Mitazalimab, a potent CD40 agonist in combination with FOLFIRINOX demonstrates changes consistent with increased immune activation in TME and peripheral blood in a preclinical pancreatic cancer tumor model

David Gomez Jimenez, Mia Thagesson, Anneli Nilsson, Y. Pico de Coaña, S. Ambarkhane, Karin Enell Smith, and Peter Ellmark; Alligator Bioscience AB, Lund, Sweden.

- Mitazalimab is a CD40-targeting FcγR-conditional agonistic monoclonal IgG1 antibody.
- Recently, mitazalimab has shown positive results in a Phase 1b/2 trial (Optimize-1, NCT04888312) in combination with mFOLFIRINOX (mFFX), demonstrating clinically relevant survival benefits in 1st line pancreatic cancer.
- Optimize-1 top-line readout demonstrated an Objective Response Rate of 40.4%, meeting primary endpoint and confirming the benefit of mitazalimab in combination with mFFX.
 - Median Overall Survival of 14.3 months at the time of analysis.
 - Median Duration of Response of 12.5 months, compared to the standard of care mFFX of 5.9 months.

Objectives:

- Investigate the anti-tumor efficacy in pancreatic tumor bearing, human CD40 transgenic (hCD40tg) mice treated with mitazalimab and FOLFIRINOX.
- Assess immune-related changes after the first, tumor priming, and second, T-cell priming, mitazalimab doses in blood, tumor and spleen of KPCY tumor bearing mice.

Mitazalimab controls pancreatic tumor growth

Mice treated with mitazalimab, displayed enhanced tumor volume control compared to vehicle group. In turn, mitazalimab's capacity to control tumor growth synergized with FOLFIRINOX, further reducing tumor volume as compared to vehicle and single agent groups. The tumor priming dose was safe and well tolerated.

A

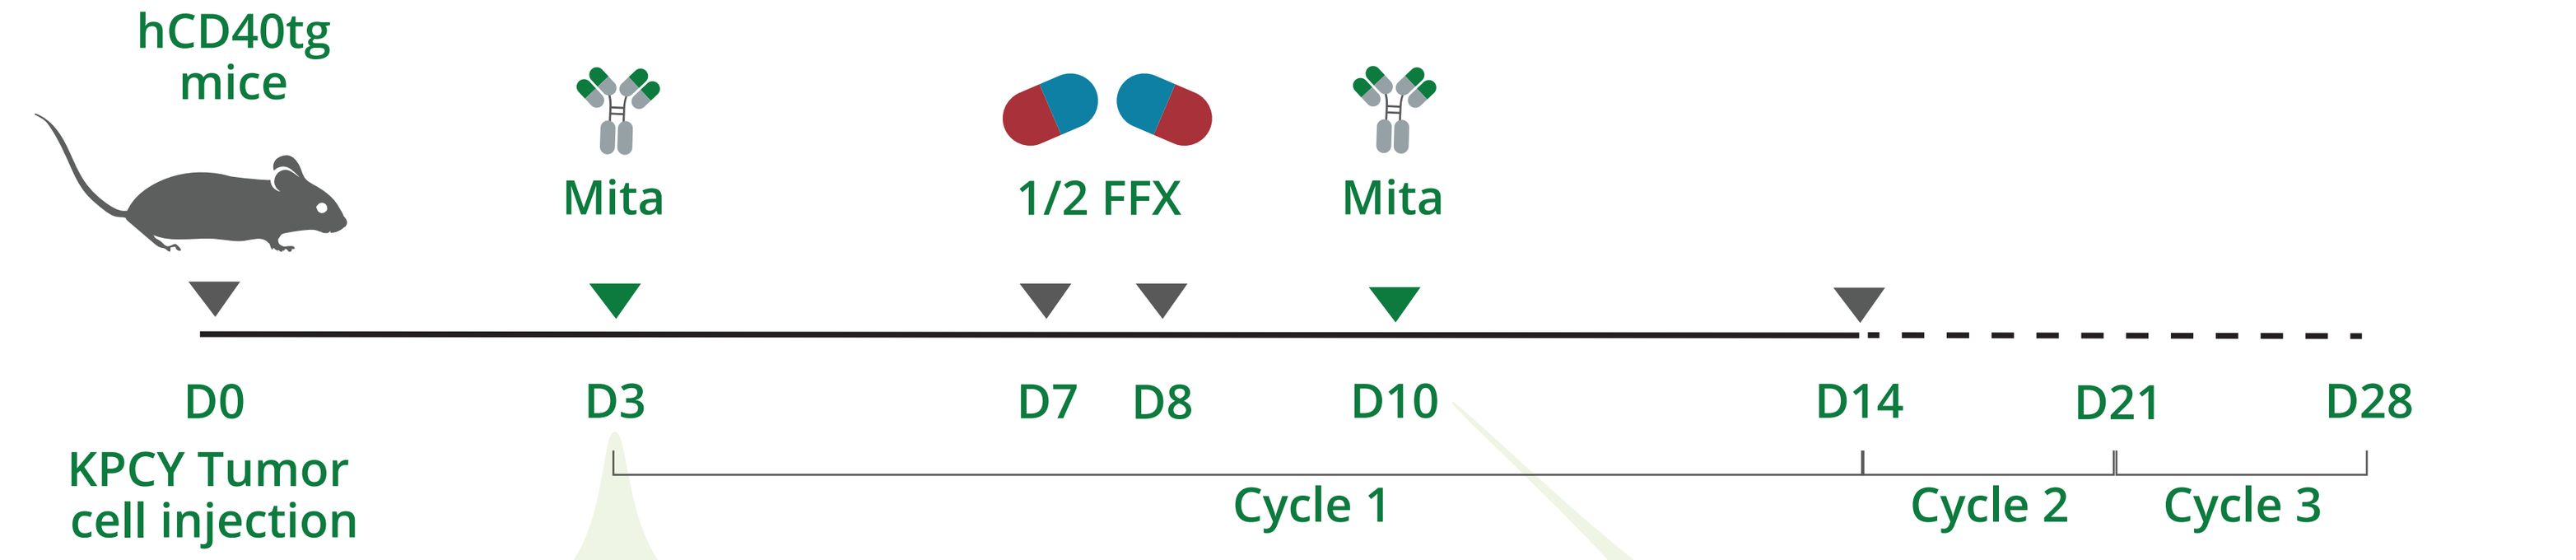
hCD40tg mice
Mita
1/2 FFX
Mita
D0
D3
D7
D8
D10
D14
D21
D28
KPCY Tumor cell injection
Cycle 1
Cycle 2
Cycle 3

B

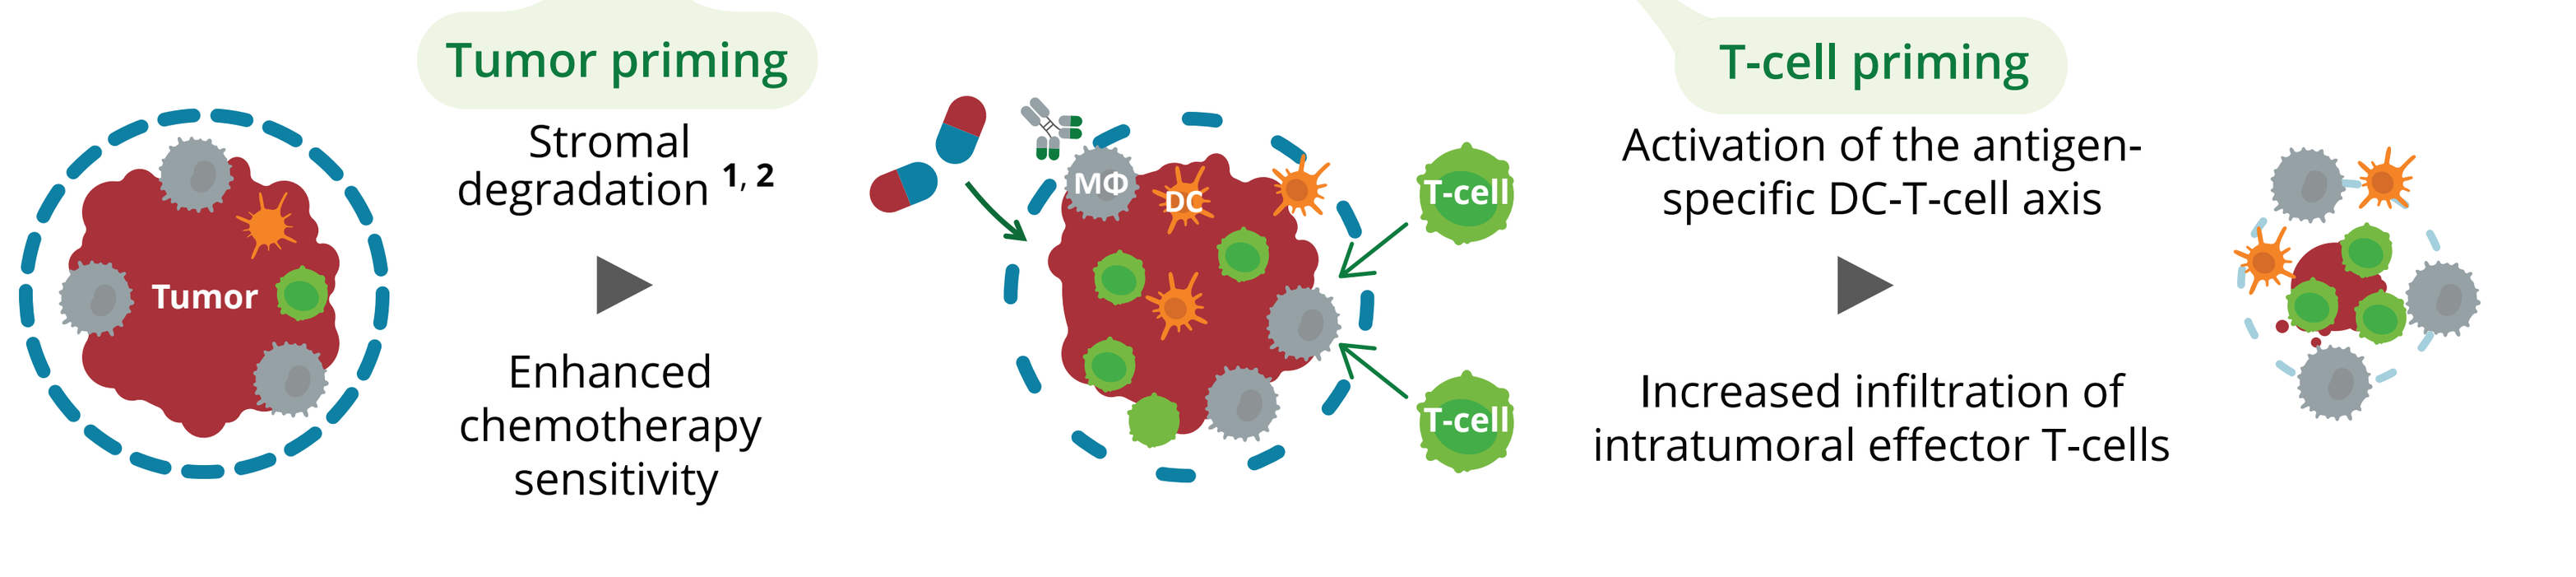
Tumor priming
Tumor
Stromal degradation 1, 2
Enhanced chemotherapy sensitivity
MΦ
DC
T-cell
T-cell
T-cell priming
Activation of the antigen-specific DC-T-cell axis
Increased infiltration of intratumoral effector T-cells

C

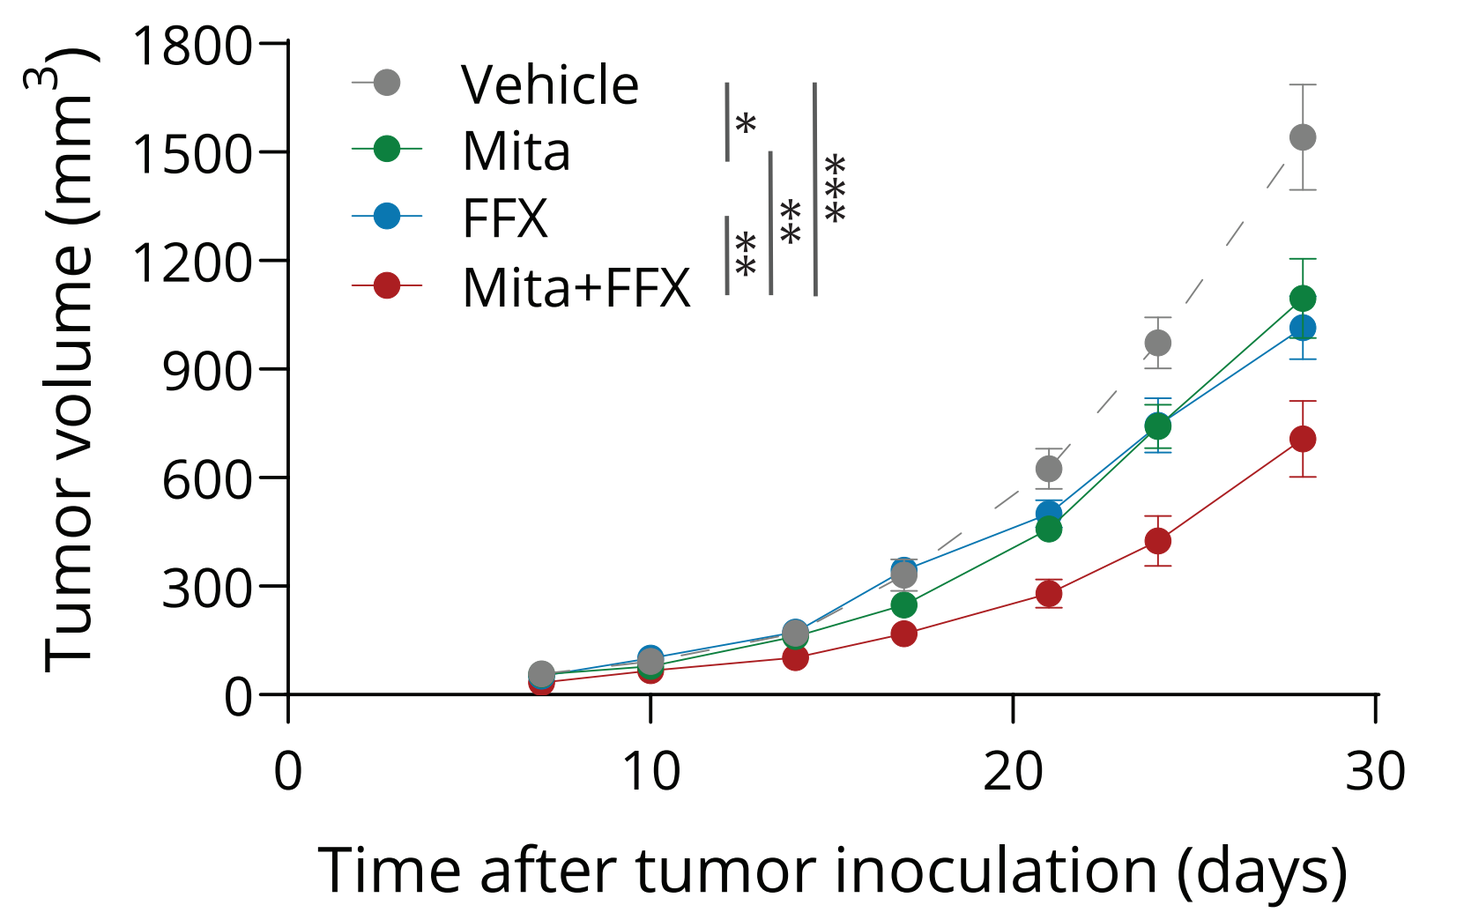
Tumor volume (mm3)
1800
1500
1200
900
600
300
0
0
10
20
30
Time after tumor inoculation (days)
Vehicle
Mita
FFX
Mita+FFX
*
**
**

Figure 1. (A) hCD40tg mice were inoculated subcutaneously with KPCY tumor cells (clone 2838c3, Trp53^{R172H}, KRAS^{G12D})[3] re-suspended in matrigel (n=40, 10/group). Mice were treated with vehicle (Dextrose), FOLFIRINOX (FFX; days 7-8, 14-15 and 21-22) and/or mitazalimab (Mita; 3, 10, 17 and 24). **(B)** Mode of action of mitazalimab in combination with FFX. **(C)** Anti-tumor efficacy of mice treated with mita, FFX or mita + FFX. Differences in tumor volume were calculated at day 21 using Mann-whitney test. * p < 0.05, ** p < 0.01 and, *** p < 0.005. MΦ: Macrophage; DC: Dendritic cell.

Conclusions:

- Mitazalimab in combination with FOLFIRINOX controls tumor growth in a mouse model challenged with the translationally relevant pancreatic tumor KPCY.
- The tumor priming dose of mitazalimab induced myeloid cell infiltration in the tumor microenvironment along with peripheral T/NK cell expansion in lymphoid tissue favouring effector T-cell movilization into circulation.
- The T-cell priming dose of mitazalimab induced intra-tumoral effector CD8⁺ T cell infiltration, and sustained effector T-cell expansion in lymphoid organs, consistent with mitazalimab's ability to mediate long term anti-tumor immunity.
- This study provides support to the unique design of the OPTIMIZE-1 Phase 2 study in first line metastatic pancreatic cancer and provides a mechanistic explanation to the unprecedented Duration of Response and Overall Survival observed in the study.

Tumor priming dose - Mitazalimab induces peripheral expansion of the T-cell compartment along with myeloid activation to sensitize the tumor

A single dose of mitazalimab elicits effector T cell differentiation in peripheral blood and lymphoid tissue.

A heterogeneous **intratumoral** infiltration of monocytic cells is seen in response to mitzalimab

In **blood**, mitazalimab induces T effector maturation and trafficking. Mitazalimab also boosts the cytotoxic profile of T/NK cells as evidenced by the upregulation of GzmB and Ki67

Mitazalimab induces T/NK cell proliferation in the **spleen**, followed by the egression of effector T-cells into blood.

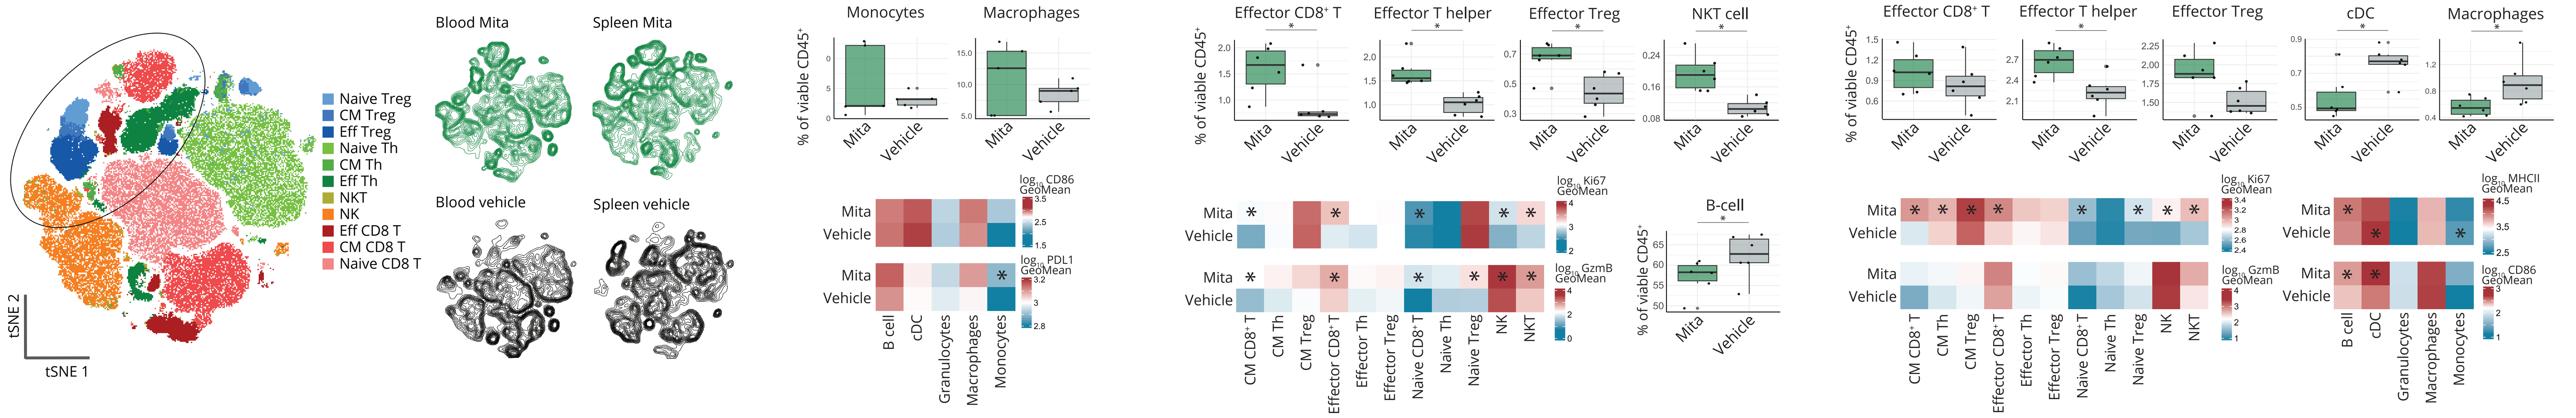
Naive Treg
CM Treg
Eff Treg
Naive Th
CM Th
Eff Th
NKT
NK
Eff CD8 T
CM CD8 T
Naive CD8 T
tSNE 1
tSNE 2
Blood Mita
Spleen Mita
Blood vehicle
Spleen vehicle
Monocytes
Macrophages
% of viable CD45+
Mita
Vehicle
log10 CD86 GeoMean
log10 PDL1 GeoMean
B cell
cDC
Granulocytes
Macrophages
Monocytes
Effector CD8+ T
Effector T helper
Effector Treg
NKT cell
B-cell
log10 Ki67 GeoMean
log10 GzmB GeoMean
CM CD8+ T
CM Th
CM Treg
Effector CD8+ T
Effector Th
Effector Treg
Naive CD8+ T
Naive Th
Naive Treg
NK
NKT
cDC
log10 MHCII GeoMean

Figure 2. Mitazalimab-induced effects 96 hours after tumor primng dose (n=24, 6/group and FC panel). Blood, spleen and tumor were dissociated to single cell suspension and analysed by flow cytometry using a T/NK or an antigen presenting cell directed-panels, on a FACS symphony instrument. tSNE plots were generated to visually compare differences in T/NK cell lineages accross treatments and organs, using the fast interpolation based tSNE method. For statistical comparison, data was tested for normality and groups were compared either using unpaired t-test or ManWhitney test. P-values were adjusted using FDR method (* p < 0.05). Heatmaps of activation markers were generated by averaging their geometric mean, followed by log10-transformation. No significant differences were found for PD1 in either of the tissues. No differences were found in tdLN (data not shown). Mita: Mitazalimab; FFX: FOLFIRINOX; cDC: conventional dendritic cell; CM: Central memory; Th: helper T-cell ; Treg: regulatory T-cell; NK: Natural killer; NKT: Natural killer T-cell.

T-cell priming dose - Mitazalimab evokes robust effector CD8⁺ T and T helper intratumoral infiltration suggesting long lasting anti-tumor immunity

Mitazalimab increases the production of effector T cells in peripheral lymphoid organs, favouring trafficking and intratumoral infiltration. In combination with FFX, mita further polarizes naive into effector CD8⁺ T cells.

Mitazalimab, with/out FFX, boosts infiltration of **intratumoral** effector CD8⁺ T-cells with increased cytotoxic and proliferative capacity.

- Upregulation PD1 by effector Thelper and increased B cell infiltration suggests formation of TLS
- Mitazalimab downregulation of PD1 by effector Tregs might hinder they supressive activity

In **blood**, mitazalimab induces proliferation of CD8⁺ T, and T helper cells, boosts GzmB production by cytotoxic cells, and induces PD1 upregulation in T helper, NK and NKT cells.

Repeated dosage of mitazalimab sustains T-cell proliferation and differentiation of effector CD8⁺ and T helper cells in the **spleen**, based on GzmB and PD1 upregulation, respectively.

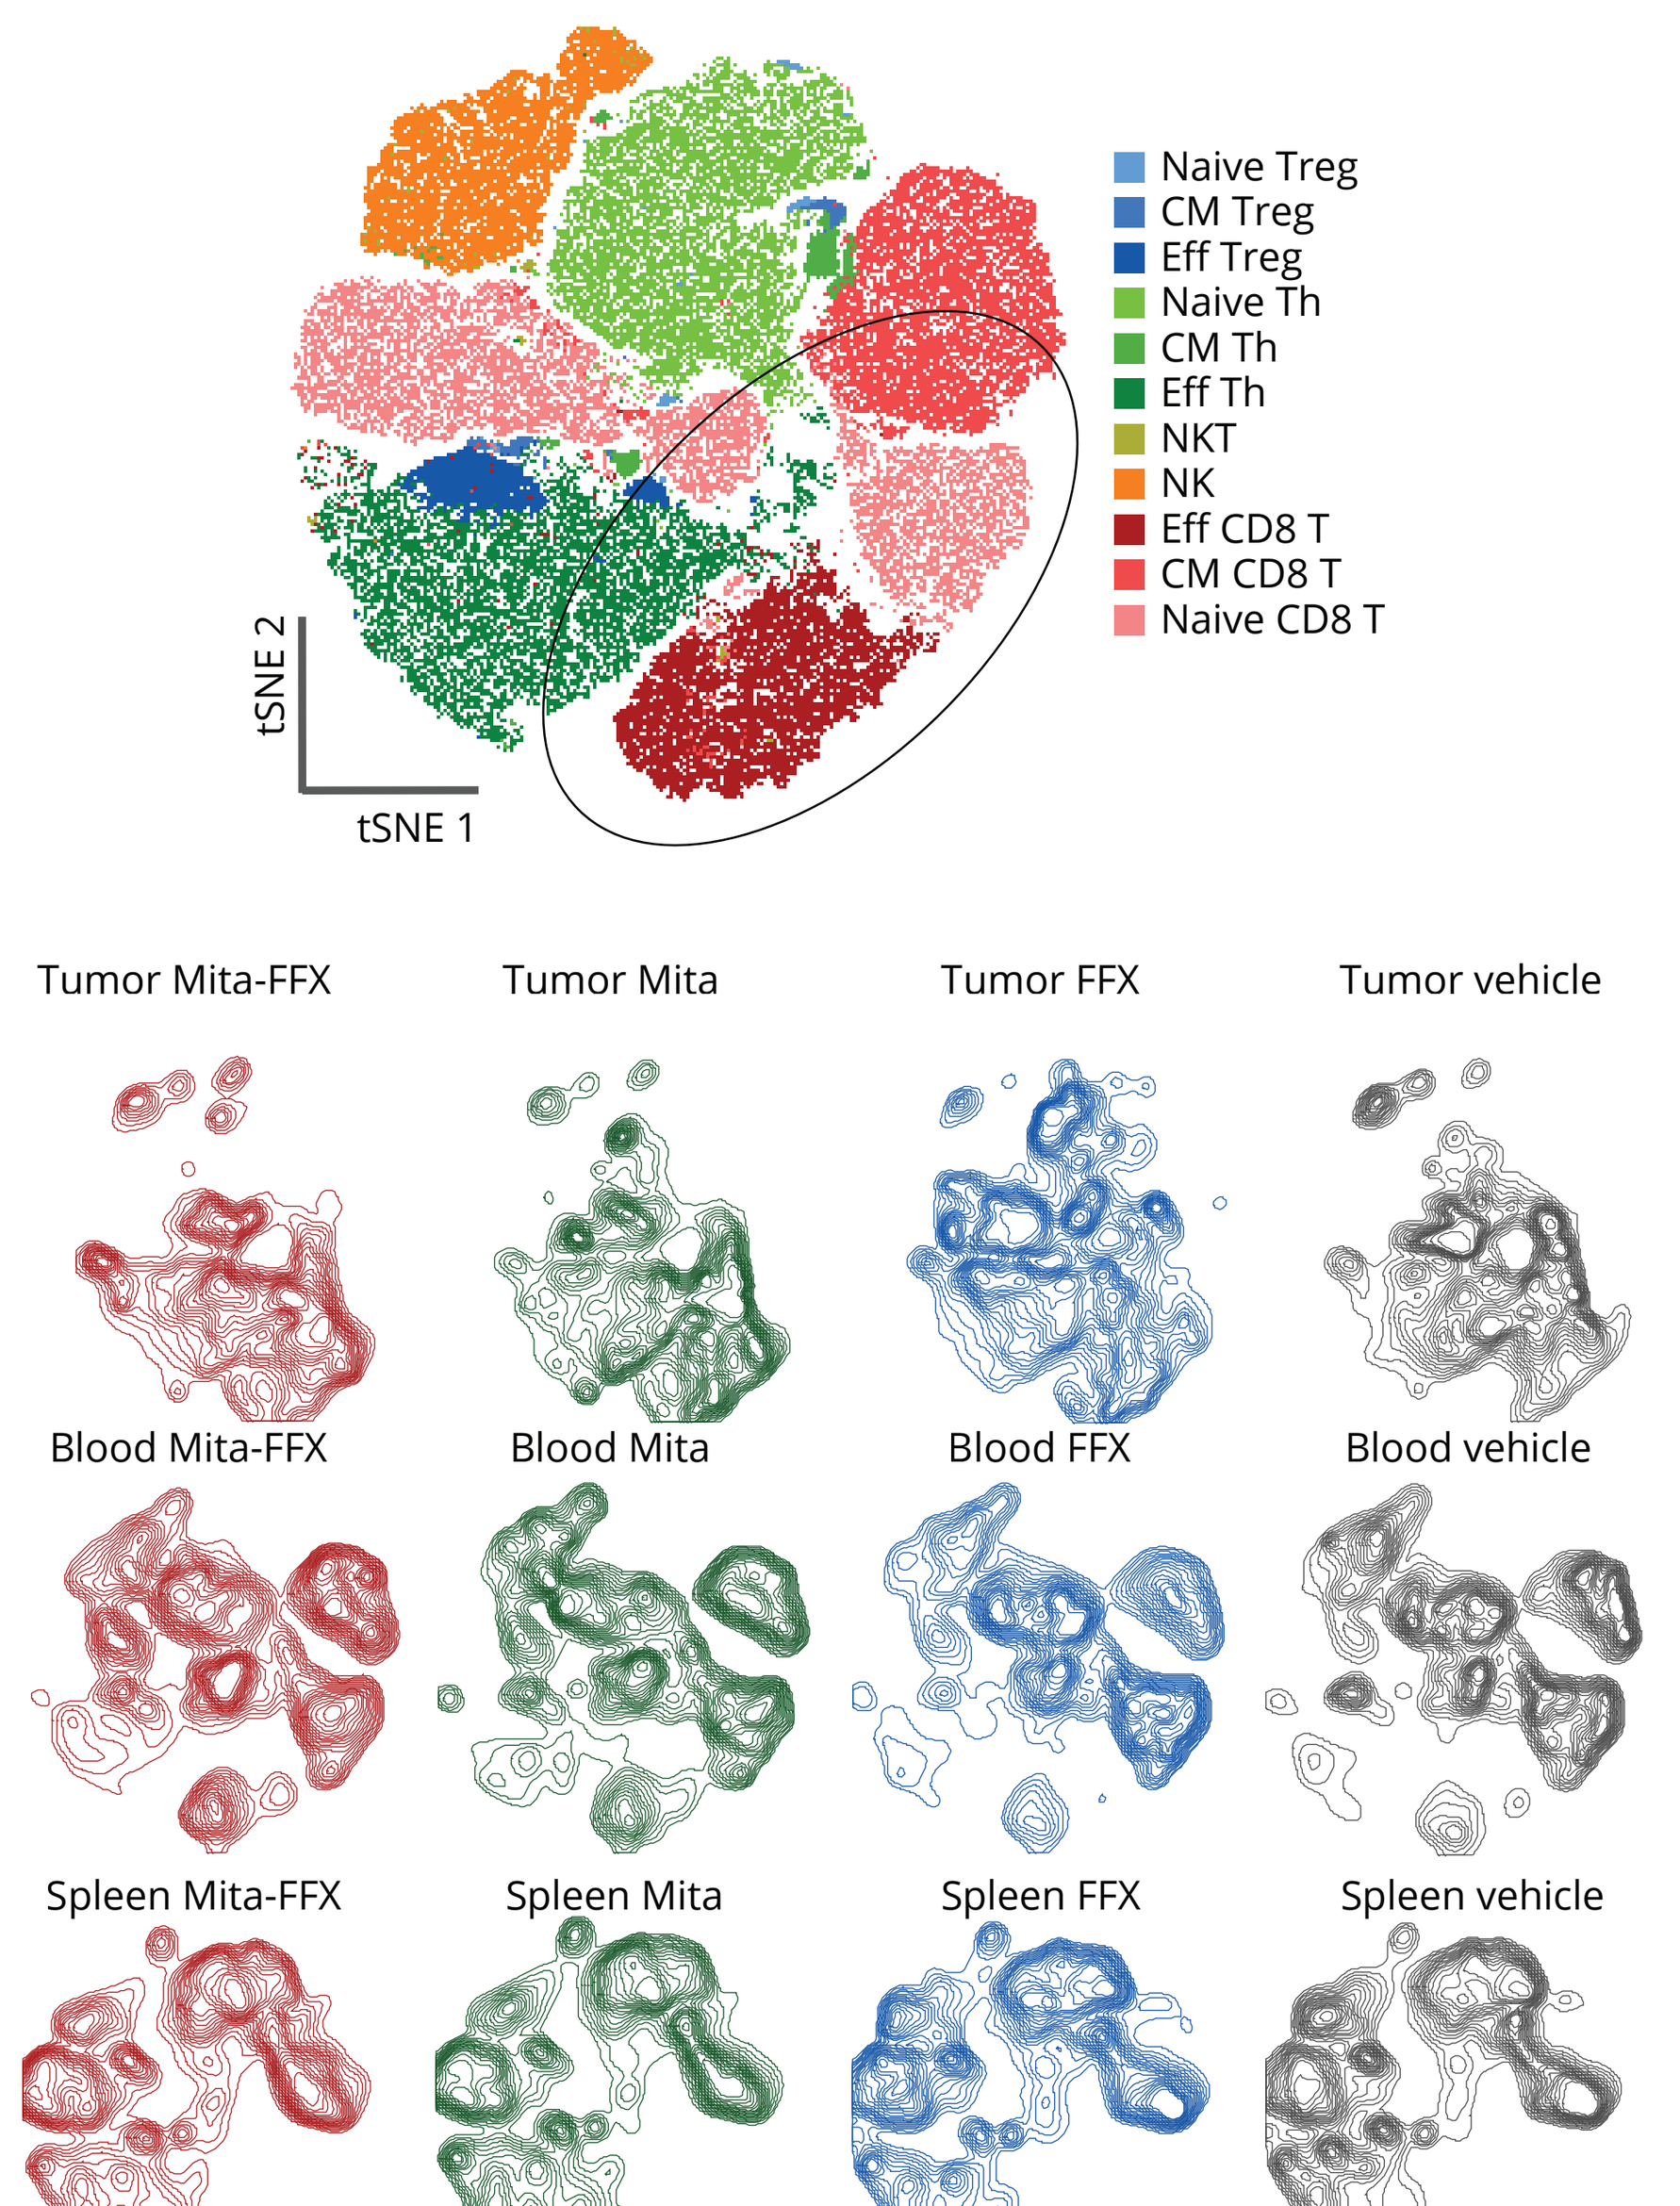
Naive Treg
CM Treg
Eff Treg
Naive Th
CM Th
Eff Th
NKT
NK
Eff CD8 T
CM CD8 T
Naive CD8 T
tSNE 1
tSNE 2
Tumor Mita-FFX
Tumor Mita
Tumor FFX
Tumor vehicle
Blood Mita-FFX
Blood Mita
Blood FFX
Blood vehicle
Spleen Mita-FFX
Spleen Mita
Spleen FFX
Spleen vehicle

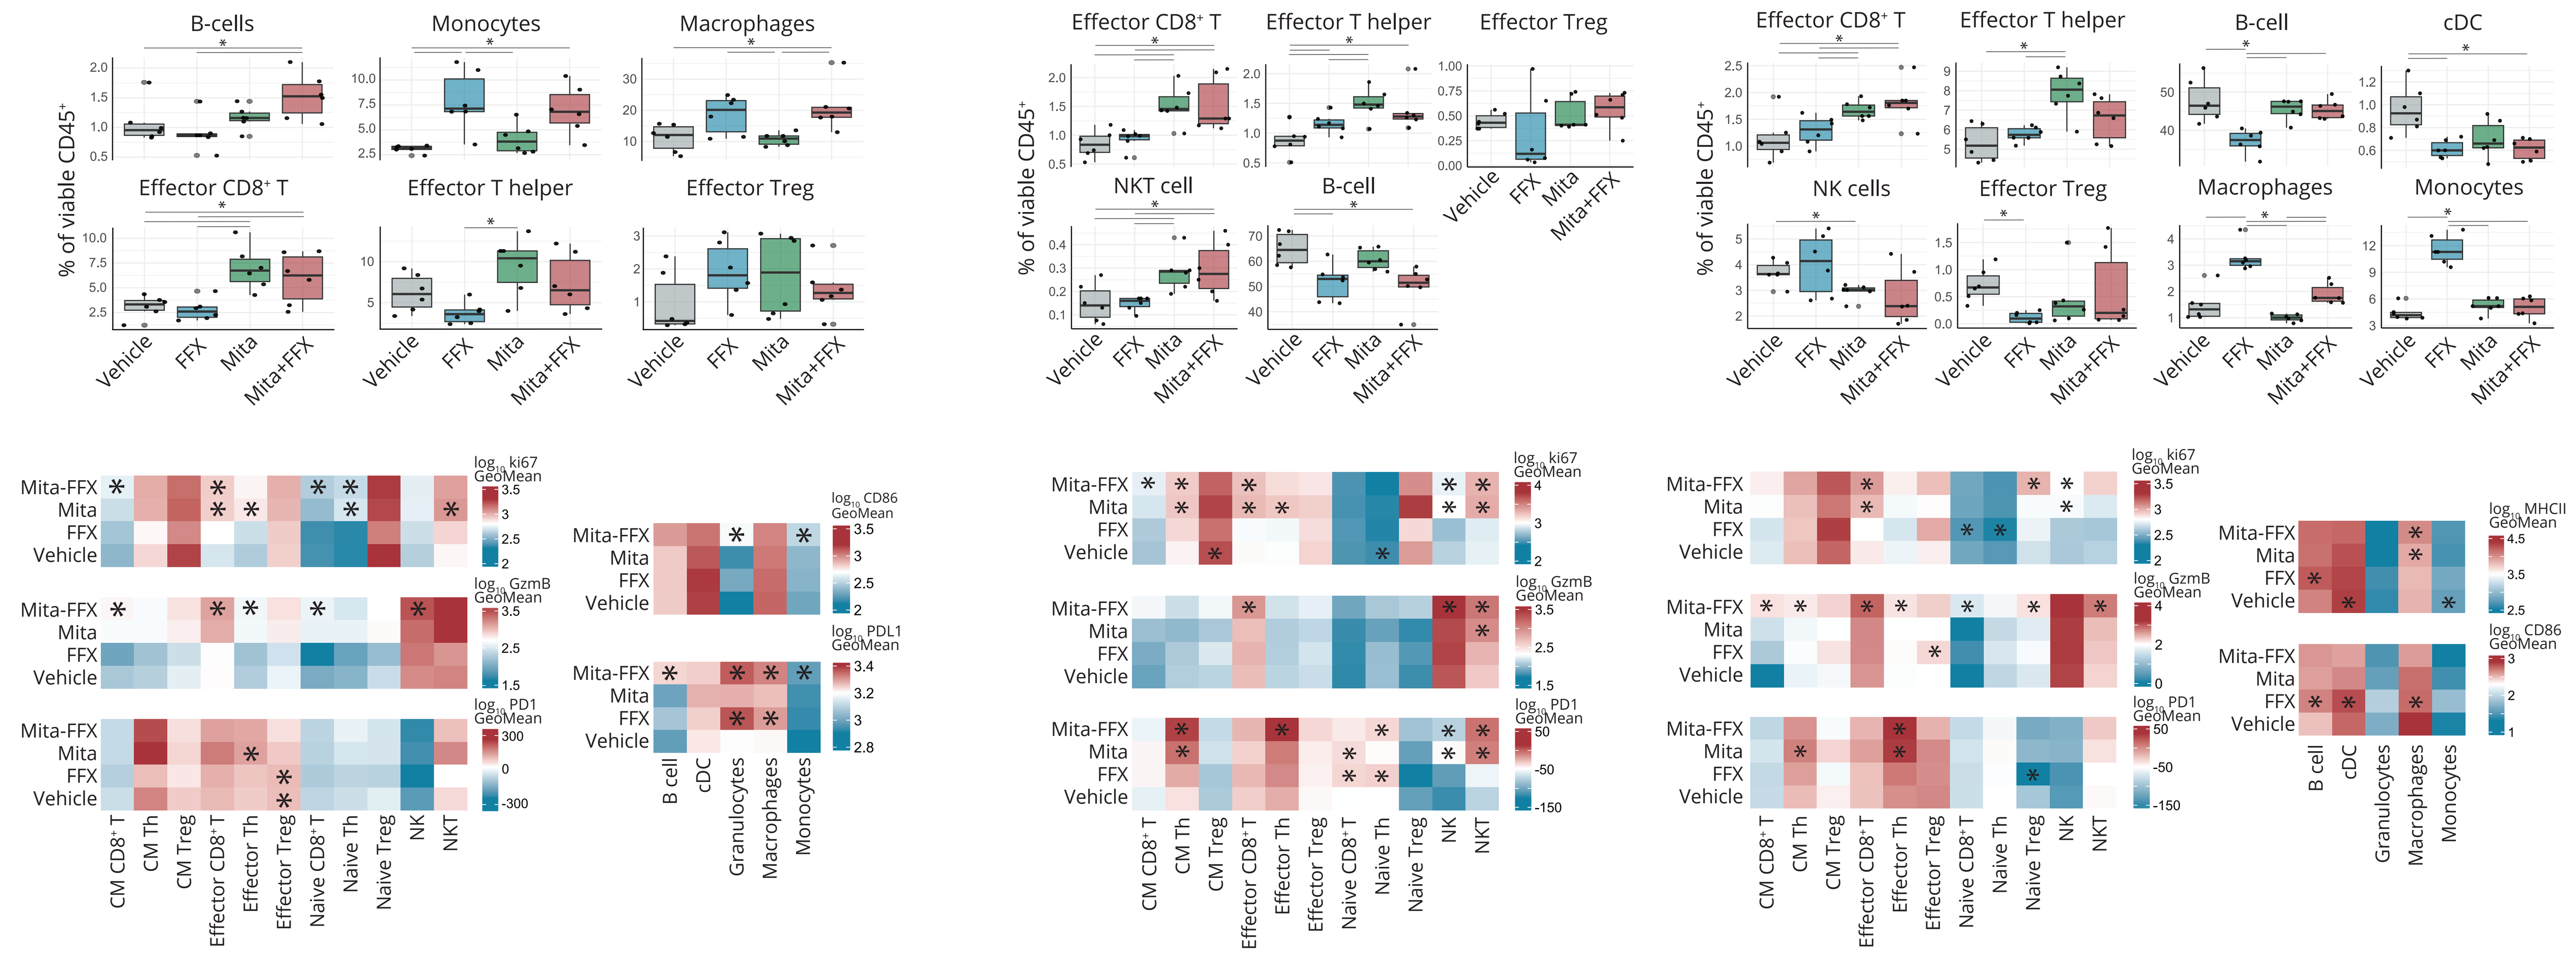
B-cells
Monocytes
Macrophages
Effector CD8+ T
Effector T helper
Effector Treg
NKT cell
B-cell
NK cells
cDC
% of viable CD45+
Vehicle
FFX
Mita
Mita+FFX
Mita-FFX
log10 ki67 GeoMean
log10 GzmB GeoMean
log10 PD1 GeoMean
log10 CD86 GeoMean
log10 PDL1 GeoMean
log10 MHCII GeoMean
CM CD8+ T
CM Th
CM Treg
Effector CD8+ T
Effector Th
Effector Treg
Naive CD8+ T
Naive Th
Naive Treg
NK
NKT
B cell
cDC
Granulocytes
Macrophages
Monocytes

Figure 3. Treatment-induced effects 96 hours after the T-cell priming dose of mitazalimab (n=48, 6/group and FC panel). Blood, spleen and tumor were dissociated to single cell suspension and analysed by flow cytometry using a T/NK or an antigen presenting cell directed-panels, on a FACS symphony instrument. tSNE plots were generated to visually compare differences in T/NK cell lineages accross treatments and organs, using the fast interpolation based tSNE method. For statistical comparison, data was tested for normality and groups were compared either using unpaired t-test or ManWhitney test. P-values were adjusted using FDR method (* p < 0.05). Heatmaps of activation markers were generated by averaging their geometric mean, followed by log10-transformation. Heatmaps only inlcude comparisons of treatment groups to vehicle group. Mita: Mitazalimab; FFX: FOLFIRINOX; cDC: conventional dendritic cell; MacroGeoMean: Geometric mean; CM: Central memory; Th: helper T-cell ; Treg: regulatory T-cell; NK: Natural killer; NKT: Natural killer T-cell.

References

1. Beatty, G.L., et al. CD40 agonists alter tumor stroma and show efficacy against pancreatic carcinoma in mice and humans. Science. 2011 Mar 25;331(6024):1612-6.
2. Long K.B., et al. IFNγ and CCL2 Cooperate to Redirect Tumor-Infiltrating Monocytes to Degrade Fibrosis and Enhance Chemotherapy Efficacy in Pancreatic Carcinoma. Cancer Discov. 2016 Apr;6(4):400-413.
3. Li, J., et al. Tumor Cell-Intrinsic Factors Underlie Heterogeneity of Immune Cell Infiltration and Response to Immunotherapy. Immunity, 2018. 49(1): p. 178-193 e7.

IAACR 20204
Session
PO.CL06.07
Antibodies 1

ALLIGATOR bioscience