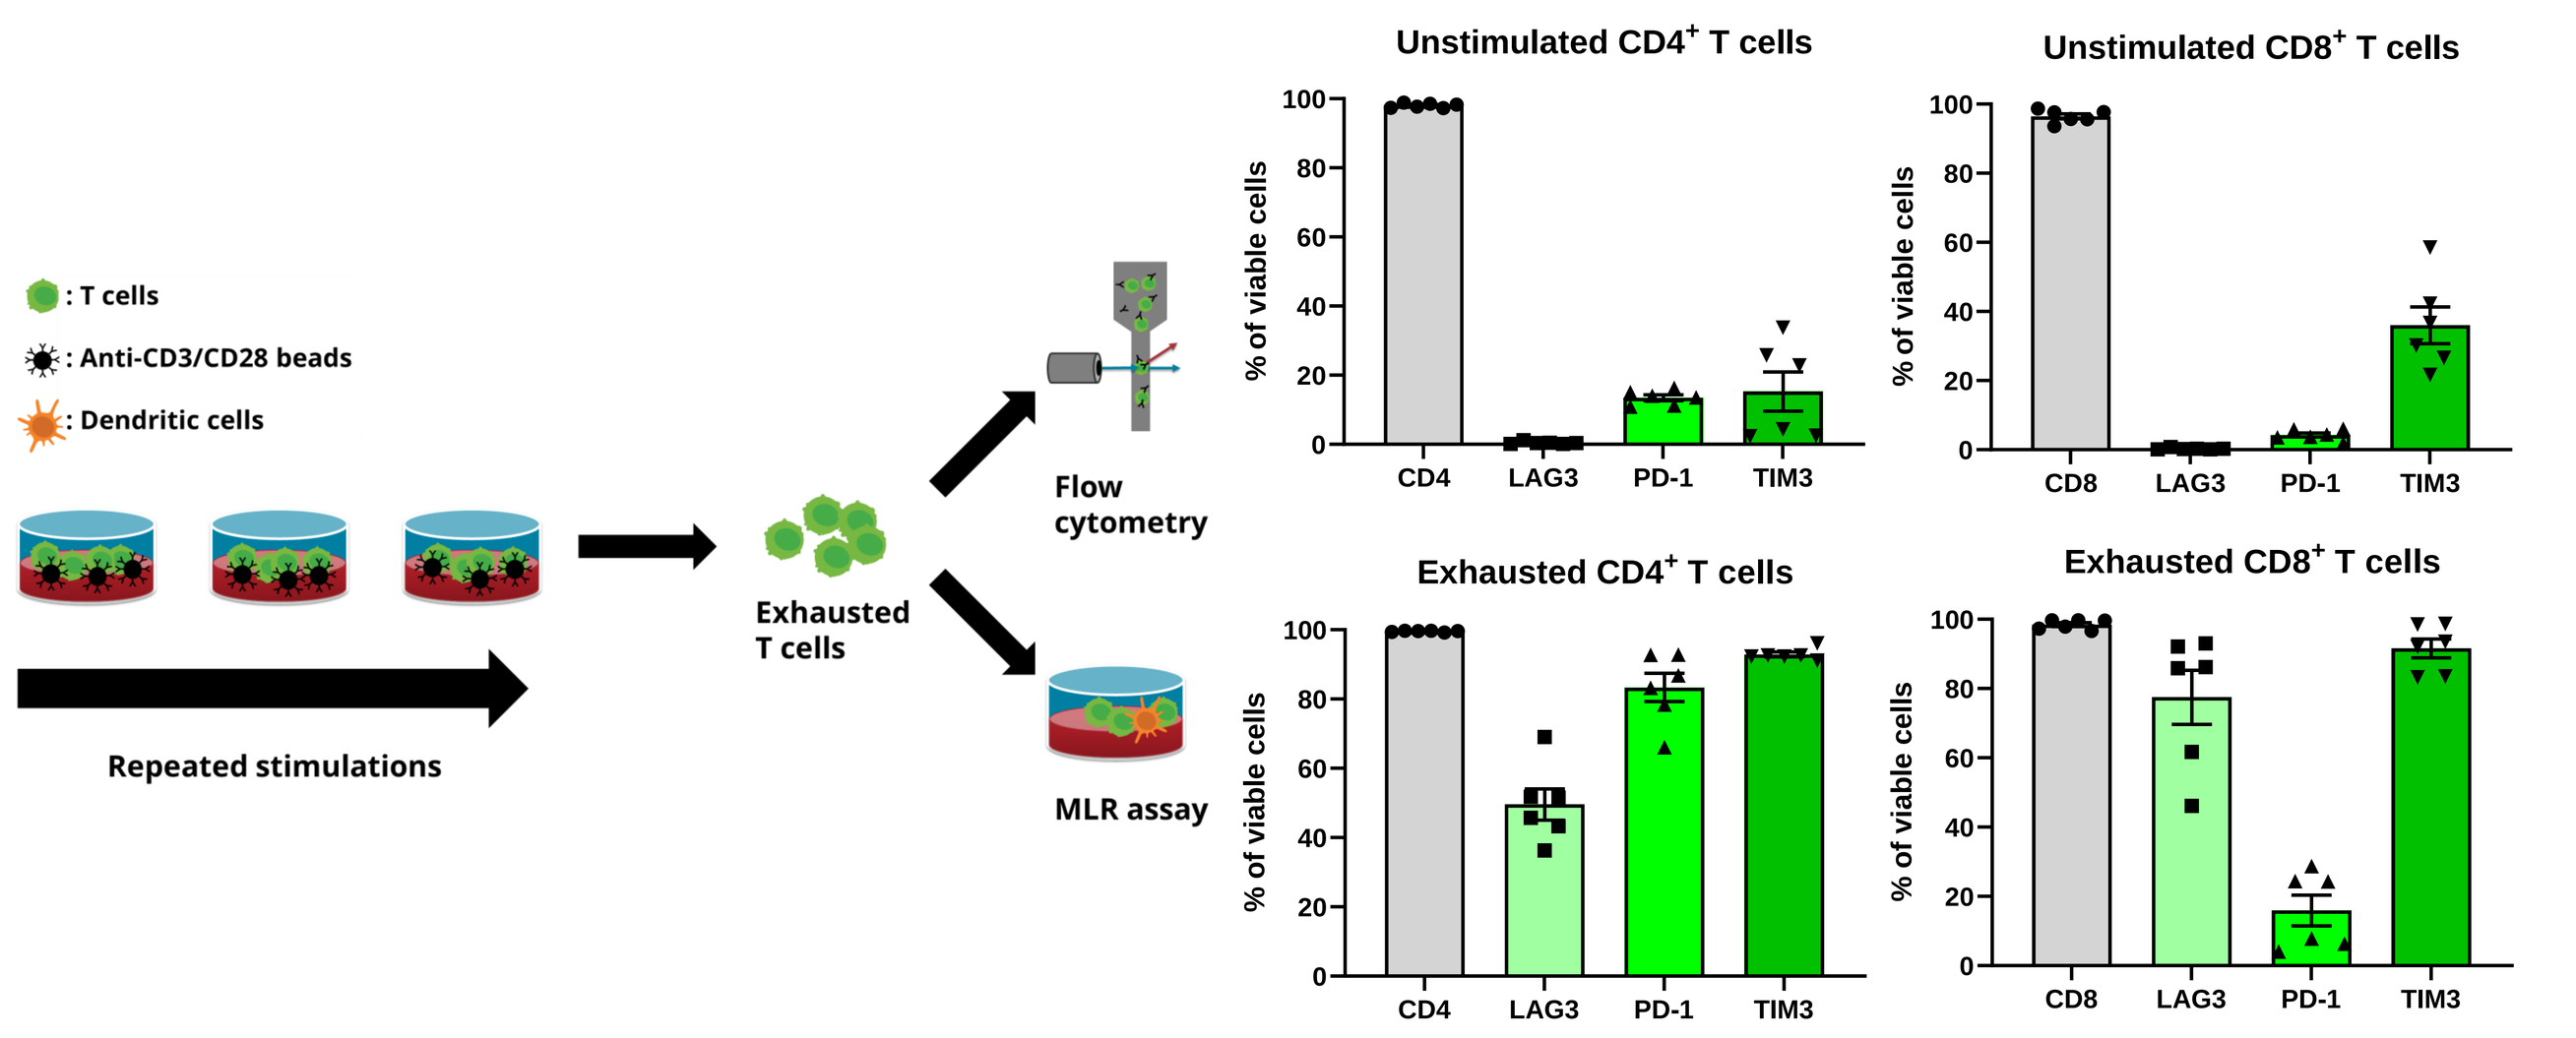
: T cells
: Anti-CD3/CD28 beads
: Dendritic cells
Repeated stimulations
Exhausted T cells
Flow cytometry
MLR assay
Unstimulated CD4+ T cells
Unstimulated CD8+ T cells
Exhausted CD4+ T cells
Exhausted CD8+ T cells
% of viable cells
CD4
CD8
LAG3
PD-1
TIM3

Figure 5. T cell exhaustion was induced by repeated stimulation of either CD4⁺ or CD8⁺ T cells with anti-CD3/CD28 dynabeads for 8 days, as previously described (Dunsford, L.S. et al., 2020, A Human In Vitro T Cell Exhaustion Model for Assessing Immuno-Oncology Therapies. In: Tan, SL. (eds) Immuno-Oncology. Methods in Pharmacology and Toxicology. Humana, New York, NY.). Naïve or exhausted CD4⁺ and CD8⁺ T cells were stained for LAG3, PD-1 and TIM3 and subjected to flow cytometry analysis or MLR assays (below).

ATOR-4066 significantly improves reactivation of exhausted T cells with increasing concentrations of anti-PD-1 in MLR assays

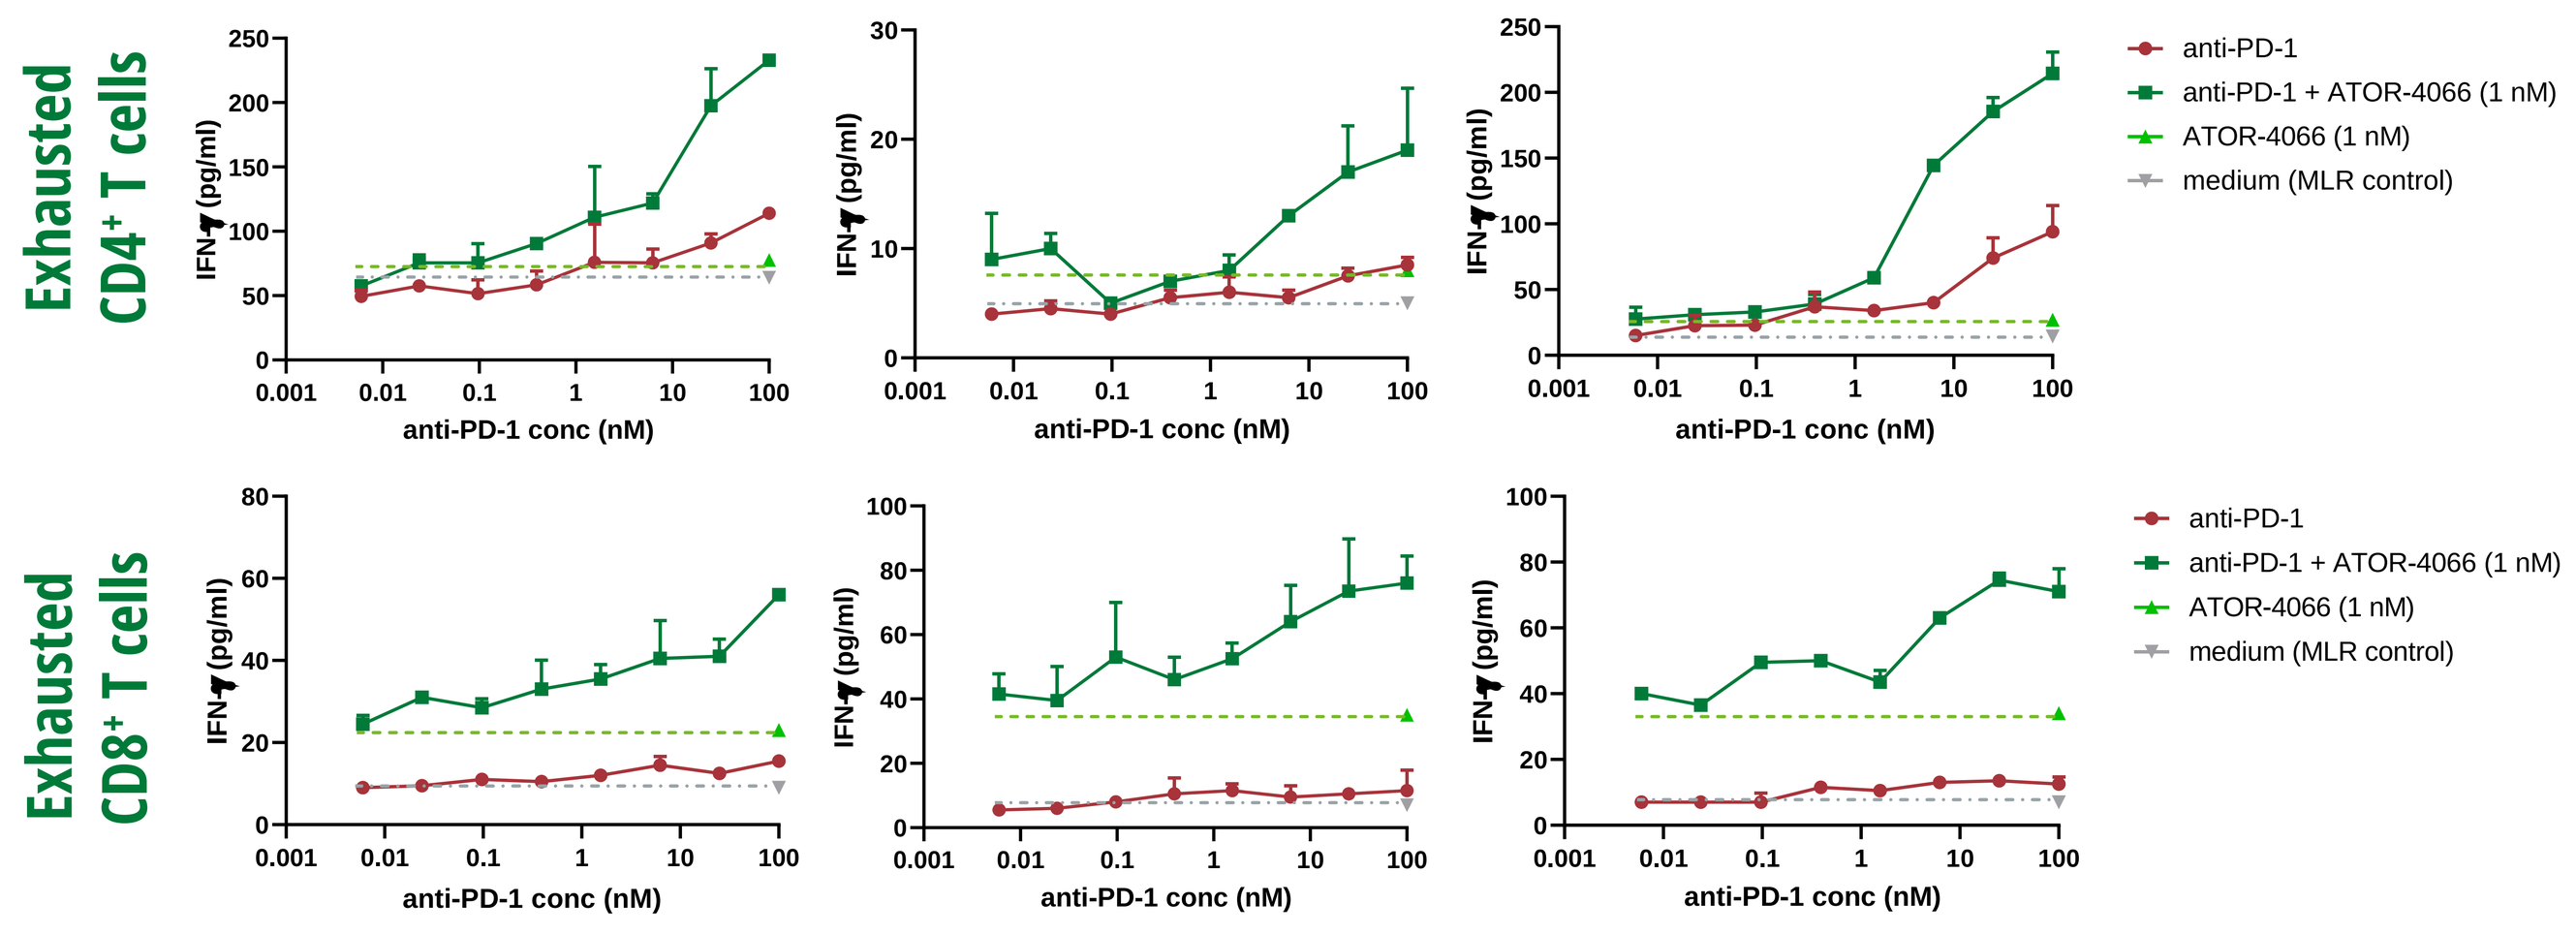
Exhausted CD4+ T cells
Exhausted CD8+ T cells
IFN-γ (pg/ml)
anti-PD-1 conc (nM)
anti-PD-1
anti-PD-1 + ATOR-4066 (1 nM)
ATOR-4066 (1 nM)
medium (MLR control)

Figure 6. Exhausted CD4⁺ or CD8⁺ T cells (100 000 cells/well) were cultured with mature moDCs (10 000 cells/well) from different blood donors and hCEACAM5-coated beads (20 000/well) and stimulated for 7 days with different concentrations of anti-PD-1 (nivolumab) alone or in combination with a fixed concentration of ATOR-4066 (1 nM). At the end of the MLR culture, supernatants were collected, and IFN-γ levels were measured by ELISA (mean±SD). Three different T cell:moDC donor pairs are depicted for exhausted CD4⁺ T cells and CD8⁺ T cells, respectively.

Reactivation of exhausted CD8⁺ T cells by increasing concentrations of ATOR-4066 alone and with anti-PD-1 in MLR assays

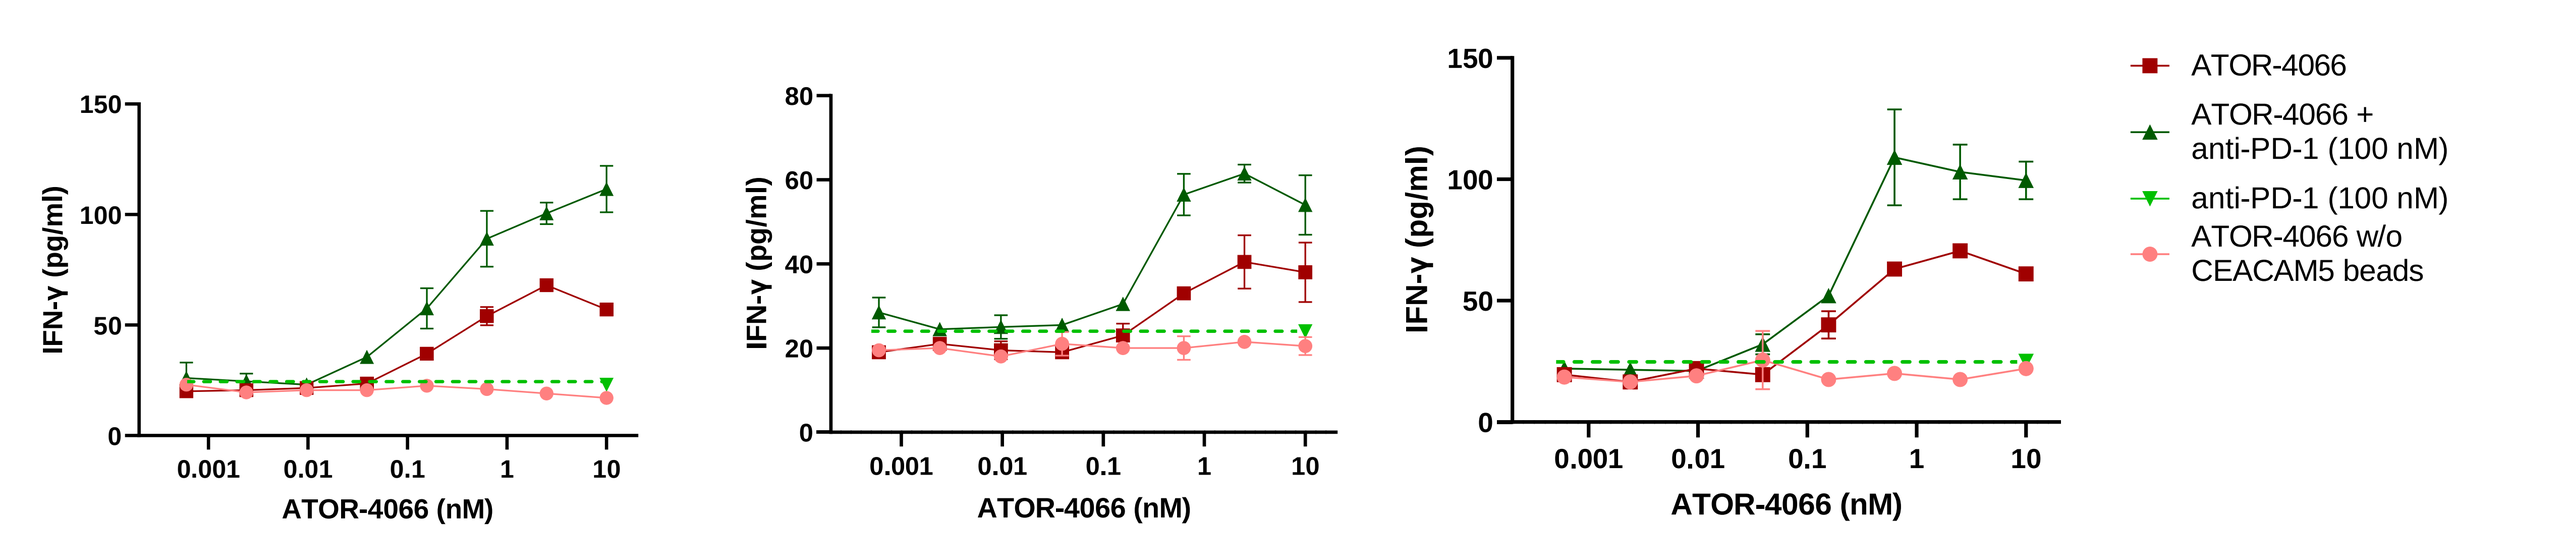
IFN-γ (pg/ml)
ATOR-4066 (nM)
ATOR-4066
ATOR-4066 + anti-PD-1 (100 nM)
anti-PD-1 (100 nM)
ATOR-4066 w/o CEACAM5 beads

Figure 7. Exhausted CD8⁺T cells (100 000 cells/well) were cultured with mature moDCs (10 000 cells/well) from different blood donors and hCEACAM5-coated beads (20 000/well) stimulated for 7 days with different concentrations of ATOR-4066 alone or in combination with a fixed concentration of anti-PD-1 (nivolumab; 100 nM). At the end of the MLR culture, supernatants were collected, and IFN-γ levels were measured by ELISA (mean±SD). Three different T cell:moDC donor pairs are depicted.

Activation of tumor-infiltrating immune cells in human primary gastric cancer tumor samples by ATOR-4066

Activation (CD83 upregulation) of CD40 expressing cell populations

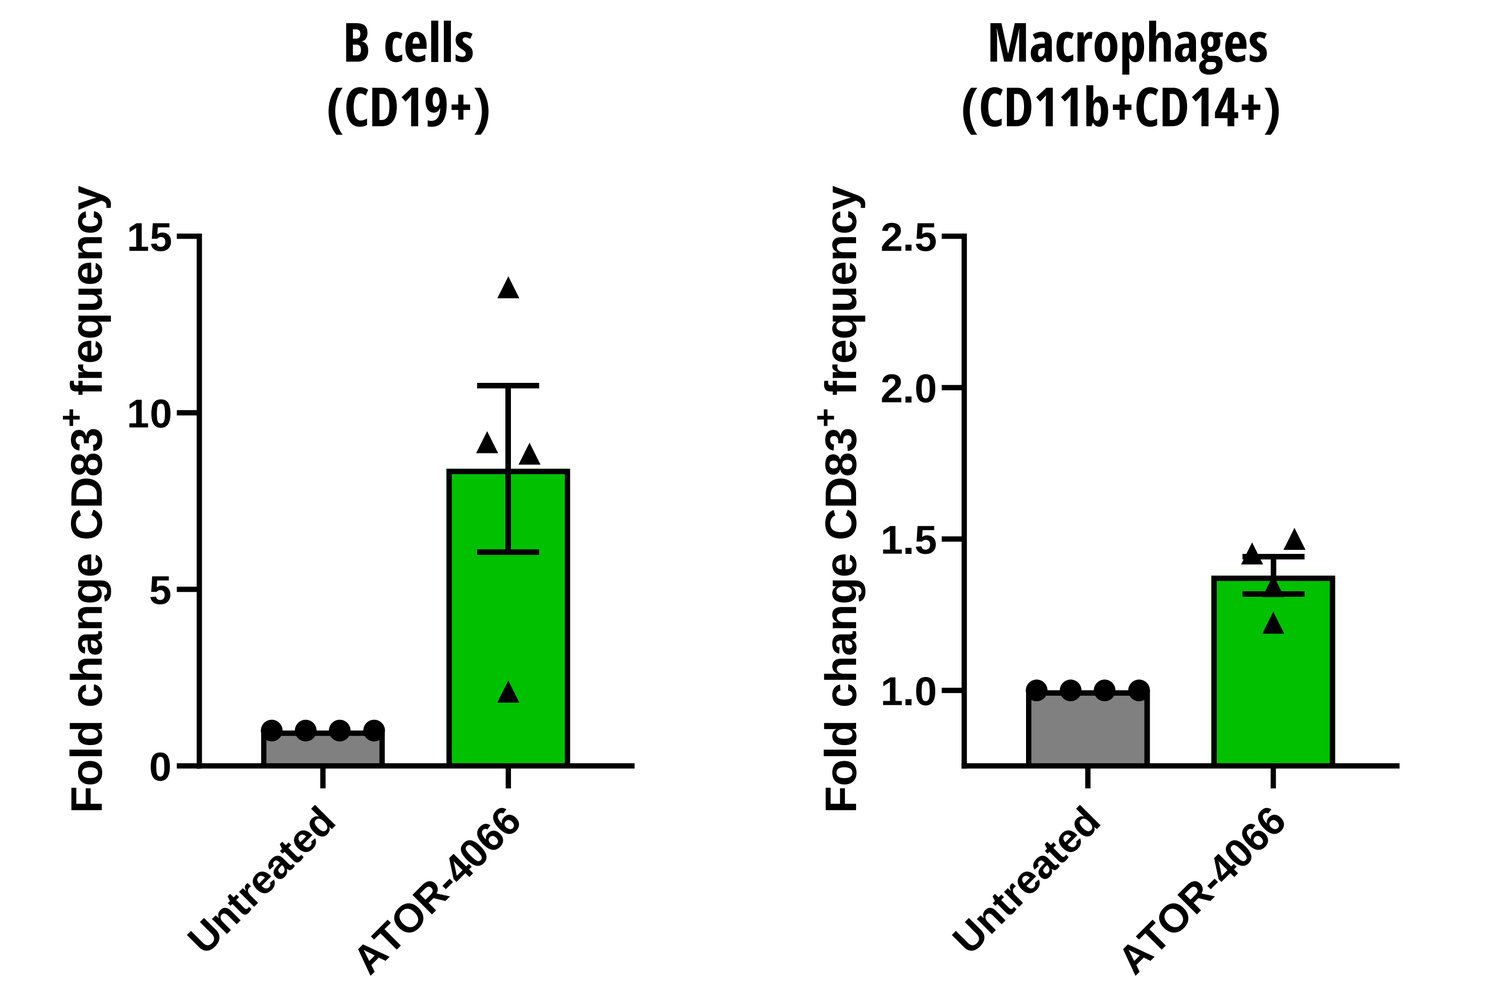
B cells (CD19+)
Macrophages (CD11b+CD14+)
Fold change CD83+ frequency
Untreated
ATOR-4066

Activation (CD25 and Granzyme B expression) of T cells

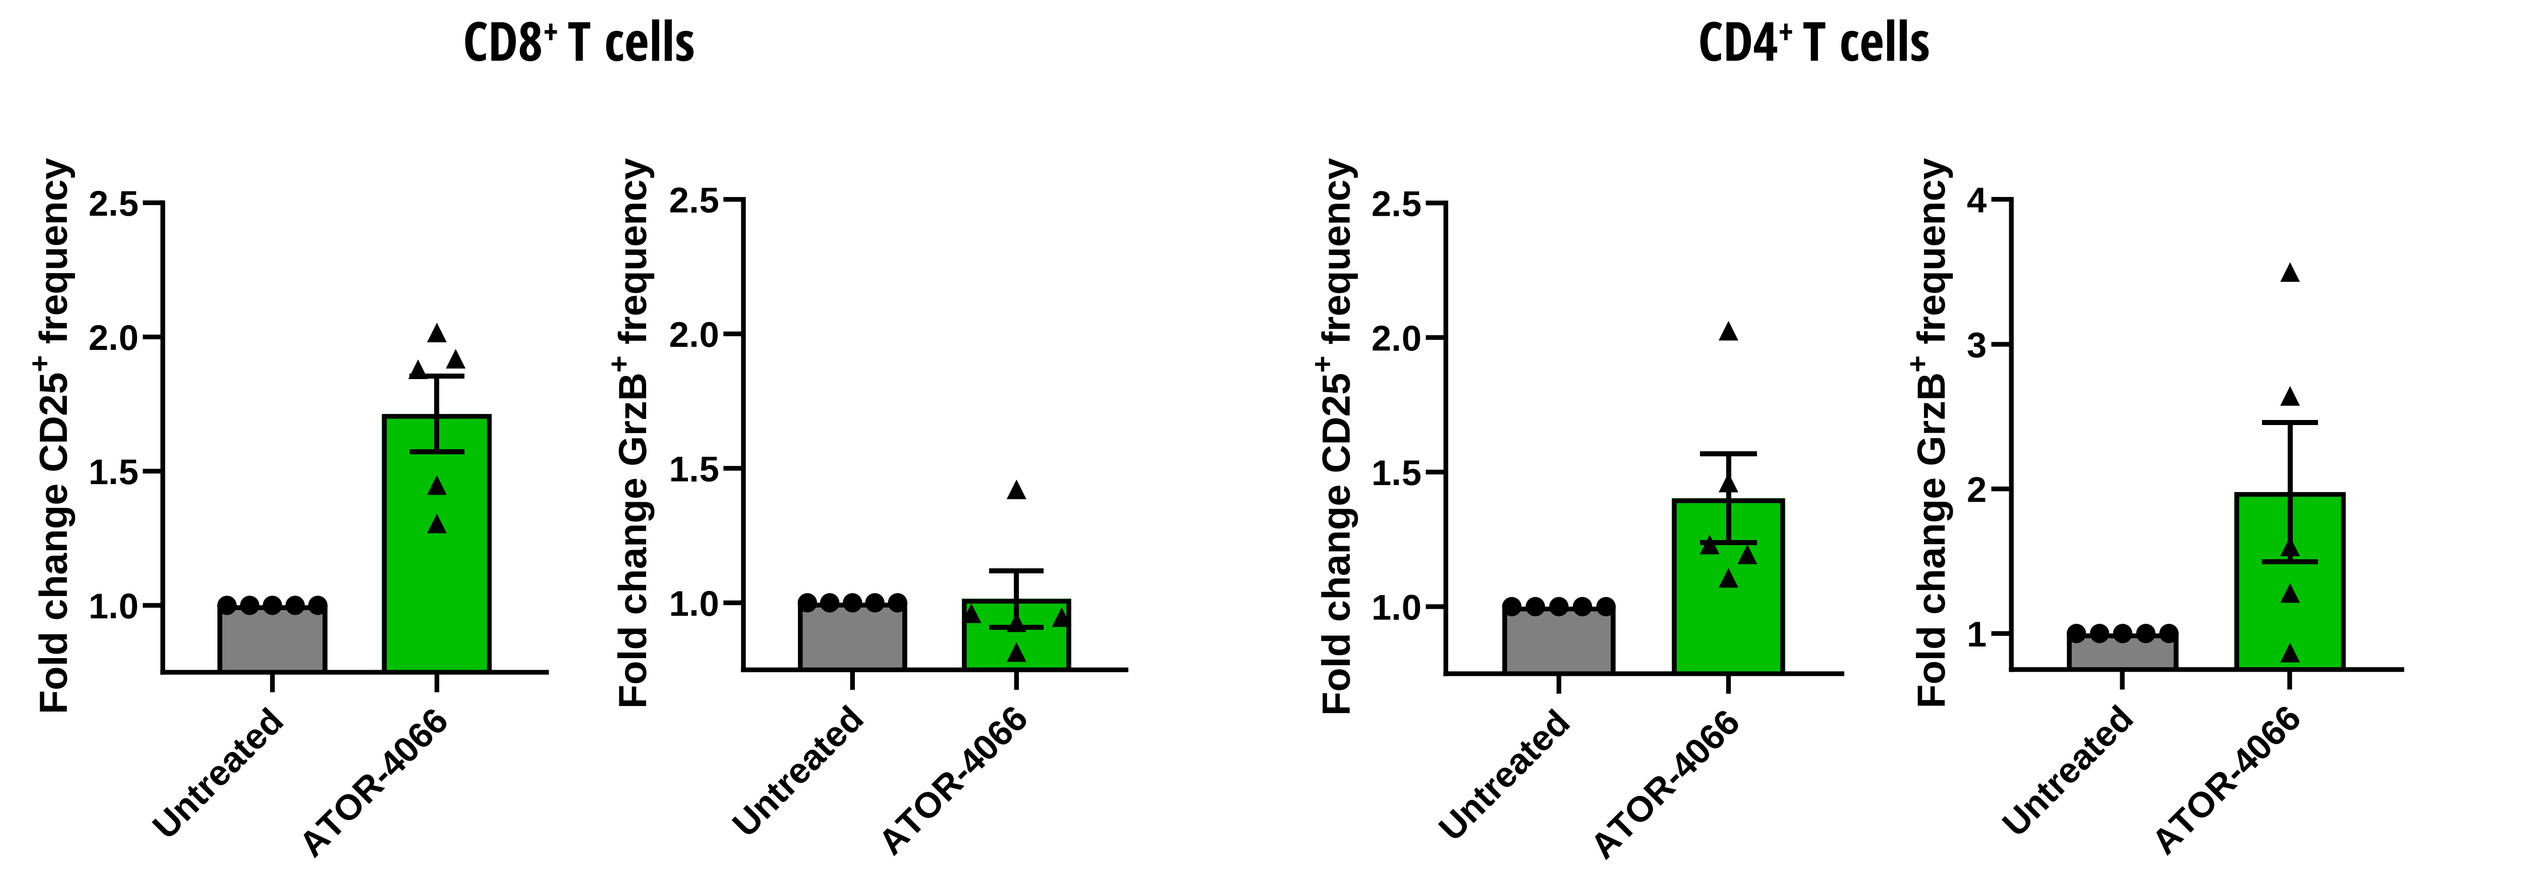
CD8+ T cells
CD4+ T cells
Fold change CD25+ frequency
Fold change GrzB+ frequency
Untreated
ATOR-4066

Figure 8. Dissociated single cells from four or five CEACAM5-positive gastric cancer tumors were analyzed for their ability to activate tumor-infiltrating immune cells following stimulation with ATOR-4066 for 72 hrs. Expression of activation markers on different cell subsets were determined by flow cytometry and depicted as fold change compared to untreated samples. Bars representing mean±SEM

SUMMARY AND CONCLUSION

- We have demonstrated that the CD40xCEACAM5 bispecific antibody ATOR-4066 induces:
 - Efficient, CEACAM5-conditional CD40 activation of human primary B cells and moDCs
 - Co-localization of CEACAM5-expressing tumor debris and CD40-expressing APCs
 - Activation of tumor-infiltrating immune cells in patient-derived dissociated tumor samples from CEACAM5+ gastric cancer patients
 - Strong anti-tumor efficacy and induction of immunological memory, also in large tumors with heterogenous CEACAM5 expression
 - Capacity to reactivate exhausted CD4 and CD8 T cells *in vitro*
 - Enhanced effect to reactivate exhausted T cells in combination with anti-PD1
- Taken together, these data show the ability of ATOR-4066 to remodel the immune microenvironment and activate tumor-infiltrating immune cells from primary human tumors expressing CEACAM5, demonstrating the promise of this new candidate drug and strongly supports further development towards the clinic.

SITC 2023 Poster

Hägerbrand et al. JITC 2022

*Corresponding author
idu@alligatorbioscience.com

ALLIGATOR bioscience