OPTIMIZE-1 primary analysis: Safety, efficacy and biomarker results of a phase 1b/2 study combining CD40 agonist mitazalimab with mFOLFIRINOX in previously untreated metastatic pancreatic ductal adenocarcinoma (mPDAC)

Abstract 4133

Jean-Luc Van Laethem[1], Ivan Borbath[2], Karen Paula Geboes[3], Philippe Alexandre Cassier[4], Aurélien Lambert[5], Emmanuel Mitry[6], Hans Prenen[7], Lorenzo Pilla[8], Jean-Frédéric Blanc[9], Inmaculada Gallego Jiménez[10], Roberto A. Pazo Cid[11], Mercedes Rodríguez Garrote[12], Jaime Feliú[13], Karin Nordbladh[14], Karin Enell Smith, David Gomez Jimenez[14], Peter Ellmark[14], Yago Pico de Coaña[14], Sumeet Vijay Ambarkhane[14], Teresa Macarulla[15]

1. Erasme University Hospital, Brussels, Belgium; 2. Saint-Luc University Clinics, Brussels, Belgium; 3. Department of Gastroenterology, Division of Digestive Oncology, Ghent University Hospital, Ghent, Belgium; 4. Centre Léon Bérard, Lyon, France; 5. Institut De Cancerologie De Lorraine, Vandoeuvre Les Nancy, France; 6. Institut Paoli-Calmettes, Marseille, France; 7. University Hospital Gasthuisberg, Leuven, Belgium; 8. Department of Gastroenterology and Gastrointestinal Oncology, Hôpital Européen Georges-Pompidou, AP-HP, Université de Paris, Paris, France; 9. Hôpital Haut-Lévêque, Pessac, France; 10. Hospital Universitario Virgen del Rocío, Sevilla, Spain; 11. Hospital Universitario Miguel Servet, Zaragoza, Spain; 12. Hospital Universitario Ramón y Cajal, Madrid, Spain; 13. Department of Medical Oncology, Hospital Universitario La Paz, Madrid, Spain; 14. Alligator Bioscience AB, Lund, Sweden; 15. Vall d'Hebron University Hospital and Vall d'Hebron Institute of Oncology, Barcelona, Spain

Mitazalimab, a CD40 agonist with best-in-class profile

- Mitazalimab is a **FcγR crosslinking dependent CD40 agonistic** antibody (IgG1) with a tumor-directed immune activation
- Mitazalimab binds with **high affinity** to a unique binding epitope on the CD40 receptor allowing for high efficacy and potency
- Mitazalimab has the **potential for superior clinical activity:** mitazalimab is the only FcγR-dependent CD40 agonist with tumor-directed activity that can be dosed at 0.9 mg/kg

Mitazalimab in pancreatic cancer

DAY 1 Lead-in
Mitazalimab activates macrophages for stromal degradation.

DAY 8 Chemotherapy
mFOLFIRINOX induces tumor cell death, releasing antigens that are presented to the immune system

DAY 10 Mitazalimab primes for immunity
Enhanced priming of tumor-specific T cells via activation of dendritic cells (DCs).

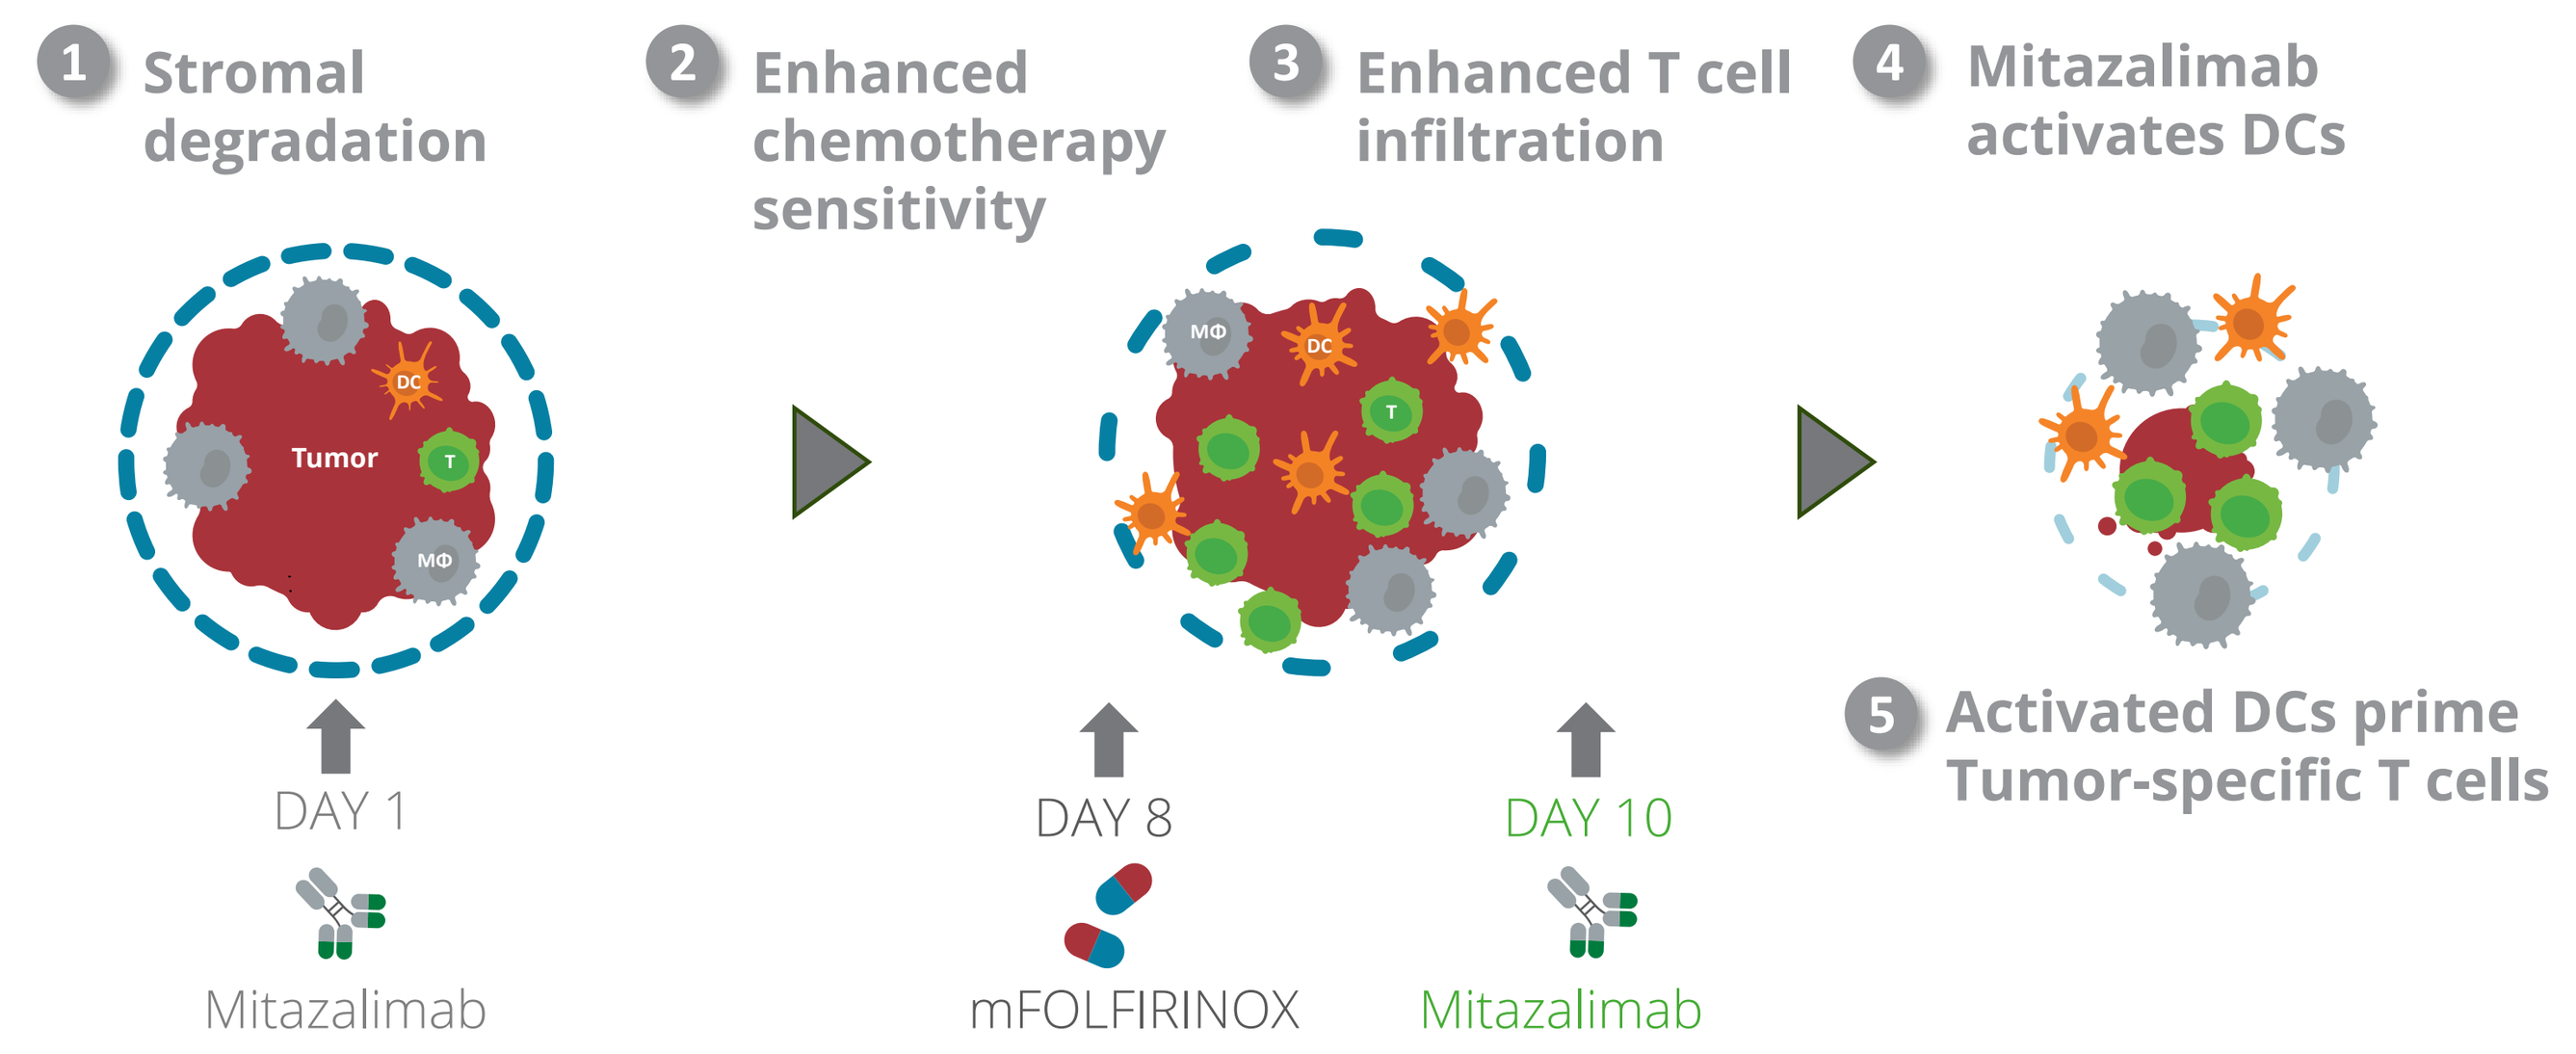
1 Stromal degradation
2 Enhanced chemotherapy sensitivity
3 Enhanced T cell infiltration
4 Mitazalimab activates DCs
5 Activated DCs prime Tumor-specific T cells
Tumor
DC
T
MΦ
DAY 1
Mitazalimab
DAY 8
mFOLFIRINOX
DAY 10
Mitazalimab

OPTIMIZE-1 study overview

OPTIMIZE-1 (NCT04888312) is a phase 1b/2, open-label, multicenter study designed to evaluate safety, tolerability, and efficacy of mitazalimab in combination with mFOLFIRINOX in adults diagnosed with previously untreated metastatic PDAC.

Key eligibility criteria include: Histologically confirmed diagnosis of metastatic PDAC, ECOG 0-1, No prior chemotherapy for PDAC

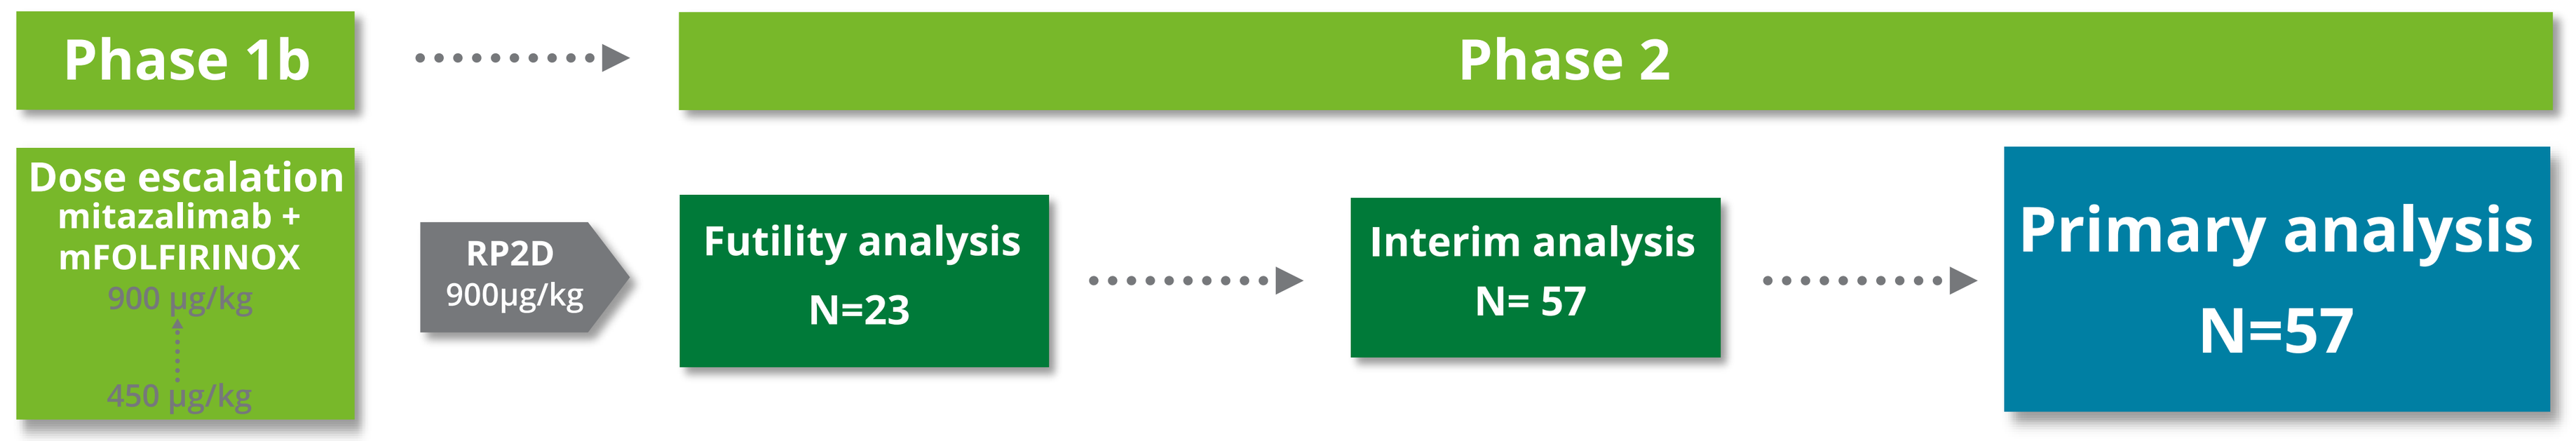
Phase 1b
Phase 2
Dose escalation mitazalimab + mFOLFIRINOX
900 μg/kg
450 μg/kg
RP2D 900μg/kg
Futility analysis N=23
Interim analysis N= 57
Primary analysis N=57

Dosing Regimen

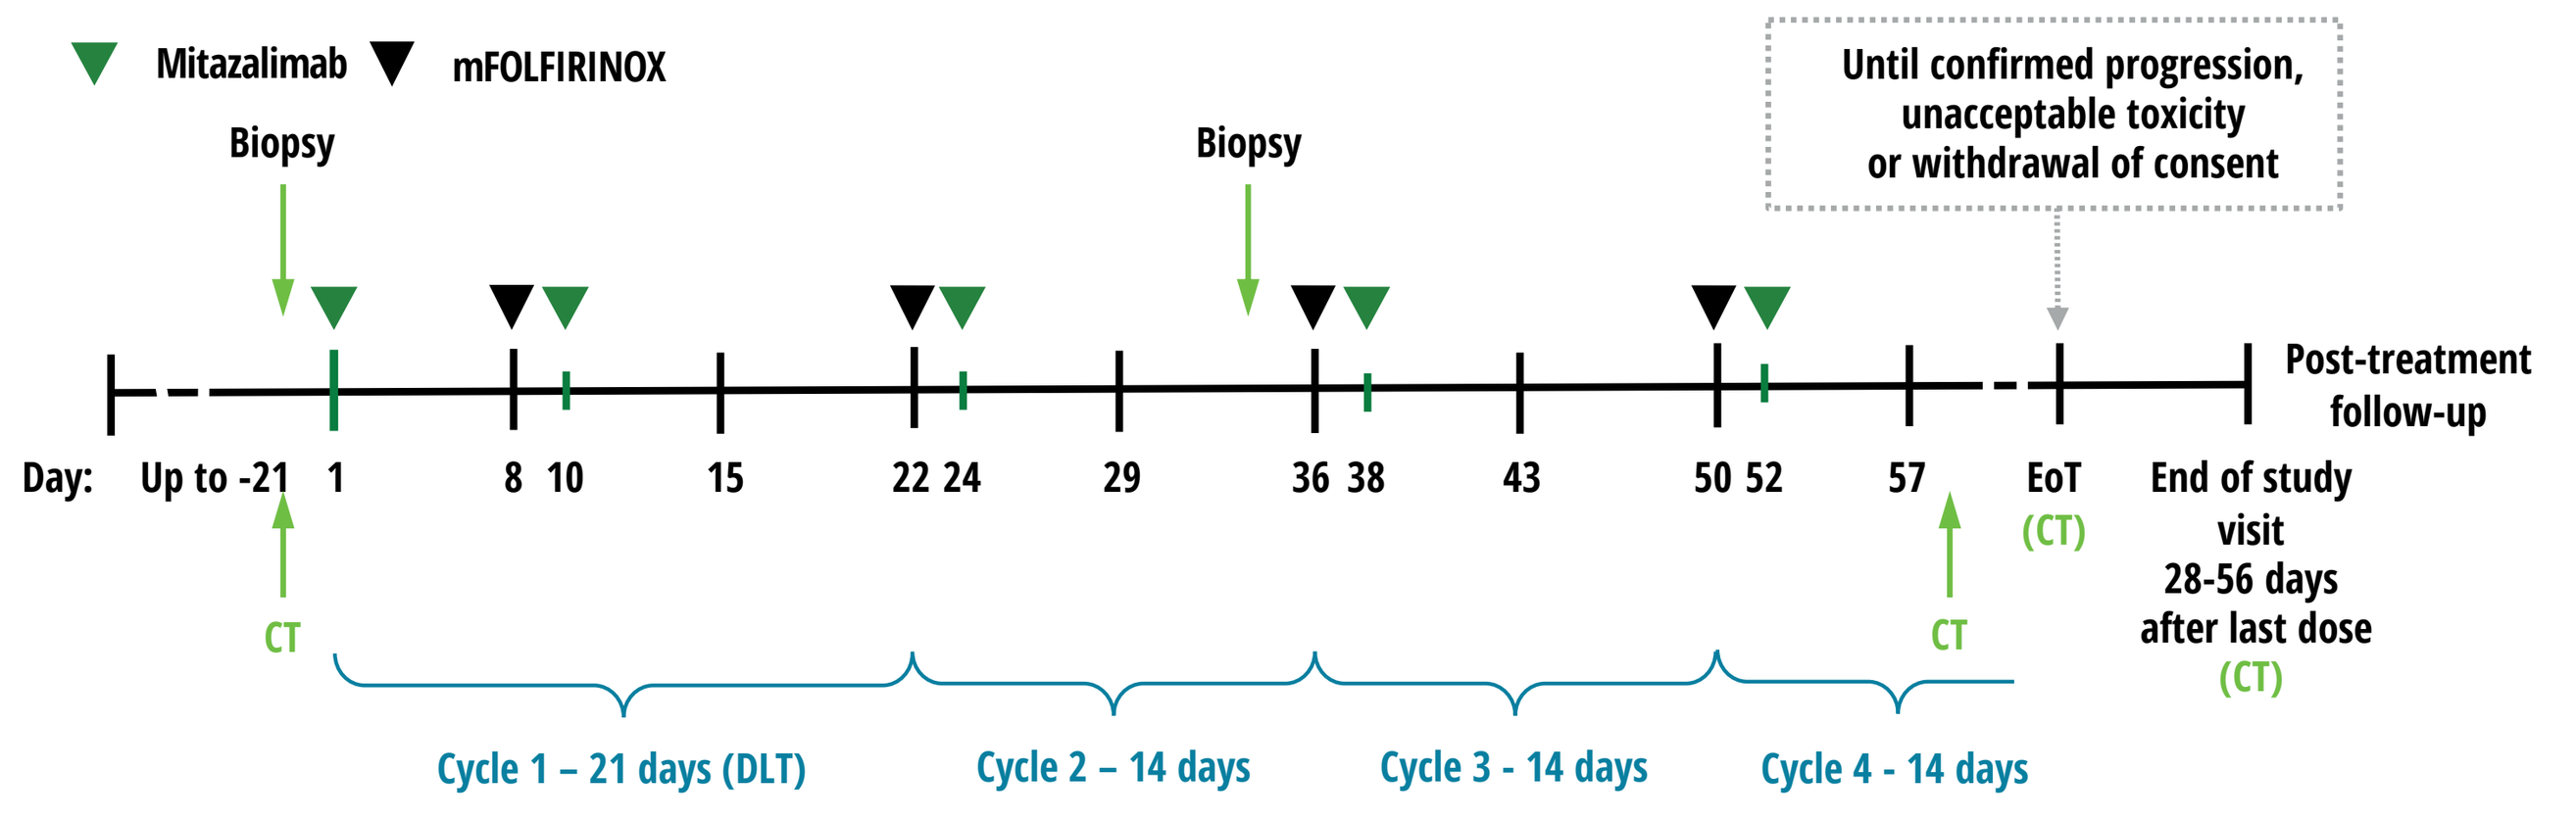
Mitazalimab
mFOLFIRINOX
Biopsy
Biopsy
Until confirmed progression, unacceptable toxicity or withdrawal of consent
Post-treatment follow-up
Day: Up to -21 1 8 10 15 22 24 29 36 38 43 50 52 57
EoT (CT)
End of study visit 28-56 days after last dose (CT)
CT
CT
Cycle 1 – 21 days (DLT)
Cycle 2 – 14 days
Cycle 3 - 14 days
Cycle 4 - 14 days

Objectives and Endpoints

Primary objective: To assess the clinical activity of mitazalimab in combination with chemotherapy as determined by ORR.

Secondary objectives include: Survival outcomes, best overall response, duration of response, disease control rate, time to next anti-cancer therapy, safety and PK analysis. PD biomarkers will be analyzed as exploratory endpoints.

Primary endpoint: ORR compared to historical FOLFIRINOX (30%)

Primary endpoint was met: Confirmed ORR 40.4% (unconfirmed ORR 50.9%)

Mitazalimab + mFOLFIRINOX demonstrated promising durability of response (median DoR 12.5 months) and overall survival (mOS 14.3 months)

These encouraging results support continued development in a confirmatory trial

Patient Baseline Characteristics

	Safety set 450 μg/kg N=5	Safety set 900 μg/kg N=65	Efficacy Set 900 μg/kg N=57
Age, years			
Under 65	2 (40.0)	38 (58.5)	35 (61.4%)
65 or above	3 (60.0)	27 (41.5)	22 (38.6%)
Gender			
Female	3 (60.0)	38 (58.5)	33 (57.9%)
Male	2 (40.0)	27 (41.5)	24 (42.1%)
Days from diagnosis to treatment start; median (IQR)	21 (20-33)	29 (21–35)	30 (21.8-35%)
ECOG performance status			
Grade 0	3 (60.0)	35 (53.8)	31 (54.4%)
Grade 1	2 (40.0)	30 (46.2)	26 (45.6%)
CA19-9 at baseline (n)	5	56	48
<100	1 (20.0)	8 (14.3)	8 (16.7%)
100-1000	0 (0.0)	13 (23.2)	13 (27.1%)
>1000	4 (80.0)	35 (62.5)	27 (56.3%)
Neutrophil/lymphocyte ratio (n)	5	64	56
<5	5 (100.0)	52 (81.3)	46 (82.1%)
>5	0 (0.0)	12 (18.8)	10 (17.9%)

	Safety set 450 μg/kg N=5	Safety set 900 μg/kg N=65	Efficacy Set 900 μg/kg N=57
Prior surgery			
Yes	1 (20.0)	2 (3.1)	2 (3.5%)
No	4 (80.0)	63 (96.9)	55 (96.5%)
Primary tumour location (n)	4	63	55
Head	0 (0.0)	29 (46.0)	25 (45.5%)
Body	1 (25.0)	22 (34.9)	21 (38.2%)
Tail	3 (75.0)	12 (19.0)	9 (16.4%)
Number of target lesions			
1	0 (0.0)	12 (18.5)	12 (17.1%)
2	2 (40.0)	19 (29.2)	21 (30.0%)
3 or more	3 (60.0)	34 (52.3)	37 (52.9%)
Sites of metastatic disease			
Liver	5 (100.0)	55 (78.6%)	42 (73.7%)
Lung	1 (20.0)	12 (17.1%)	9 (15.8%)
Peritoneal	1 (20.0)	10 (14.3%)	9 (15.8%)
Lymph Node	2 (40.0)	14 (20.0%)	10 (17.5%)
Other	2 (40.0)	20 (28.6%)	16 (28.1%)

Primary Endpoint

Overall response	Efficacy Evaluable (n=57)
ORR, n, % (90% CI)	29, 50.9% (39.3-62.4)
ORR (confirmed), n, % (90% CI)	23, 40.4% (29.4-52.1)
Best Overall Response (confirmed) n (%)	
Complete Response	1 (1.8%)
Partial Response	22 (38.6%)
Stable Disease	22 (38.6%)
Progressive Disease	12 (26.3%)
Disease Control Rate	45 (78.9%)

At cutoff (November 14, 2023), 57 patients treated with mFOLFIRINOX + 900 μg/kg mitazalimab had received ≥2 cycles and were evaluable for efficacy analysis.

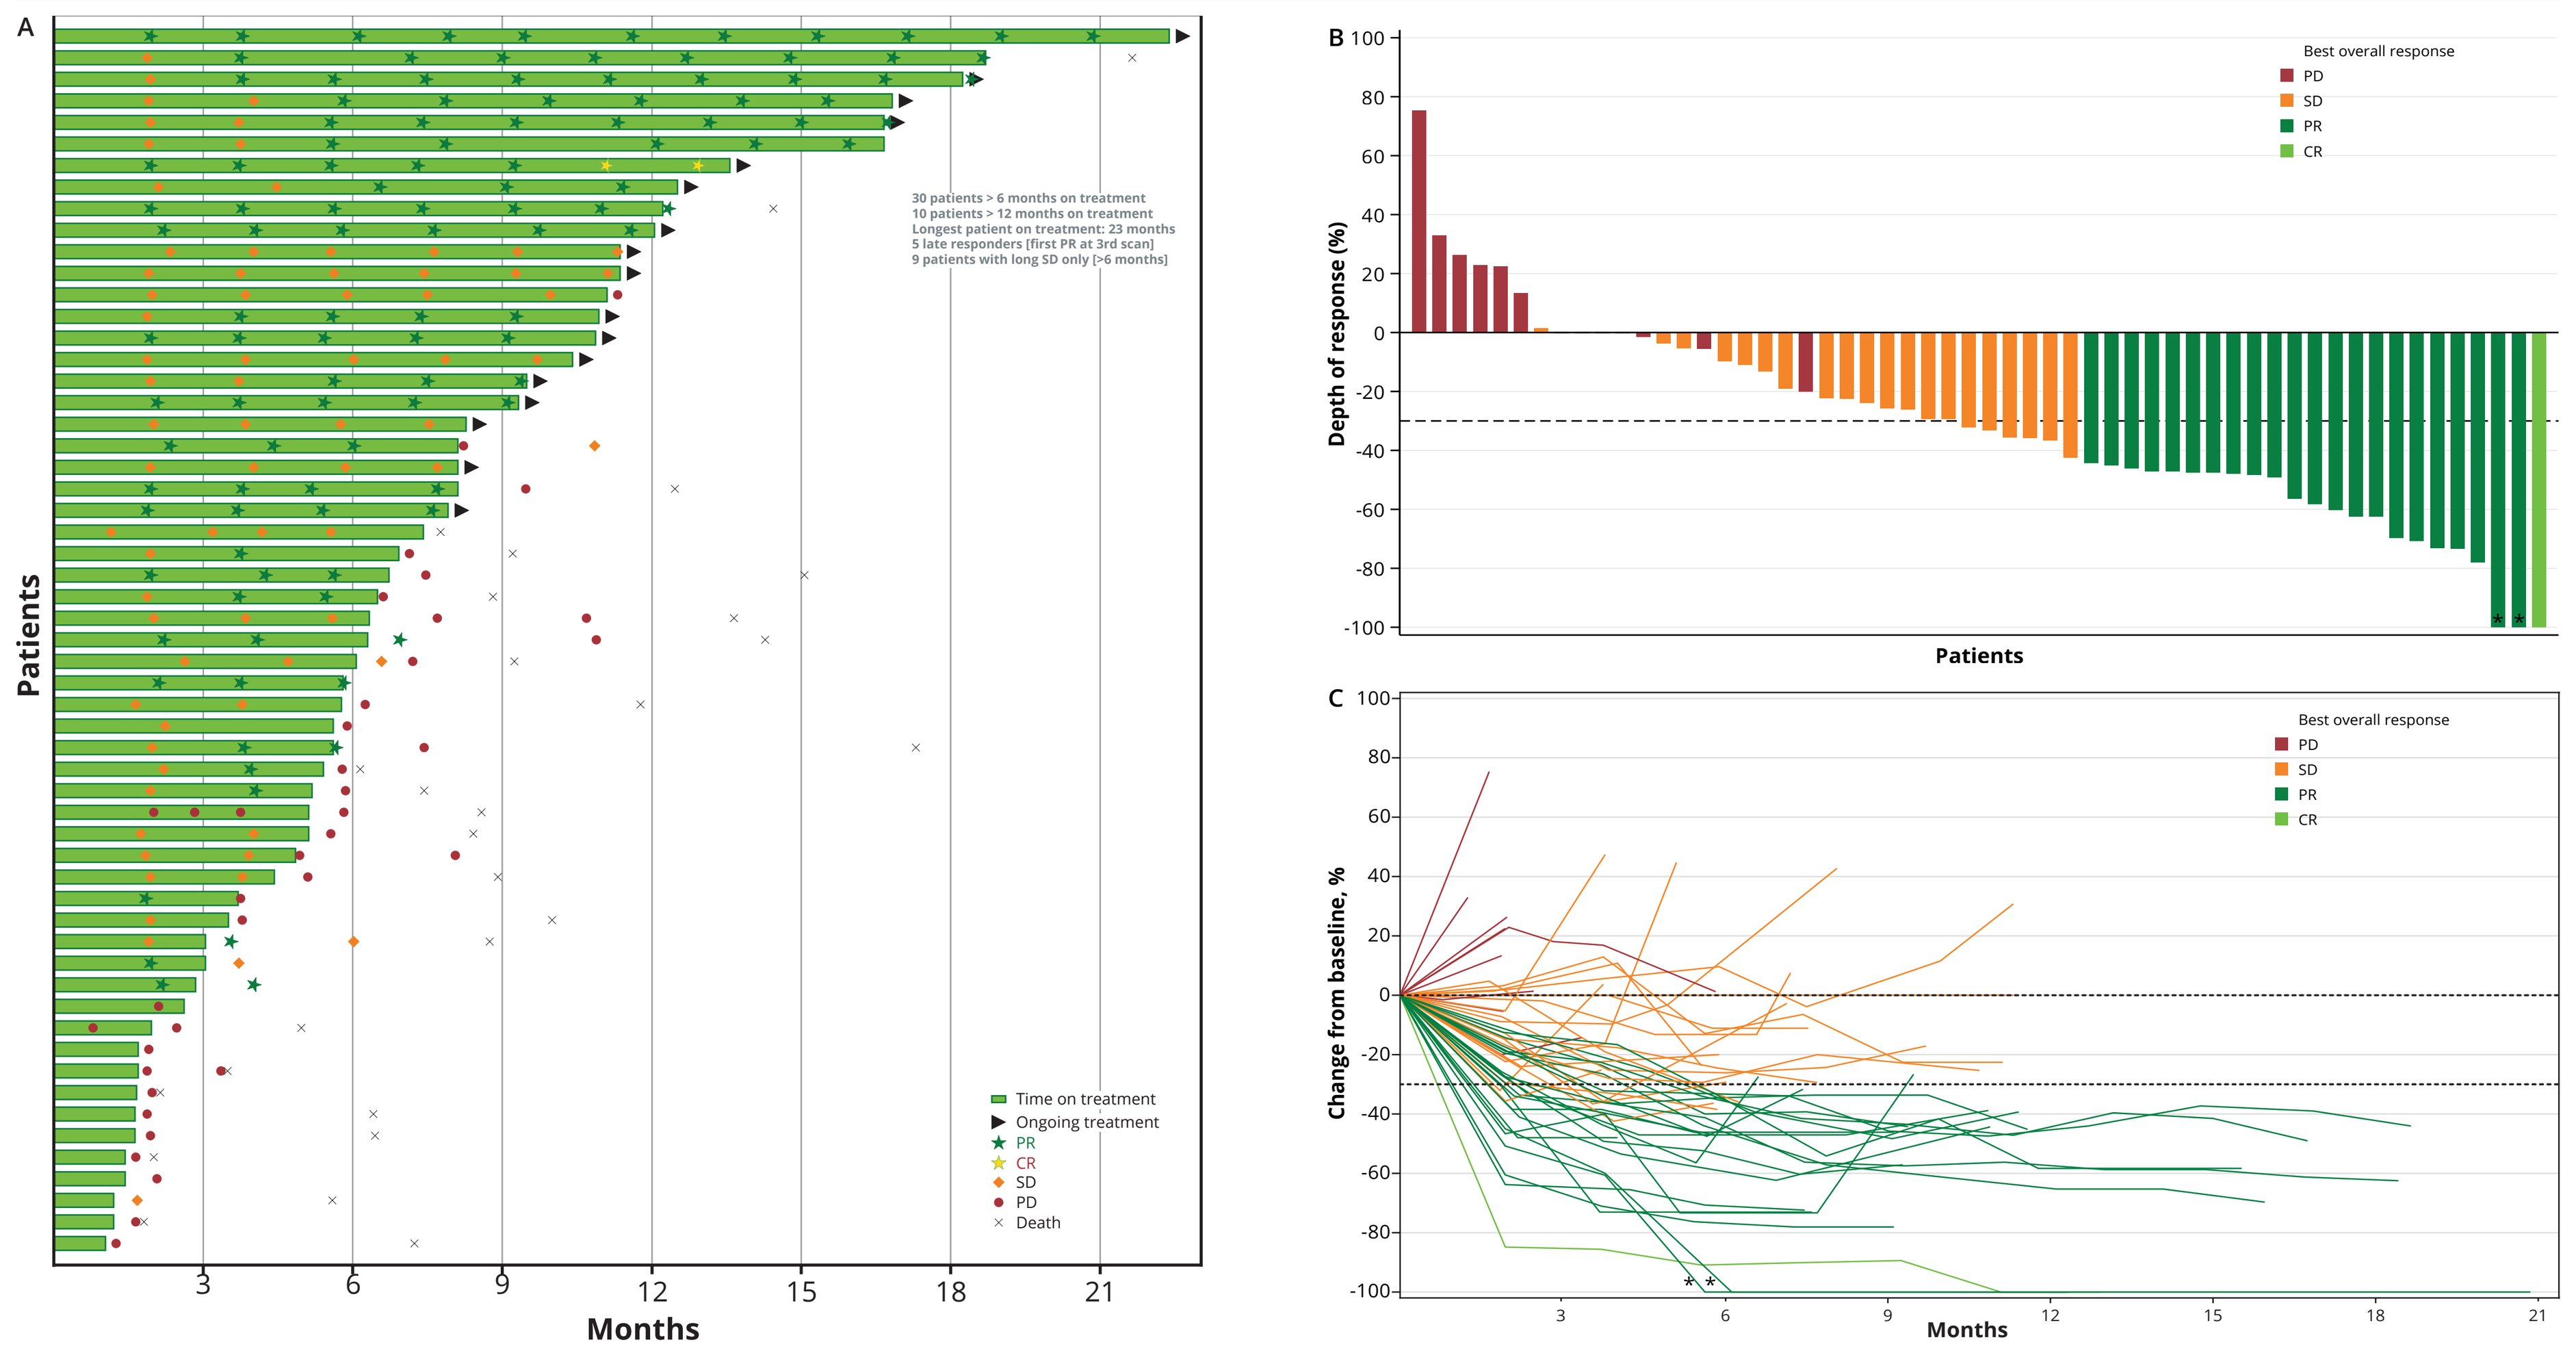
A
Patients
Months
30 patients > 6 months on treatment
10 patients > 12 months on treatment
Longest patient on treatment: 23 months
5 late responders [first PR at 3rd scan]
9 patients with long SD only [>6 months]
Time on treatment
Ongoing treatment
PR
CR
SD
PD
Death
B
Depth of response (%)
Patients
Best overall response
PD
SD
PR
CR
C
Change from baseline, %
Months
Best overall response
PD
SD
PR
CR

(A) Swimmer plot showing the individual patient treatment durations for all activity-evaluable patients. (B) Waterfall plot showing the maximum percentage change in the summed tumour size relative to the sum of baseline in individual patients' target lesions by best overall response. (C) Spider plot showing percent change from baseline in summed tumour target lesions over time. *Two partial responders demonstrated a complete response in target lesions

Safety

- At cutoff, 70 patients were treated with mFOLFIRINOX and 450 μg/kg (N=5) or 900 μg/kg (N=65) mitazalimab and evaluated for safety
- Mitazalimab + mFOLFIRINOX has a manageable safety profile
- The overall safety profile is consistent with mFOLFIRINOX chemotherapy
- No new safety signals or evidence of additive toxicity
- Eleven patients (15.7%) experienced mitazalimab related grade >3 AEs
- Four (5.7%) patients discontinued study treatment due to TEAEs, one with fatigue, chest pain, and headache, one with general deterioration in physical health, one with pneumonia, and one with an IRR (pruritus)

Patients with Grade ≥3 AEs (any causality) n (%)	N=70
Any Grade ≥3 AEs	**55 (78.6)**
Occurring in ≥5% patients:	
Neutropenia	18 (25.7)
Hypokalaemia	11 (15.7)
Anaemia	8 (11.4)
Thrombocytopenia	8 (11.4)
Fatigue	7 (10.0)
Diarrhoea	6 (8.6)
Neuropathy peripheral	6 (8.6)
Nausea	4 (5.7)
Asthenia	4 (5.7)

Overall Survival

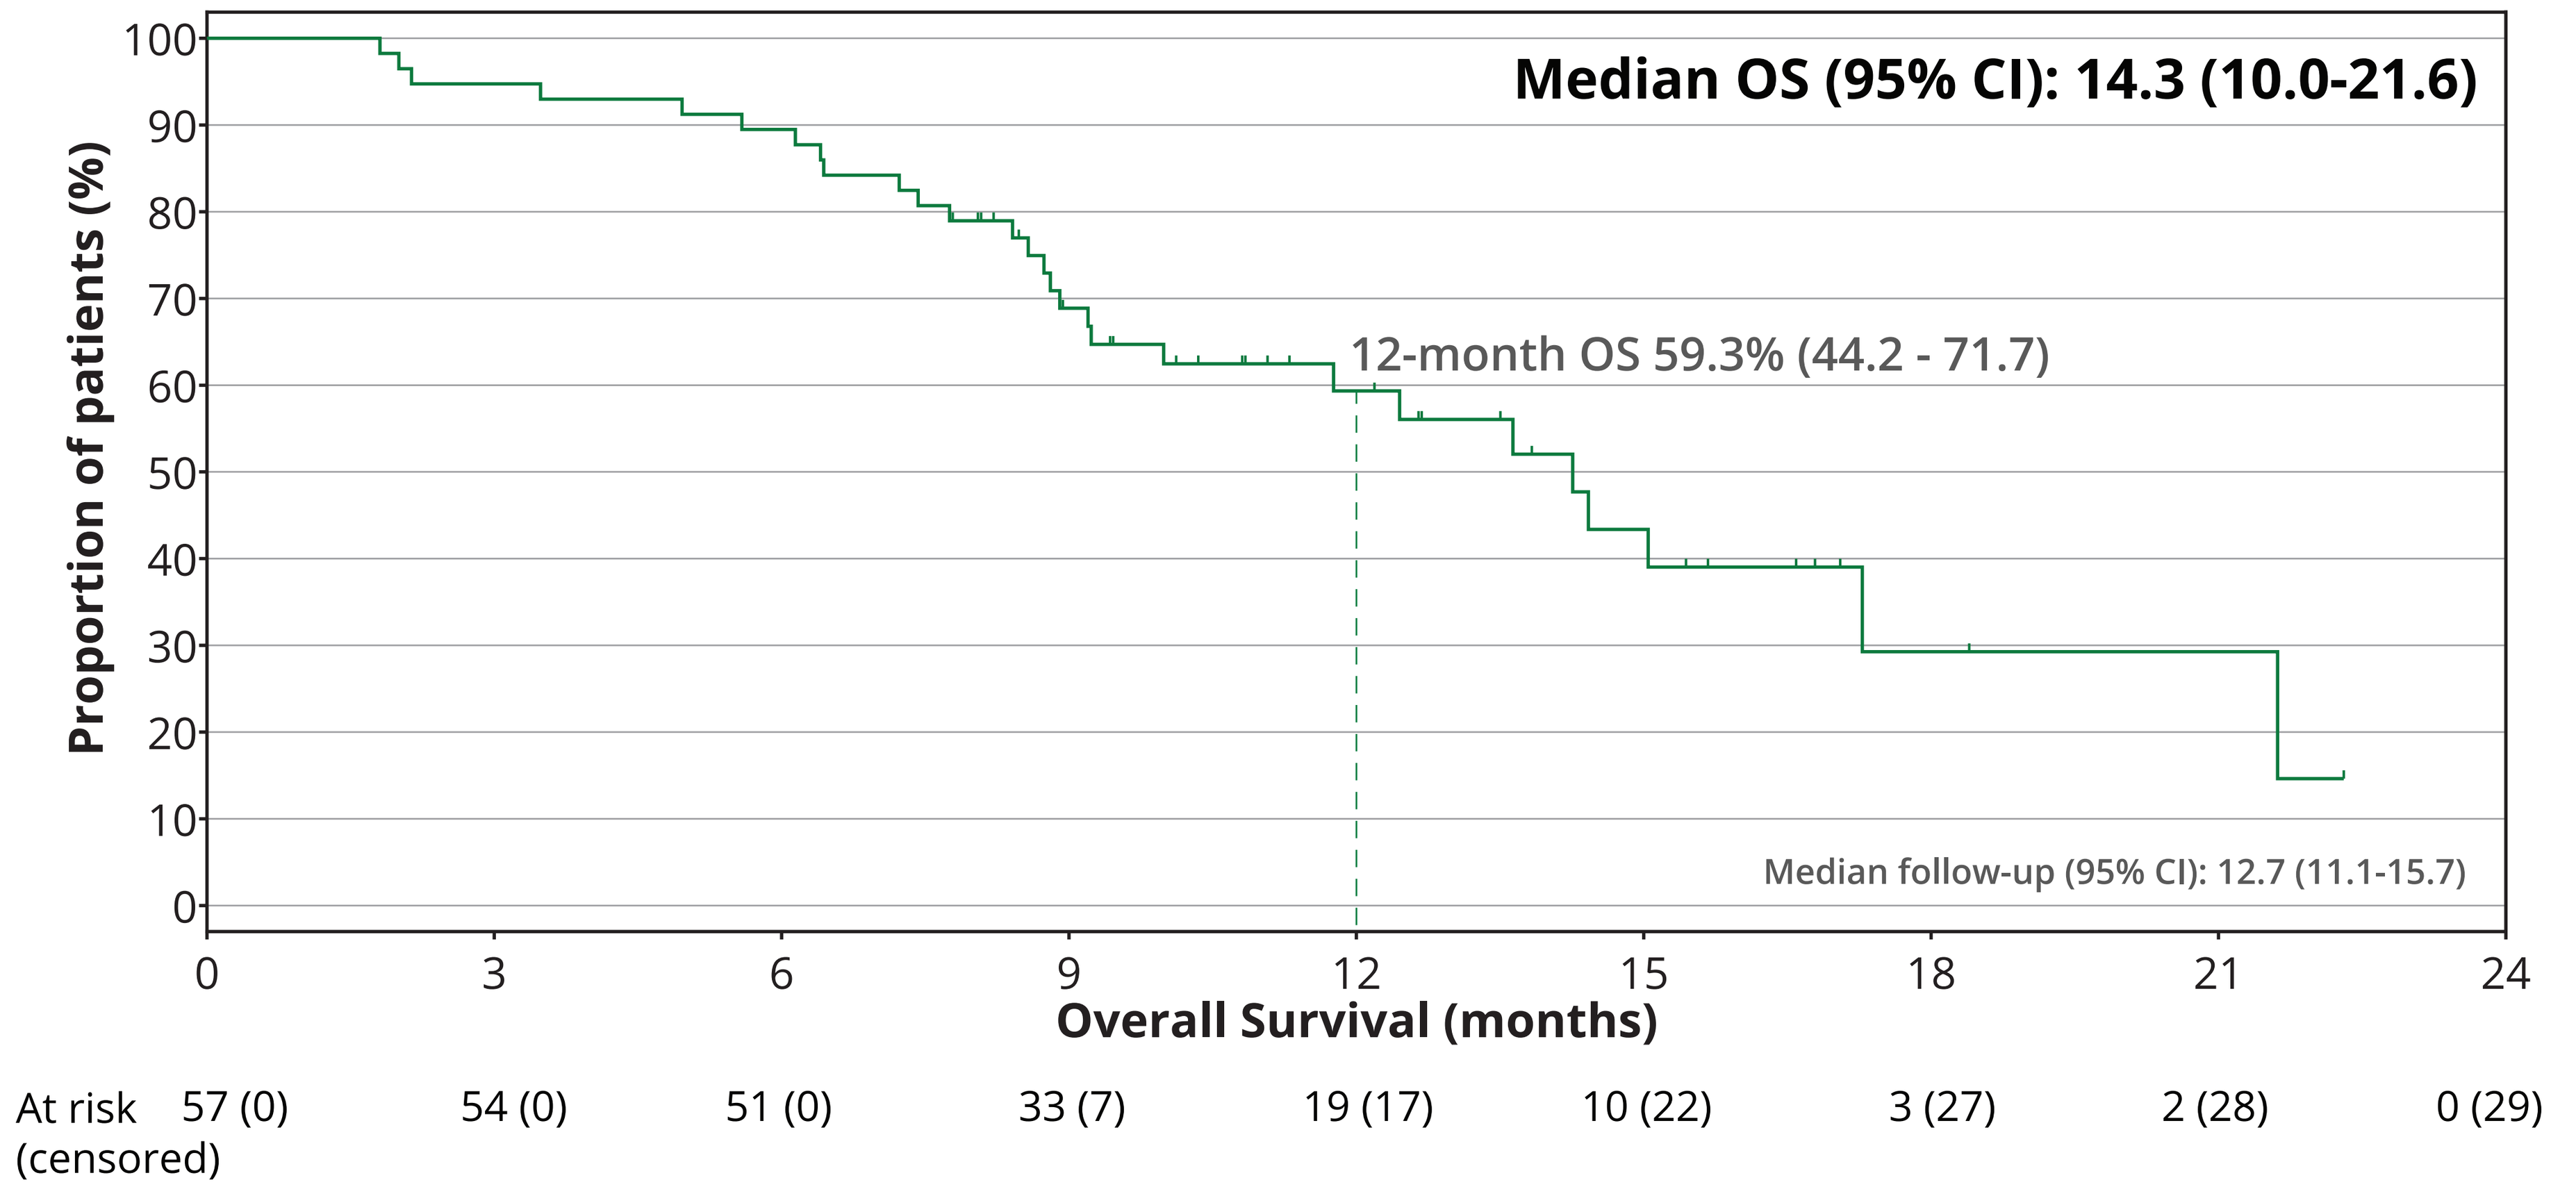
Median OS (95% CI): 14.3 (10.0-21.6)
12-month OS 59.3% (44.2 - 71.7)
Median follow-up (95% CI): 12.7 (11.1-15.7)
Proportion of patients (%)
Overall Survival (months)
At risk (censored) 57 (0) 54 (0) 51 (0) 33 (7) 19 (17) 10 (22) 3 (27) 2 (28) 0 (29)

Duration of Response

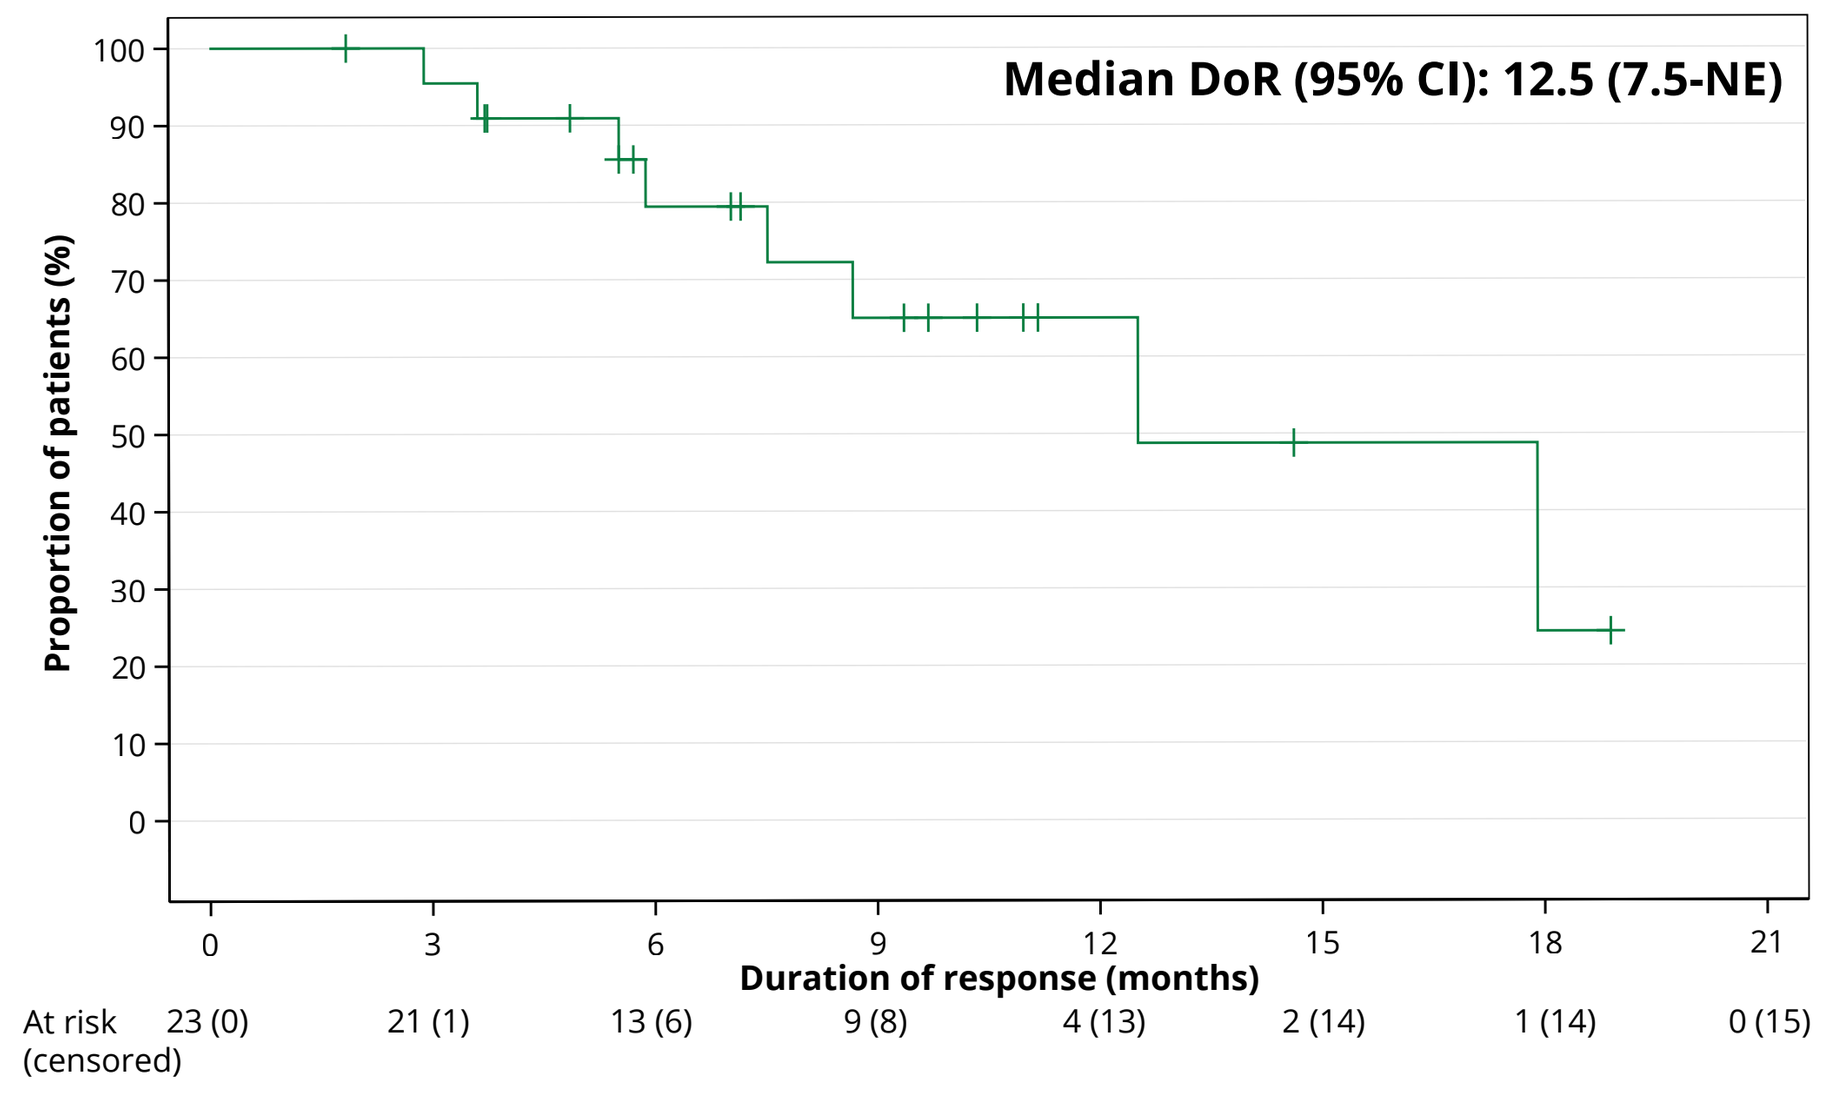
Median DoR (95% CI): 12.5 (7.5-NE)
Proportion of patients (%)
Duration of response (months)
At risk (censored) 23 (0) 21 (1) 13 (6) 9 (8) 4 (13) 2 (14) 1 (14) 0 (15)

Progression Free Survival

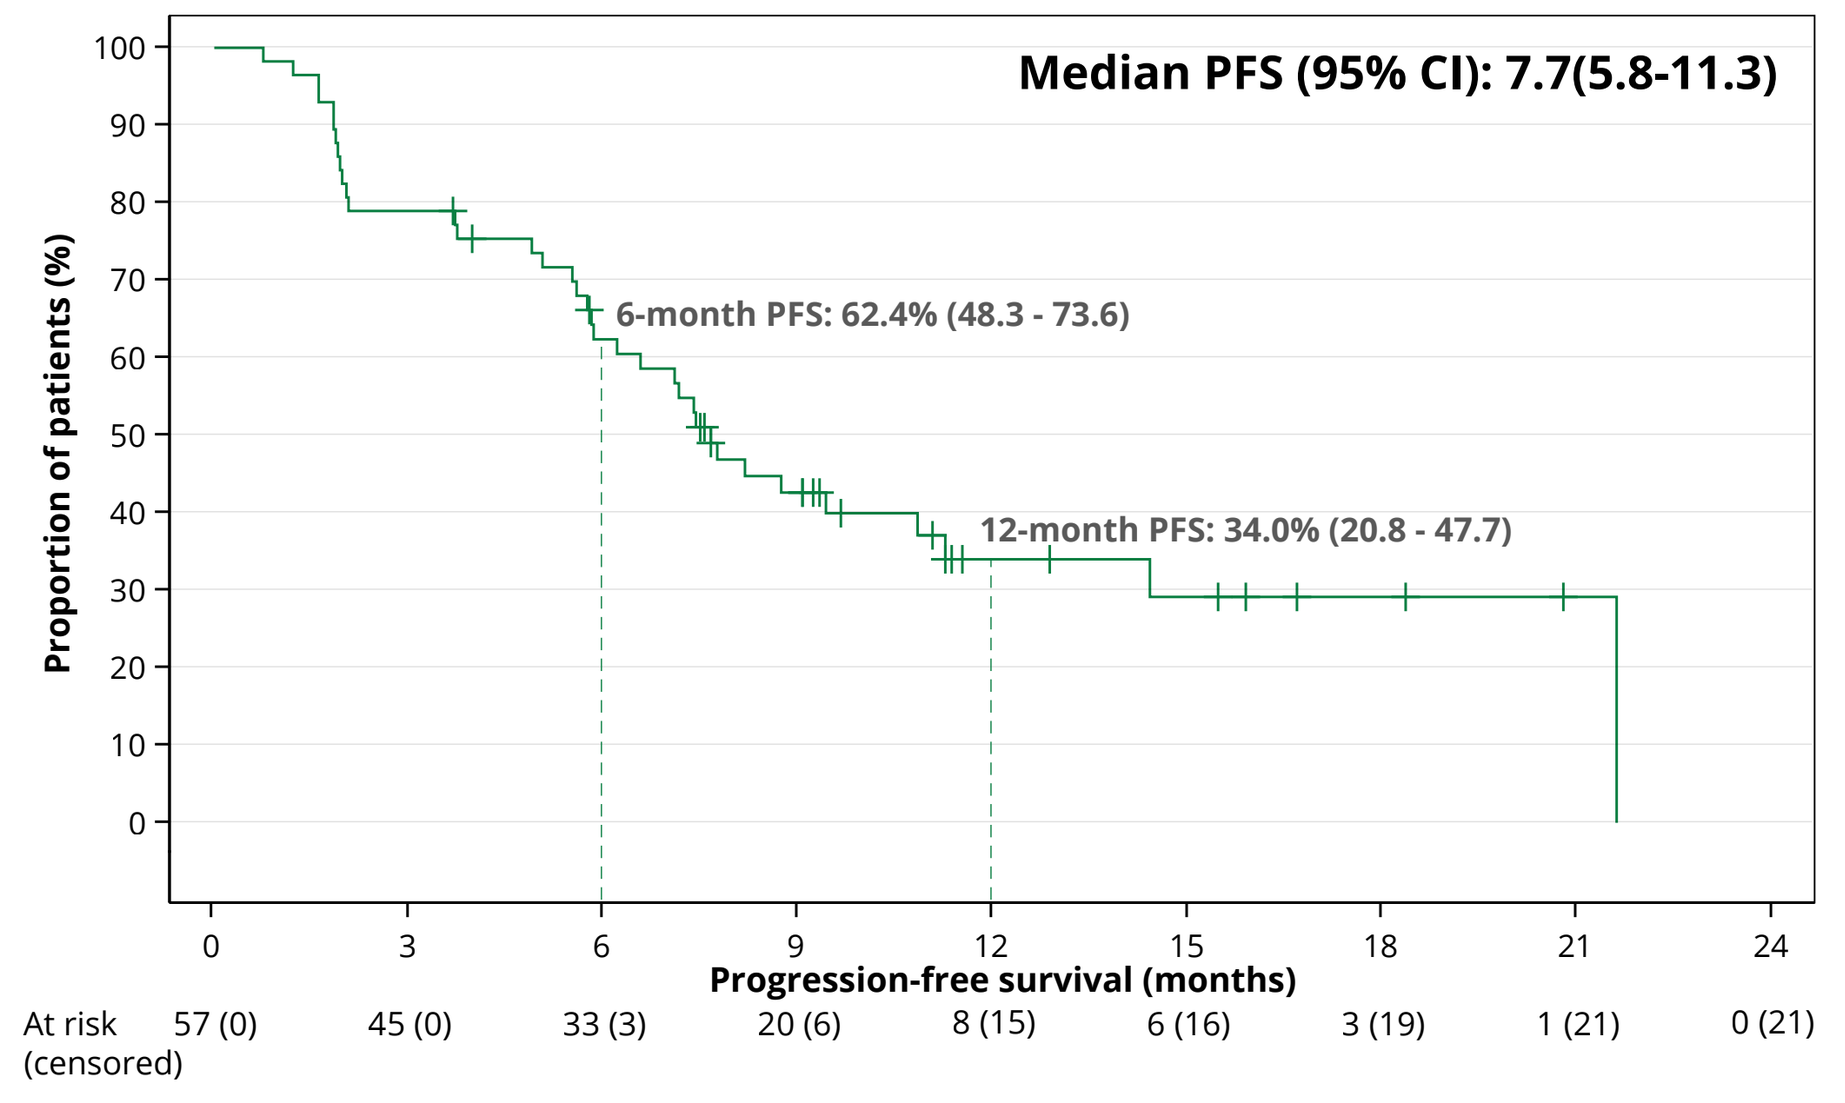
Median PFS (95% CI): 7.7(5.8-11.3)
6-month PFS: 62.4% (48.3 - 73.6)
12-month PFS: 34.0% (20.8 - 47.7)
Proportion of patients (%)
Progression-free survival (months)
At risk (censored) 57 (0) 45 (0) 33 (3) 20 (6) 8 (15) 6 (16) 3 (19) 1 (21) 0 (21)

KRAS mutation subgroup analysis

n(%(95%CI))	KRAS[a] G12D (n=20)	KRAS G12V (n=21)	KRAS G12R (n=5)	Efficacy Evaluable (n=57)
ORR (confirmed)	5* (25 (10.4 - 45.6))	13 (61.9 (41.7 - 79.4))	3 (60 (18.9 – 92.4))	23* (40.4(29.4 - 52.1))
Median DoR, mo	NE	12.5 (7.5-NE)	NE	12.5(7.5-NE)
Median PFS, mo	5.7 (1.9 - 8.2)	10.8 (6.2- NE)	7.8 (5.7- NE)	7.7 (5.8 - 11.3)
6 mo PFS, %	45 (23.1 - 64.7)	76.2 (51.9 - 89.3)	80 (20.4 - 96.9)	62.4 (48.3 - 73.6)
Median OS, mo	9.2 (6.4- NE)	14.4 (11.8- NE)	15 (6.1- NE)	14.3 (10 - 21.6)
12 mo OS, %	39 (16.9 - 60.8)	69.3 (43.2 - 85.2)	60(12.6 - 88.2)	59.3 (44.2 - 71.7)

*1 CR- [a]KRAS mutations not detected in 11pts- NE=Not Estimable

ACKNOWLEDGEMENTS:
We would like to show our gratitude to the patients, their families and the clinical research staff who are making this trial possible.

Correspondence to: yas@alligatorbioscience.com

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from the author of this poster.